Poster

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from AACR or the author of this poster.

#CT057

Phase 2, Multicenter Open-label Basket Trial of *nab*-Sirolimus for Patients With Inactivating Alterations in *TSC1* or *TSC2* (PRECISION I)

Jordi Rodon Ahnert, MD, PhD[1]; Brian Schulte, MD[2]; Michael J. Demeure, MD[3]; Dustin Deming, MD[4]; Noah Federman, MD[5]; Meredith A. McKean, MD[6]; Elizabeth Lee, MD[7]; Alexander Spira, MD, PhD[8]; David J. Kwiatkowski, MD, PhD[9]; Maen Hussein, MD[10]; Erlinda M. Gordon, MD[11]; David Crockett, MD[12]; Kristen Ganjoo, MD[13]; Lee D. Cranmer, MD, PhD[14]; Li Ding, MS, MA[15]; Anita N. Schmid, PhD[15]; Willis H. Navarro, MD[15]; Loretta M. Itri, MD[15]; Gopa Iyer, MD[16]

[1]MD Anderson Cancer Center, Houston, TX; [2]UCSF Helen Diller Family Comprehensive Cancer Center, San Francisco, CA; [3]Hoag Memorial Hospital Presbyterian, Newport Beach, CA; [4]University of Wisconsin, Madison, WI; [5]University of California, Los Angeles, CA; [6]Sarah Cannon and HCA Research Institute, Nashville, TN; [7]Dana-Farber Cancer Institute, Boston, MA; [8]Virginia Cancer Specialists, Fairfax, VA; [9]Brigham and Women's Hospital, Boston, MA; [10]Florida Cancer Specialists North Division, St. Petersburg, FL; [11]Sarcoma Oncology Research Center, Santa Monica, CA; [12]Nebraska Cancer Specialists, Omaha, NE; [13]Stanford Cancer Center, Stanford, CA; [14]Fred Hutchinson Cancer Center, Seattle, WA; [15]Aadi Bioscience, Pacific Palisades, CA; [16]Memorial Sloan Kettering Cancer Center, New York, NY

Trial Summary

Copies of this trial summary through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

INTRODUCTION

- Tuberous sclerosis complex subunit 1 or 2 (*TSC1* or *TSC2*) are critical negative regulators of mechanistic target of rapamycin (mTOR) complex 1 activation[1] (**Figure 1**)

Figure 1. PI3K-Akt-mTOR Pathway[2,3]

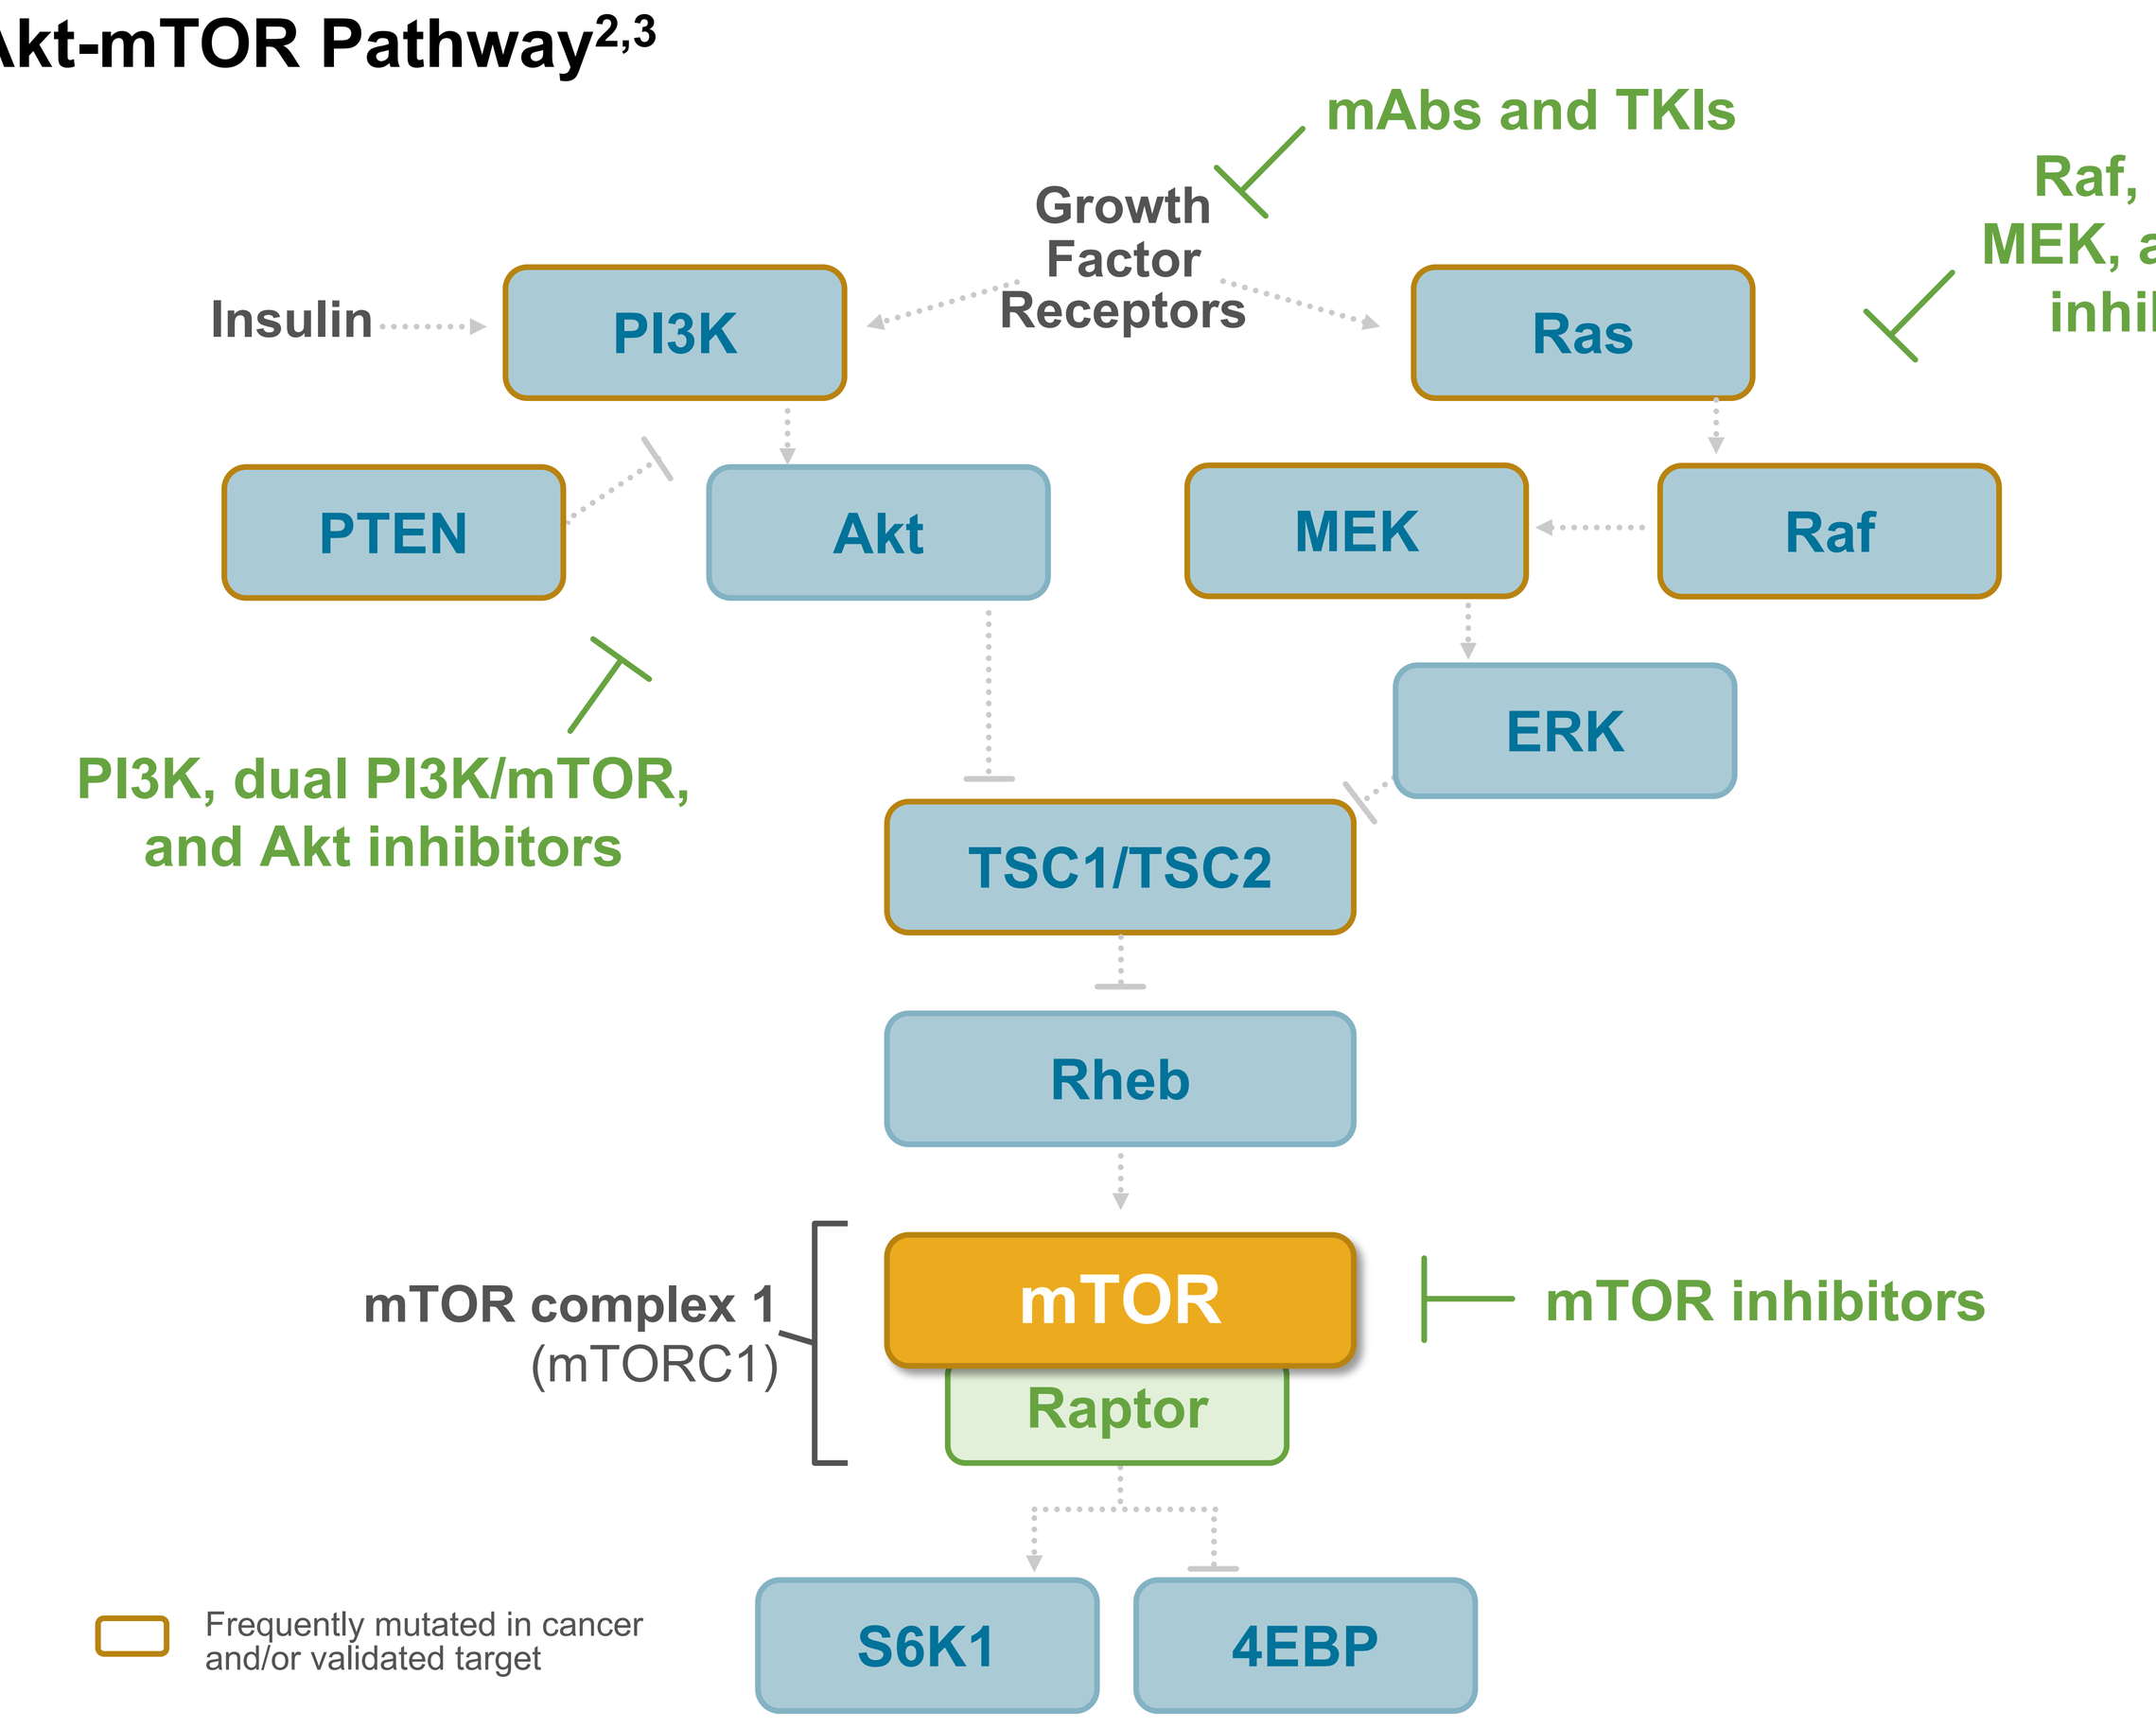

mAbs and TKIs
Growth Factor Receptors
Insulin
PI3K
Ras
Raf, B-Raf, MEK, and ERK inhibitors
PTEN
Akt
MEK
Raf
ERK
PI3K, dual PI3K/mTOR, and Akt inhibitors
TSC1/TSC2
Rheb
mTOR complex 1 (mTORC1)
mTOR
Raptor
mTOR inhibitors
Frequently mutated in cancer and/or validated target
S6K1
4EBP

4EBPI, eukaryotic translation initiation factor 4E-binding protein; Akt, protein kinase B; Erk, extracellular signal-regulated kinase; Mek, mitogen-activated protein kinase kinase; mTOR, mechanistic target of rapamycin; mTORC1, mTOR complex 1; *nab*, nanoparticle albumin-bound; PI3K, phosphatidylinositol 3-kinase; PTEN, phosphatase and tensin homolog; Raf, rapidly accelerated fibrosarcoma; Raptor, regulatory-associated protein of mTOR; Ras, rat sarcoma virus homolog; Rheb, Ras homolog enriched in brain; S6K, ribosomal S6 kinase; *TSC1/TSC2*, tuberous sclerosis complex subunit 1 or 2.

- Inactivating alterations in *TSC1* and/or *TSC2* have been observed in several types of cancer, but no treatment options exist specifically for patients with *TSC1* or *TSC2* inactivating alterations (**Table 1**)

Table 1. Estimated Frequency of Definite Impact *TSC1* or *TSC2* Alterations

Tumor Type	*TSC1* Alterations, %[a]	*TSC2* Alterations, %	*TSC1* or *TSC2* Combined, %
Bladder	6.33	1.70	8.03
Hepatobiliary	1.27	3.31	4.58
Endometrial	2.10	1.22	3.32
Soft tissue sarcoma	1.28	1.71	2.99
Ovarian	1.85	0.92	2.77
Esophagogastric	0.65	1.46	2.11
Non-small cell lung cancer	0.77	1.16	1.93
Colorectal carcinoma	0.99	0.39	1.38
Breast	0.41	0.10	0.51

[a]The proportion of patients with definite impact alterations (ie, alterations known to have a biological impact, including frameshift, nonsense, and splice-site mutations and deep deletions) derived from analysis of TCGA and cBioPortal by Gulati et al. (Data on file). *TSC1*, *TSC2*, tuberous sclerosis complex subunit 1, 2.

- The utility of oral mTOR inhibitors (mTORis), such as sirolimus, as pan-cancer agents may be restricted by low bioavailability and dose-limiting toxicity[2,4]
- To improve the pharmacologic properties of sirolimus, *nab*-sirolimus, a nanoparticle form of human albumin-bound sirolimus, was developed for intravenous (IV) use
- In preclinical animal models, *nab*-sirolimus demonstrated significantly higher intratumor drug concentrations, greater tumor growth inhibition, improved survival, and greater inhibition of the downstream marker of mTOR activity, phosphorylated-S6 (pS6) ribosomal protein, relative to equal weekly doses of sirolimus and everolimus (**Figure 2**)

Figure 2. (A) Intratumor Drug Concentration, (B) Tumor Growth Inhibition, (C) Survival, and (D) mTOR Activity

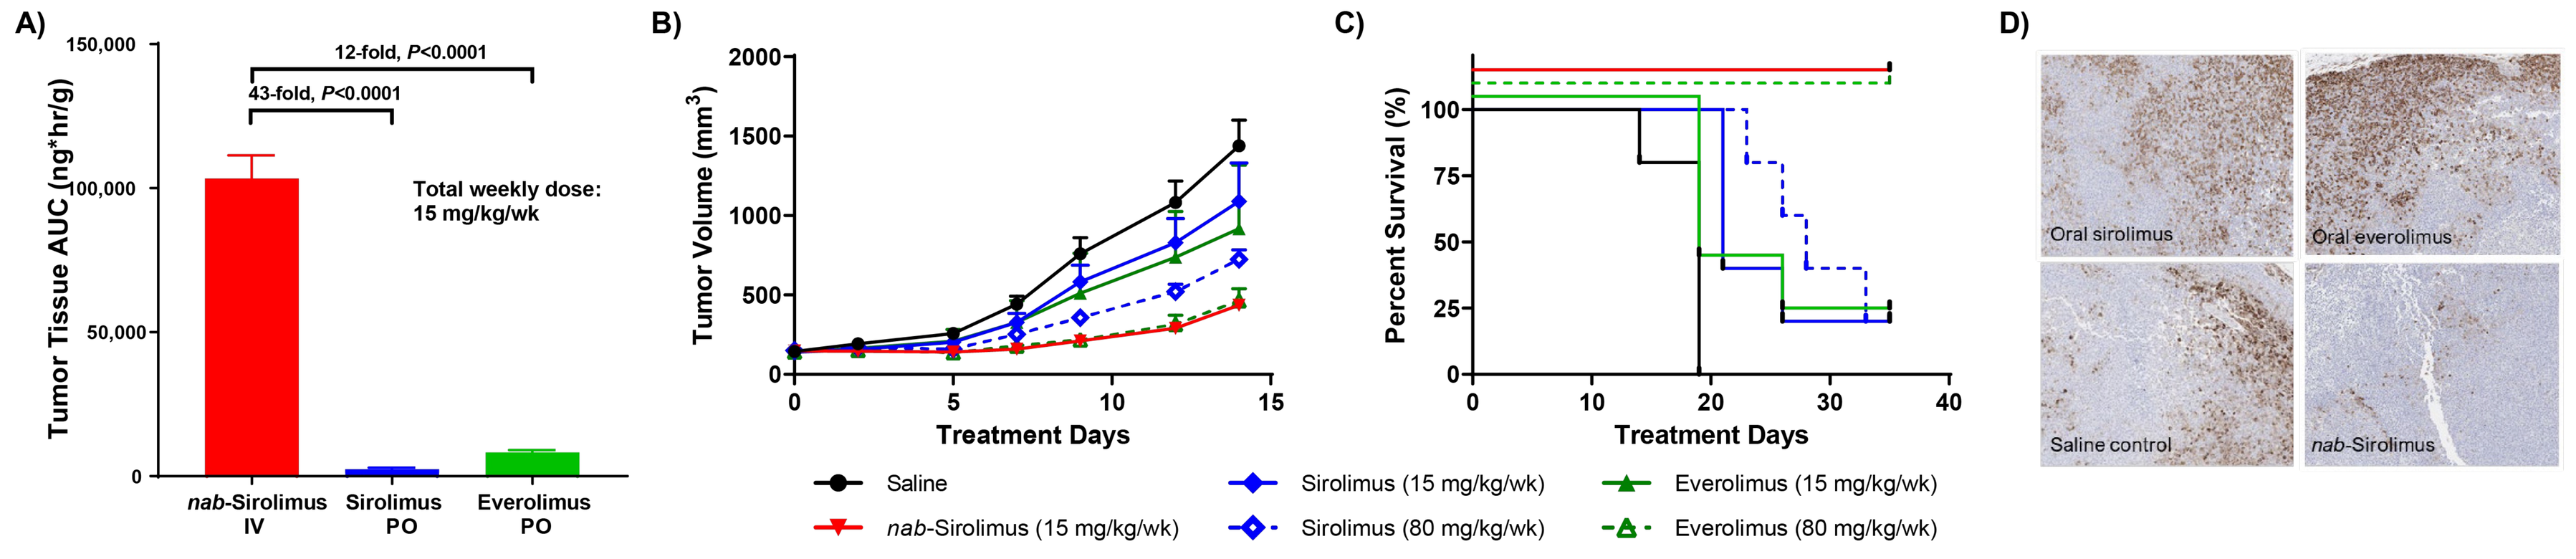

A)
Tumor Tissue AUC (ng*hr/g)
150,000
100,000
50,000
0
12-fold, P<0.0001
43-fold, P<0.0001
Total weekly dose: 15 mg/kg/wk
nab-Sirolimus IV
Sirolimus PO
Everolimus PO
B)
Tumor Volume (mm3)
2000
1500
1000
500
0
0
5
10
15
Treatment Days
C)
Percent Survival (%)
100
75
50
25
0
0
10
20
30
40
Treatment Days
D)
Oral sirolimus
Oral everolimus
Saline control
nab-Sirolimus
Saline
nab-Sirolimus (15 mg/kg/wk)
Sirolimus (15 mg/kg/wk)
Sirolimus (80 mg/kg/wk)
Everolimus (15 mg/kg/wk)
Everolimus (80 mg/kg/wk)

AUC, area under the curve; IV, intravenous, *nab*, nanoparticle albumin-bound; PO, per os (orally); pS6, ribosomal protein S6.

- *nab*-Sirolimus is a novel albumin-bound mTOR inhibitor (mTORi) and is approved in the United States for the treatment of adult patients with locally advanced, unresectable or metastatic malignant perivascular epithelioid cell tumor (PEComa)[5] based on clinical efficacy and safety results from the AMPECT trial (NCT02494570)[6]
- An exploratory biomarker analysis from AMPECT demonstrated that known inactivating alterations in *TSC1* or *TSC2* were associated with response to *nab*-sirolimus in PEComa, suggesting *nab*-sirolimus might be beneficial for patients with other solid tumors harboring *TSC1* or *TSC2* alterations (**Figure 3**)[6,7]
 - Most treatment-emergent adverse events (TEAEs) were grade 1/2 and were manageable for long-term treatment; no grade ≥4 treatment-related TEAEs were observed
 - The overall safety profile was consistent with other mTORis, with no new or unexpected safety signals emerging in the AMPECT trial

Figure 3. Percent Change in Target Tumor Measurements Over Time

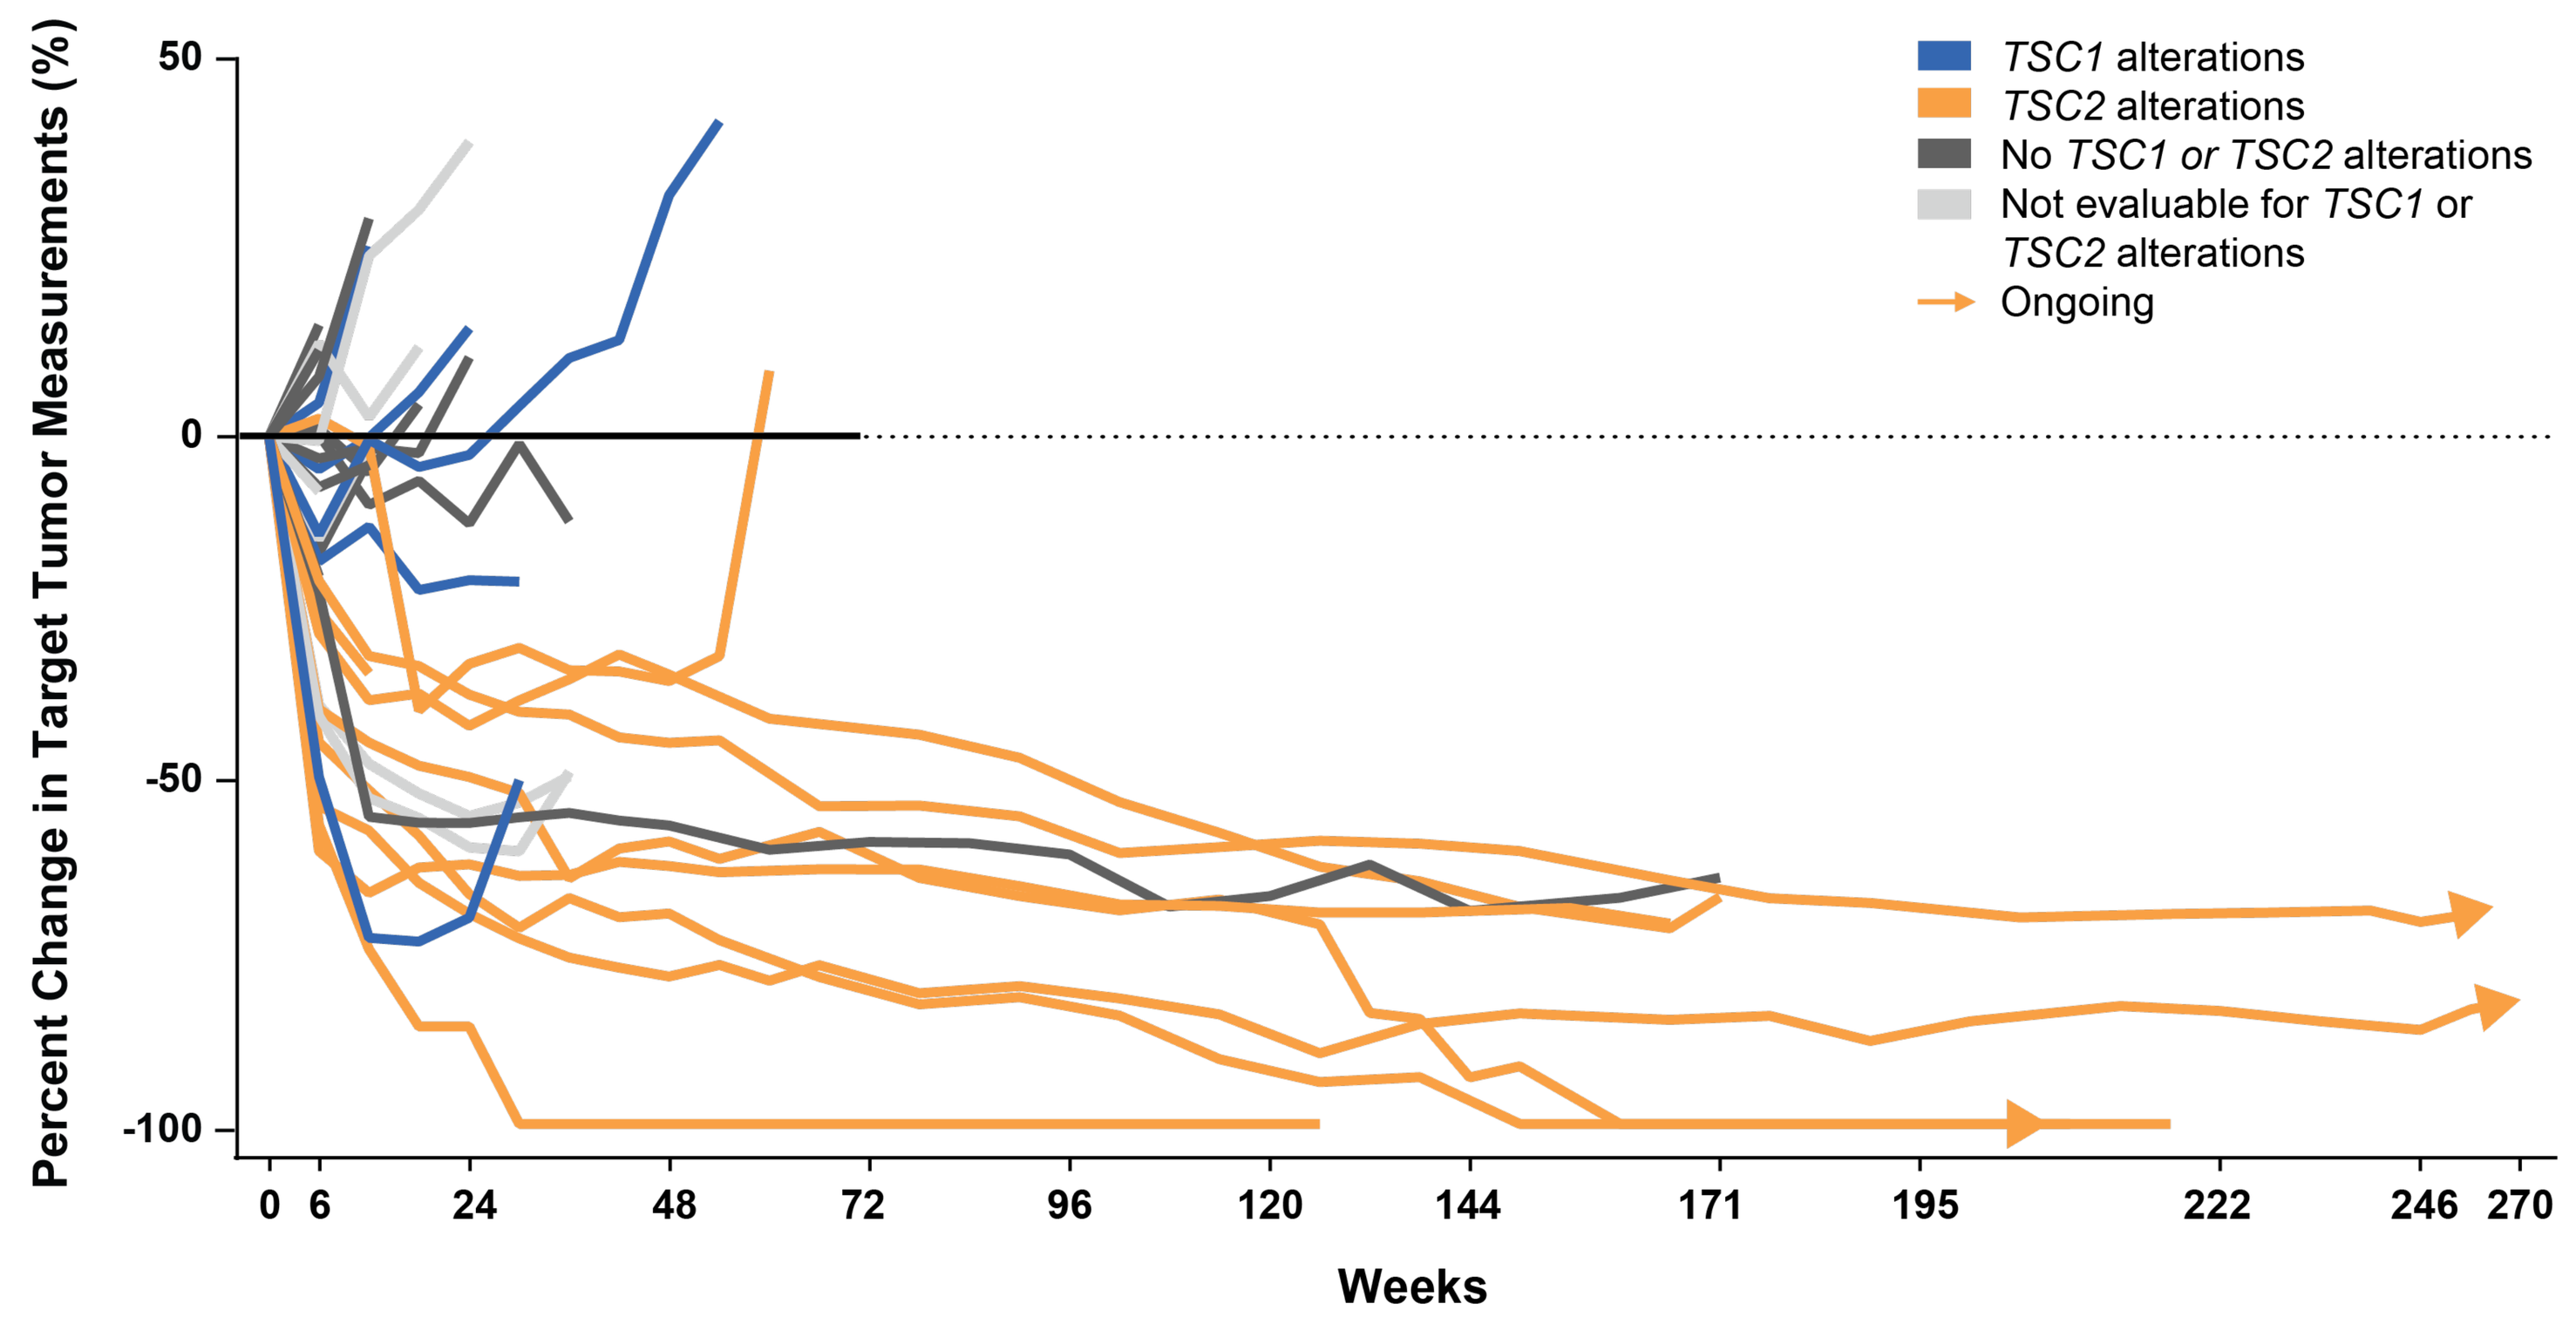

Percent Change in Target Tumor Measurements (%)
50
0
-50
-100
0
6
24
48
72
96
120
144
171
195
222
246
270
Weeks
TSC1 alterations
TSC2 alterations
No TSC1 or TSC2 alterations
Not evaluable for TSC1 or TSC2 alterations
Ongoing

- Patients with various malignancies bearing *TSC1* or *TSC2* alterations treated with *nab*-sirolimus as part of the expanded access program (NCT03817515) showed evidence of response (partial response in 5/7 patients) and manageable toxicities[8]
- The phase 2 PRECISION I trial was initiated to evaluate the potential of mTOR inhibition with *nab*-sirolimus for the treatment of patients with solid tumors harboring *TSC1* or *TSC2* inactivating alterations

STUDY DESIGN

- PRECISION I (NCT05103358) is a prospective, phase 2, open-label, multi-institution basket trial to determine the efficacy and safety profile of *nab*-sirolimus in patients with malignant solid tumors with pathogenic inactivating alterations in *TSC1* (Arm A) or *TSC2* (Arm B) (**Figure 4**)

Figure 4. Study Design

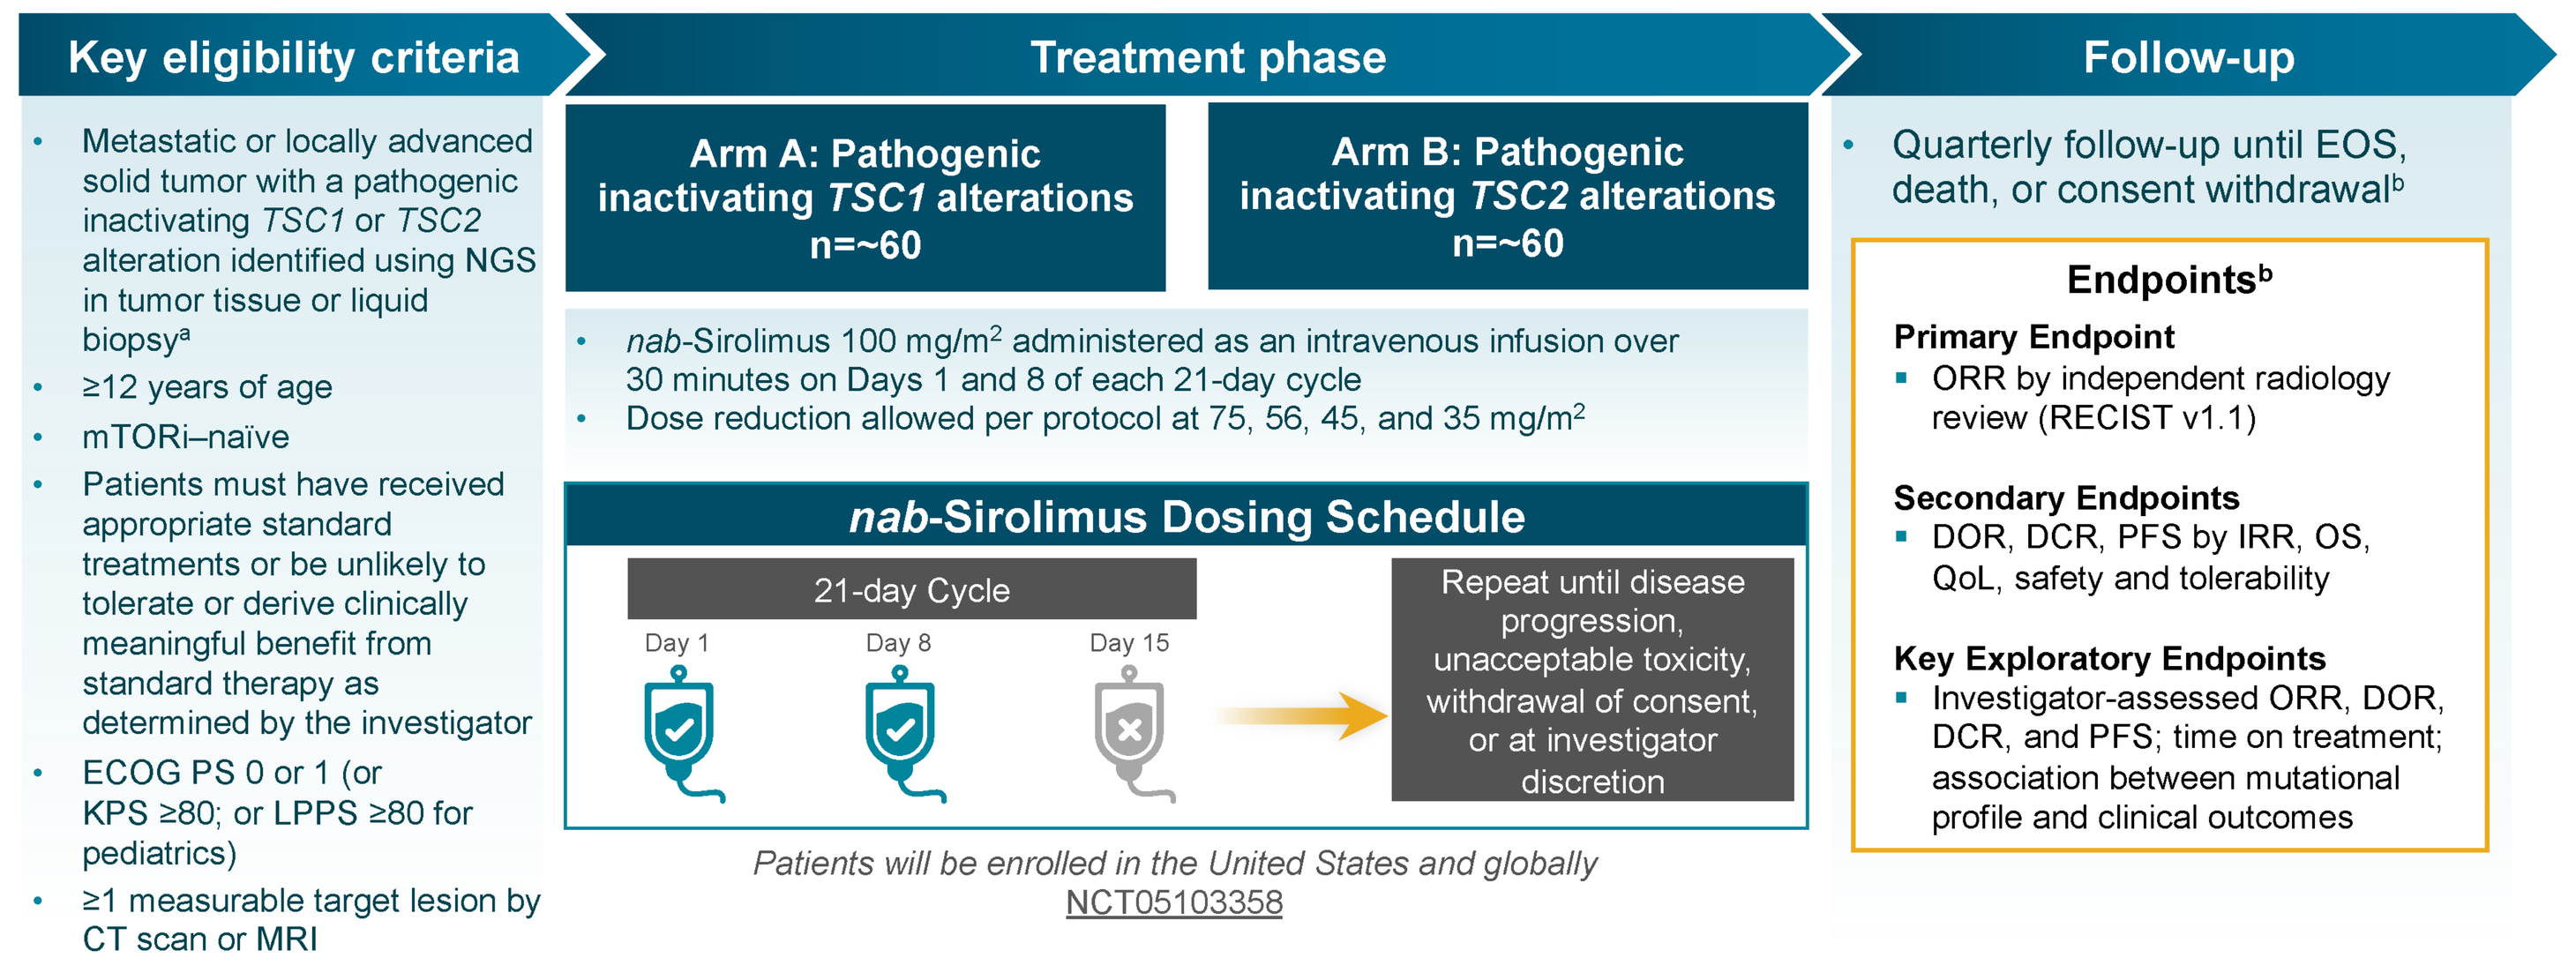

Key eligibility criteria
Metastatic or locally advanced solid tumor with a pathogenic inactivating TSC1 or TSC2 alteration identified using NGS in tumor tissue or liquid biopsy[a]
≥12 years of age
mTORi–naïve
Patients must have received appropriate standard treatments or be unlikely to tolerate or derive clinically meaningful benefit from standard therapy as determined by the investigator
ECOG PS 0 or 1 (or KPS ≥80; or LPPS ≥80 for pediatrics)
≥1 measurable target lesion by CT scan or MRI
Treatment phase
Arm A: Pathogenic inactivating TSC1 alterations n=~60
Arm B: Pathogenic inactivating TSC2 alterations n=~60
nab-Sirolimus 100 mg/m2 administered as an intravenous infusion over 30 minutes on Days 1 and 8 of each 21-day cycle
Dose reduction allowed per protocol at 75, 56, 45, and 35 mg/m2
nab-Sirolimus Dosing Schedule
21-day Cycle
Day 1
Day 8
Day 15
Repeat until disease progression, unacceptable toxicity, withdrawal of consent, or at investigator discretion
Patients will be enrolled in the United States and globally
NCT05103358
Follow-up
Quarterly follow-up until EOS, death, or consent withdrawal[b]
Endpoints[b]
Primary Endpoint
ORR by independent radiology review (RECIST v1.1)
Secondary Endpoints
DOR, DCR, PFS by IRR, OS, QoL, safety and tolerability
Key Exploratory Endpoints
Investigator-assessed ORR, DOR, DCR, and PFS; time on treatment; association between mutational profile and clinical outcomes

[a]Central confirmation of *TSC1* and *TSC2* pathogenic inactivating alterations is via evaluation of NGS reports. Patients will be enrolled only after central confirmation of eligibility. *TSC1* and *TSC2* alterations should be identified using NGS in tumor tissue or liquid biopsy and must be determined by analytically validated NGS tests performed in CLIA-certified laboratories.
[b]Follow-up is for survival and initiation of anticancer therapy. Follow-up is initiated after the EOS visit. CLIA, Clinical Laboratory Improvement Amendments; DCR, disease control rate; DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; EOS, end of study; IRR, interrater reliability; KPS, Karnofsky Performance Scale; LPPS, Lansky Play-Performance Scale; mTORi, mechanistic target of rapamycin inhibitor; *nab*, nanoparticle albumin-bound; NGS, next-generation sequencing; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PS, performance status; QOL, quality of life; RECIST, Response Evaluation Criteria for Solid Tumors; *TSC1*/*TSC2*, tuberous sclerosis complex 1 or 2. Reproduced from Schulte et al.[7]

Table 2. Key Inclusion and Exclusion Criteria

Key Inclusion Criteria	Key Exclusion Criteria
≥12 years of age	Prior treatment with mTOR inhibitor
Metastatic or locally advanced malignant solid tumor with a pathogenic inactivating *TSC1* or *TSC2* alteration, where surgical resection is not an option, or likely to result in severe morbidity	Recent infection requiring systemic anti-infective treatment
Received appropriate standard treatments or be unlikely to tolerate or derive clinically meaningful benefit from standard therapy as determined by the investigator	Primary brain tumor or PEComa
ECOG performance status of 0 or 1, KPS ≥80, or Lansky play-performance scale for pediatric patients ≥80	Severe and/or uncontrolled medical or psychiatric conditions
≥1 measurable target lesion by CT scan or MRI	For patients on strong inhibitors, inducers, and known CYP3A4 substrates, discontinuation is required ≥5 half-lives prior to receiving the first dose of *nab*-sirolimus

The full list of inclusion and exclusion criteria is available at https://www.clinicaltrials.gov/ct2/show/NCT05103358.
CT, computed tomography; CYP3A4, cytochrome P450 3A4; ECOG, Eastern Cooperative Oncology Group; KPS, Karnofsky performance status; mTOR, mechanistic target of rapamycin; MRI, magnetic resonance imaging; *nab*, nanoparticle albumin-bound; PEComa, perivascular epithelioid tumor; *TSC1/TSC2*, tuberous sclerosis complex subunit 1 or 2.

- Partnerships with leading next-generation sequencing companies (Foundation Medicine, Tempus, and Caris) and US Oncology will facilitate identification of patients with qualifying inactivating *TSC1* or *TSC2* alterations and expand access to the study through just-in-time trial locations and accelerated site activation (**Figure 5**)

Figure 5. Expanding PRECISION I Access and Enrollment Through Strategic Partnerships

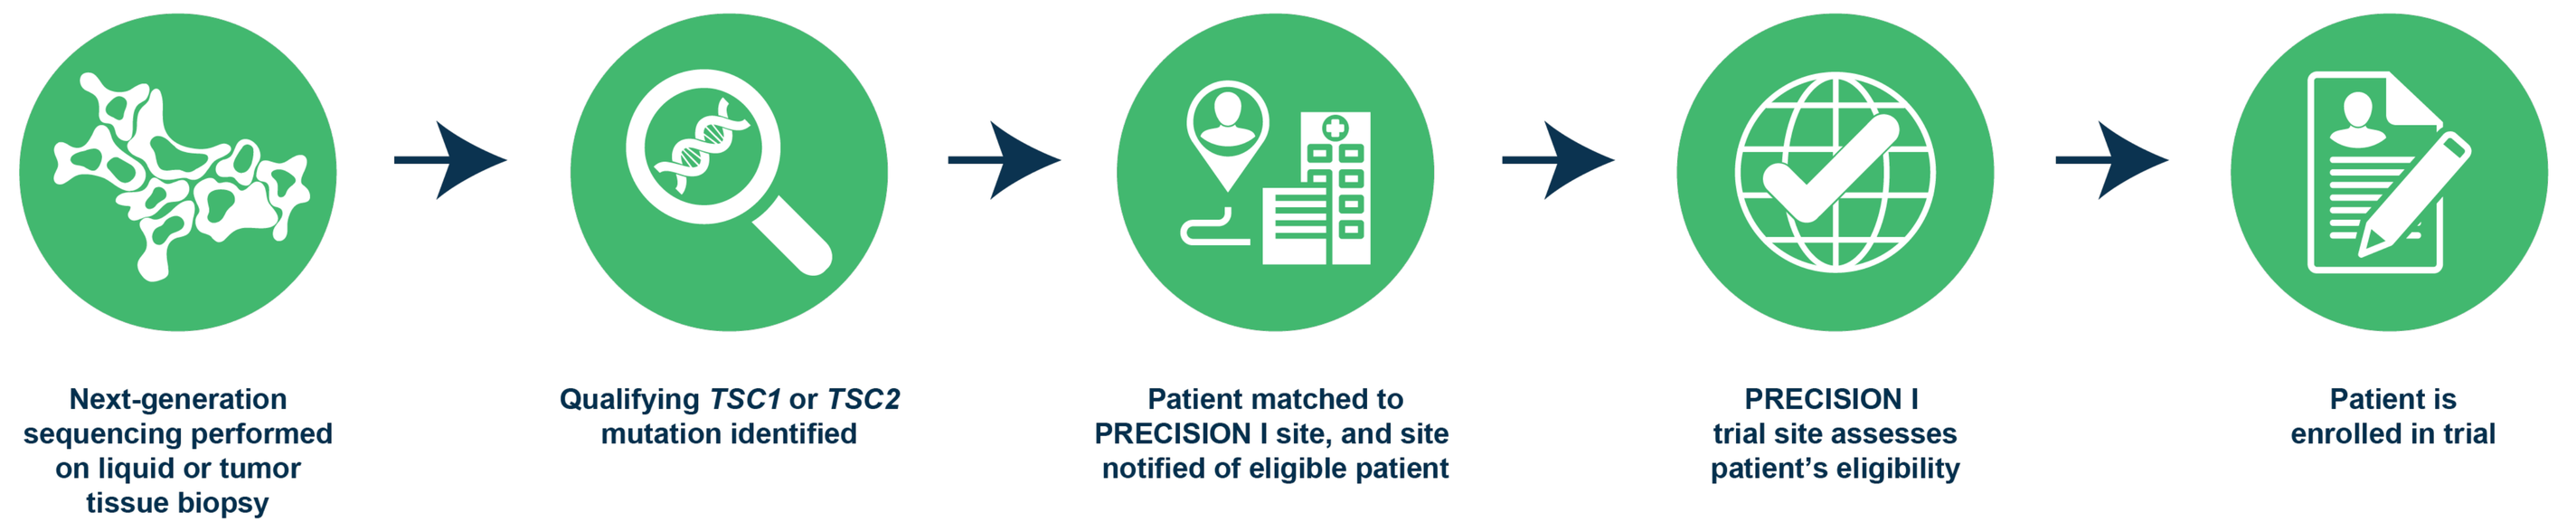

Next-generation sequencing performed on liquid or tumor tissue biopsy
Qualifying TSC1 or TSC2 mutation identified
Patient matched to PRECISION I site, and site notified of eligible patient
PRECISION I trial site assesses patient's eligibility
Patient is enrolled in trial

SUMMARY

- Available data from the AMPECT exploratory analysis and an expanded access program suggest acceptable efficacy and safety of *nab*-sirolimus, an mTORi with enhanced antitumor activity, in patients with solid tumors harboring inactivating alterations in *TSC1* and/or *TSC2*
- PRECISION I is a registrational basket trial for patients with solid tumors driven by *TSC1* or *TSC2* alterations; enrollment began in March 2022
- This trial is designed to evaluate the efficacy, safety, and tolerability of *nab*-sirolimus in a patient population with advanced malignancies and limited therapeutic options
- Collaboration with leading next-generation sequencing vendors will expedite the identification of patients with qualifying *TSC1* or *TSC2* alterations; study access will be facilitated through a "just-in-time" approach to trial location activation

REFERENCES

1. He Y, et al. *Signal Transduct Target Ther*. 2021;6(1):425. **2.** Saxton RA, et al. *Cell*. 2017;169(2):361-371. **3.** Hou S, et al. AACR-NCI-EORTC, October 7-10, 2021 [virtual]. Poster P138. **4.** Pavavra F, et al. *Oxid Med Cell*. 2017;9820181. **5.** FYARRO (sirolimus albumin-bound particles for injectable suspension). Package insert. Aadi Bioscience, Inc.: Pacific Palisades, CA; 2021. **6.** Wagner AJ, et al. *J Clin Oncol*. 2021;39(33):3660-3670. **7.** Schulte B, et al. CTOS, November 16-19, 2022; Vancouver, BC. Poster 313. **8.** Dickson MA, et al. ASCO, June 4-8, 2021, Virtual.

DISCLOSURES: JRA: Research funding: Bayer, Novartis; personal fees: Eli Lilly, Peptomyc, and Servier. **MJD:** Consulting fees: Bayer, Eli Lilly, On Cusp Therapeutics, and TD2. **NF:** Consulting and speakers' fees: Bayer. **AS:** Honoraria: ARIAD Pharmaceuticals, Clovis Oncology, Novartis, Roche; consulting fees: ARIAD Pharmaceuticals, Clovis Oncology, CytomX Therapeutics, Genentech, Roche; research funding: AbbVie, Astellas Pharma, AstraZeneca, BeiGene, Boehringer Ingelheim, Clovis Oncology, MedImmune, Merck, Merrimack, Newlink Genetics, Novartis, Roche; travel fees: ARIAD Pharmaceuticals, Roche. **DJK:** Research funding: Aadi Bioscience, Genentech, Revolution Medicines; consulting fees: Aadi Bioscience, Bridgebio, Genentech, Guidepoint. **MH:** Consulting fees: Guardant Health; speakers' fees: AMAG Pharmaceuticals, Boehringer Ingelheim, Bristol Myers Squibb, Heron, Incyte, Pfizer; stock ownership: Celgene. **EMG:** Stock and other ownership interests: Counterpoint Biomedica, Delta NextGene; research funding: Bristol Myers Squibb. **KNG:** Consulting fees: Daiichi Sankyo, Deciphera, Foundation Medicine. **LDC:** Research funding: Aadi Bioscience, Advenchen Laboratories, CBA Research, Exelixis, Iterion Therapeutics, Lilly, Merck, Monopar, Philogen, Zentalis; consulting fee: Aadi Bioscience. **LD, ANS, WHN, and LMI:** Employment and stock ownership: Aadi Bioscience, Inc. **BS, DD, MAM, EL, DGC, and GI:** no conflicts of interest.

ACKNOWLEDGMENTS: Medical writing and editorial assistance were provided by Cynthia D. Gioiello, PharmD, and Stephen Bublitz, ELS, of MedVal Scientific Information Services, LLC, and were funded by Aadi Bioscience, Inc.

Correspondence: MedInfo@AadiBio.com

Presented at the American Association for Cancer Research Annual Meeting, April 14-19, 2023, Orlando, FL