Inactivating Alterations in *TSC1* and *TSC2*, Co-Mutations, and Genomic Instability in Advanced Cancers: Analysis of a Real-World Patient Population Using the Foundation Medicine Genomic Database

C019

David J. Kwiatkowski, MD, PhD[1]; Norma A. Palma, PhD[2]; Willis H. Navarro, MD[2]; Gopa Iyer, MD[3]

[1]Brigham and Women's Hospital, Boston, MA; [2]Aadi Bioscience, Pacific Palisades, CA; [3]Memorial Sloan Kettering Cancer Center, New York, NY

Objective

- To characterize *TSC1* and *TSC2* alterations across a large real-world patient population with advanced cancer using data from tumor tissue biopsies

KEY FINDINGS

In a large real-world database of patients with advanced cancer, known/likely inactivating alterations in *TSC1* or *TSC2* occurred across common tumor types at rates as high as 9.6% (urinary bladder tumors), and in 1.9% of patients overall

Most known/likely inactivating alterations in *TSC1* and *TSC2* occurred in the context of low tumor mutational burden and/or microsatellite stable status, suggesting that these are unlikely to be passenger mutations and instead may potentially serve as driver mutations

Low frequencies of co-occurring targetable oncogenic drivers also suggest that therapeutic targeting of *TSC1* and *TSC2* inactivating alterations may be a potentially effective treatment approach in this patient population. This hypothesis is being tested in the currently enrolling PRECISION 1 study (NCT05103358)

Presented at the AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics; Boston, MA; October 11-15, 2023

Correspondence: MedInfo@AadiBio.com

BACKGROUND

- The tumor suppressor genes *TSC1* and *TSC2* are critical negative regulators of mTOR activity whose inactivation can lead to tumor cell growth[1–4] (**Figure 1**)

Figure 1. Dysregulation of the PI3K-AKT-mTOR pathway via inactivation of the TSC1-TSC2 complex can lead to overactivation and tumorigenesis

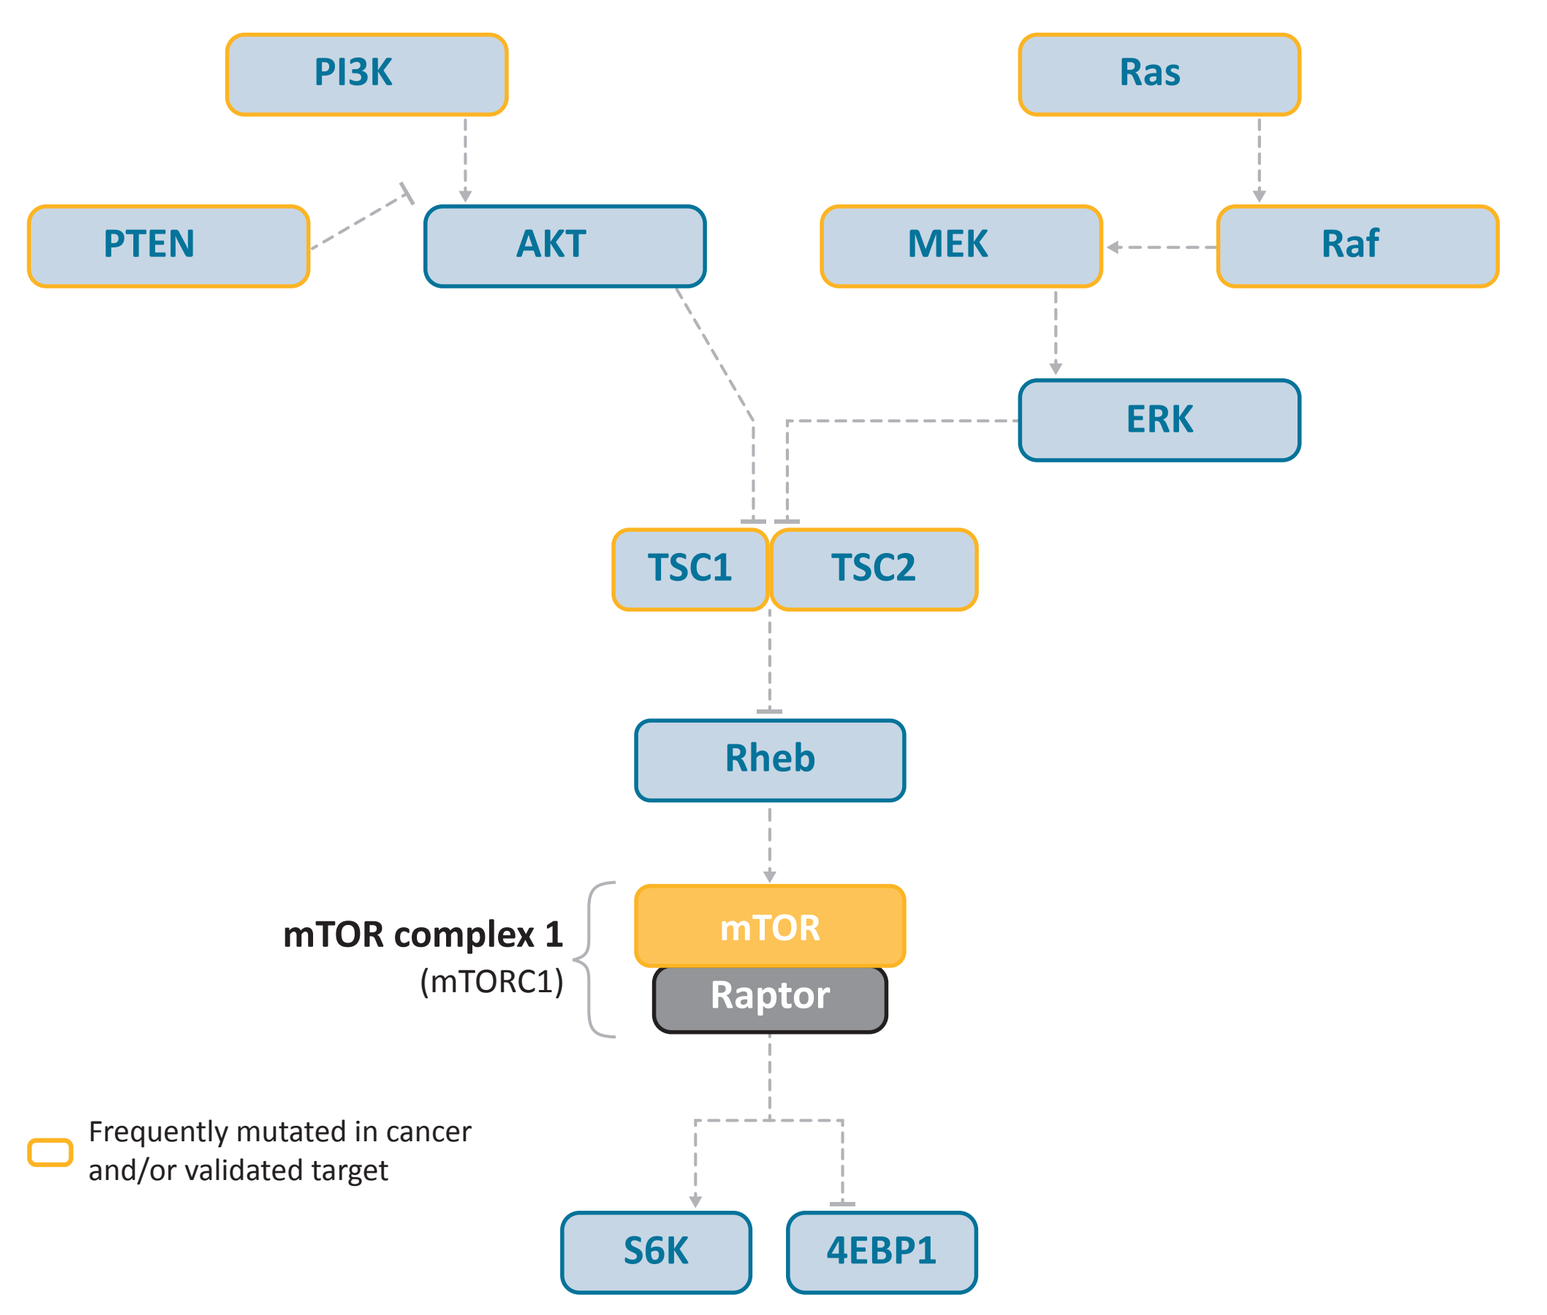
PI3K
Ras
PTEN
AKT
MEK
Raf
ERK
TSC1
TSC2
Rheb
mTOR complex 1
(mTORC1)
mTOR
Raptor
Frequently mutated in cancer and/or validated target
S6K
4EBP1

4EBP, eukaryotic translation initiation factor 4E (eIF4E) -binding protein; **AKT**, protein kinase B; **ERK**, extracellular signal-regulated kinase; **MEK**, mitogen-activated protein kinase kinase; **mTOR**, mammalian/mechanistic target of rapamycin; **mTORC1**, mTOR complex 1; **PI3K**, phosphatidylinositol 3-kinase; **PTEN**, phosphatase and tensin homolog; **Raf**, rapidly accelerated fibrosarcoma; **Raptor**, regulatory-associated protein of mTOR; **Ras**, rat sarcoma virus homolog; **Rheb**, Ras homolog enriched in brain; **S6K**, ribosomal S6 kinase; **TSC1/TSC2**, tuberous sclerosis complex subunit 1 or 2.

- *nab*-Sirolimus, a nanoparticle albumin-bound, IV administered mTOR inhibitor, is approved in the United States for treatment of adult patients with advanced malignant PEComa[5] based on the safety and efficacy demonstrated in the AMPECT trial (NCT02494570)[6]
- Of patients with known inactivating alterations in *TSC1* or *TSC2*, 9 of 14 (64.3%) with malignant PEComa in AMPECT[6] and 5 of 7 (71.4%) in the pan-tumor Expanded Access Program (NCT03817515)[7] had confirmed responses to the mTOR inhibitor *nab*-sirolimus and manageable toxicities
- PRECISION 1 (NCT05103358), a currently enrolling tumor-agnostic study, is assessing the clinical benefit of *nab*-sirolimus in patients with malignant solid tumors harboring inactivating alterations in *TSC1* and/or *TSC2*
- *TSC1* and/or *TSC2* mutational data from a real-world genomic database were analyzed to characterize and enumerate the frequency of these alterations across tumor types

METHODS

- Next-generation sequencing (NGS) data (from 2010 to 2022) from the Foundation Medicine genomic database of patients primarily with advanced cancer, exclusive of hematological malignancies, were analyzed using the FoundationInsights™ web-based platform
- *TSC1* and *TSC2* variants were categorized according to likelihood of inactivation—"known", "likely", "ambiguous", or "unknown"—based on Foundation Medicine's assessments for identification. Assessment included listing in the Catalogue of Somatic Mutations in Cancer, existing relevant literature to support categorization, and internal evidence to determine the reported functional status
 - Known inactivating variants comprised:
 - Recurrent somatic short variants
 - Copy number alterations involving genes that are recurrently amplified/deleted
 - Rearrangements involving known fusion partners, or other functional events
 - Likely variant calls included short variants or rearrangements that disrupt tumor suppressor genes or are located in known hotspot regions, or other functional events
 - Ambiguous variant calls included non-focal amplifications
 - Unknown variants were classified as those with unknown somatic or functional status, and insufficient data in the literature
- Genomic *TSC1* and *TSC2* variants were then categorized as short variants (substitutions, insertions, and deletions), copy number alterations, or genomic rearrangements
- Patient samples with known/likely inactivating alterations in *TSC1* and *TSC2* were further characterized for co-mutations, microsatellite instability status, and tumor mutational burden (TMB)

RESULTS

- As of March 29, 2022, a total of 438,974 patients with advanced cancer had NGS data for tumor tissue available in the Foundation Medicine database and accessible via the FoundationInsights™ web-based platform (**Figure 2**)
- Of these patients, 18,719 (4.3%) had *TSC1* or *TSC2* variants of unknown significance (VUS), primarily missense (~89%), that were not known to be inactivating
- In all, 8464 (1.9%) patients had known/likely inactivating alterations in *TSC1* or *TSC2*, and 65 of these 8464 (0.8%) patients had co-occurring *TSC1* and *TSC2* inactivating alterations
- Of the known/likely inactivating alterations, 52.1% were *TSC1* and 48.6% were *TSC2*; alterations included short variants (85.0%), rearrangements (10.8%), and copy number deletions (17.9%)

Figure 2. Patients with *TSC1* or *TSC2* alterations in the Foundation Medicine genomic database

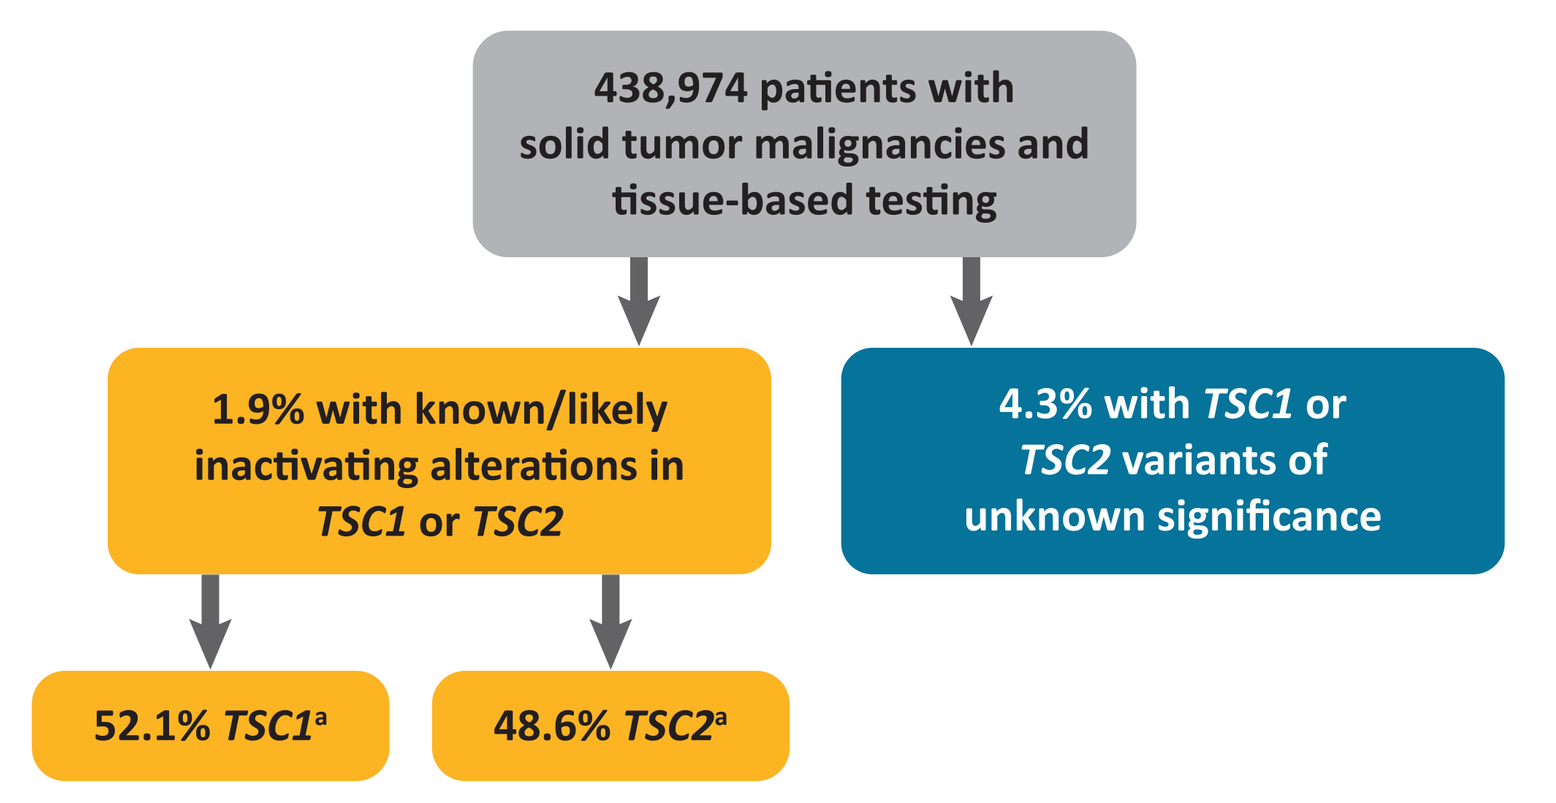
438,974 patients with solid tumor malignancies and tissue-based testing
1.9% with known/likely inactivating alterations in TSC1 or TSC2
4.3% with TSC1 or TSC2 variants of unknown significance
52.1% TSC1a
48.6% TSC2a

[a]Percentages add up to more than 100% due to 65 (0.8%) patients harboring more than one *TSC1* or *TSC2* mutation type and/or mutations in both *TSC1* and *TSC2*.

- Of the *TSC1* alterations identified (n=11,757), 4569 (38.9%) were known/likely inactivating alterations, and 7188 (61.1%) were VUS (**Figure 3A**)
 - Of the known/likely inactivating alterations in *TSC1*, 105 (2.3%) were missense short variants, 3690 (80.8%) were other types of short variants (e.g., insertion, deletion, nonsense and splice variants), 346 (7.6%) were rearrangements, and 428 (9.4%) were copy number deletions
- Of the *TSC2* alterations identified (n=17,757), 4340 (24.4%) were known/likely inactivating alterations, and 13,417 (75.6%) were VUS (**Figure 3B**)
 - Of the known/likely inactivating alterations in *TSC2*, 499 (11.5%) were missense short variants, 2775 (63.9%) were other types of short variants, 613 (14.1%) were rearrangements, and 453 (10.4%) were copy number deletions

Figure 3. Distribution of the types of *TSC1* (A) and *TSC2* (B) alterations

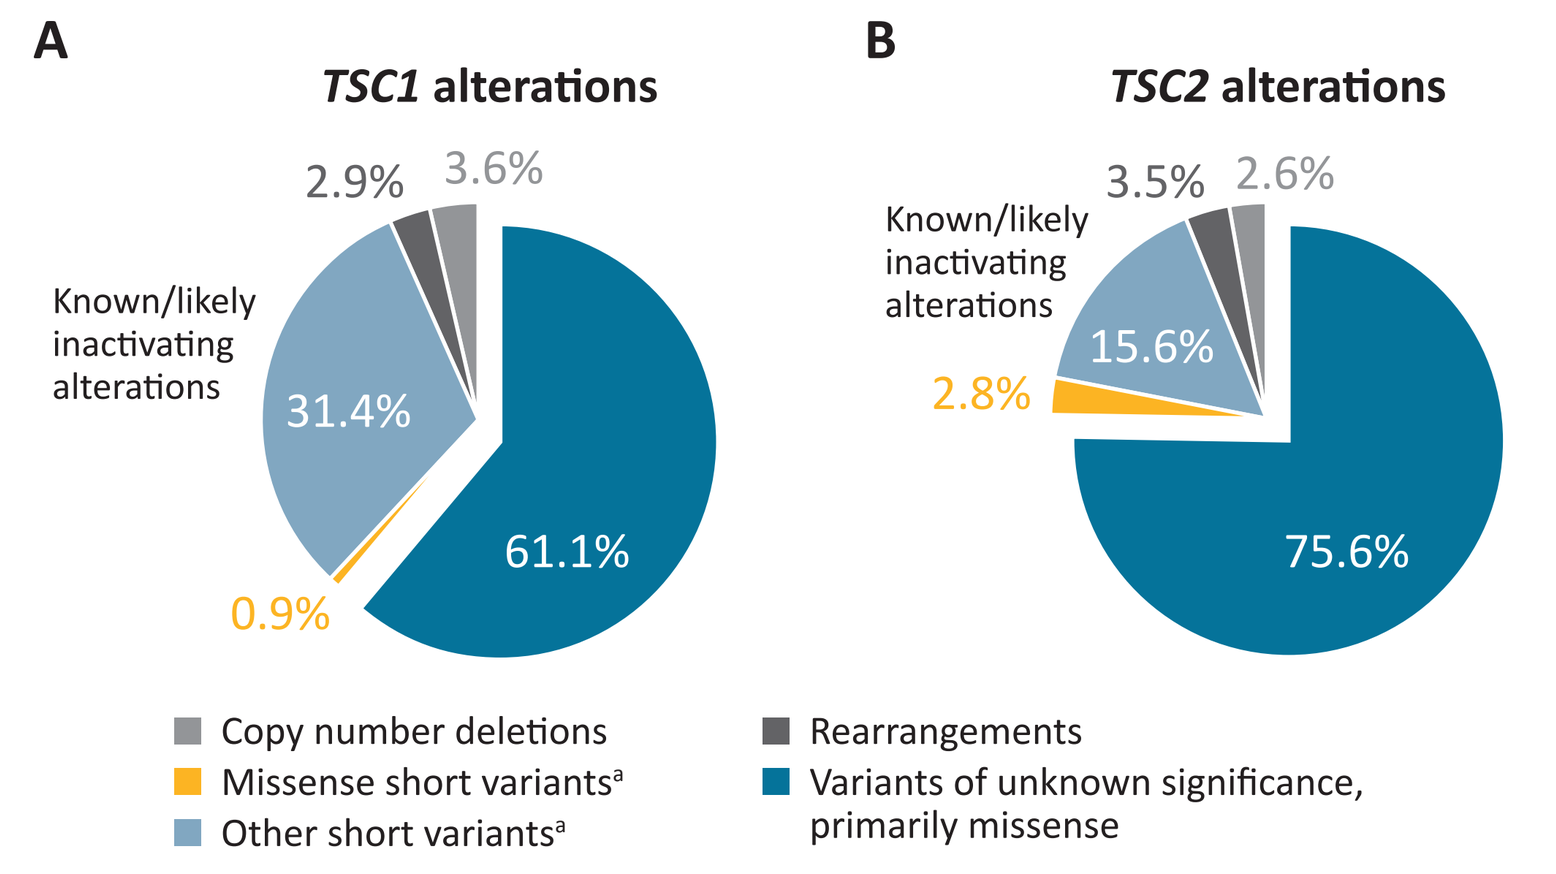
A
TSC1 alterations
2.9%
3.6%
Known/likely inactivating alterations
31.4%
61.1%
0.9%
B
TSC2 alterations
3.5%
2.6%
Known/likely inactivating alterations
15.6%
2.8%
75.6%
Copy number deletions
Missense short variantsa
Other short variantsa
Rearrangements
Variants of unknown significance, primarily missense

Note: Percentages do not add up to 100 due to rounding.
[a]Insertion, deletion, nonsense, and splice variants.

Figure 4. Inactivating alterations in *TSC1* and *TSC2* genes occurred in multiple solid tumor types

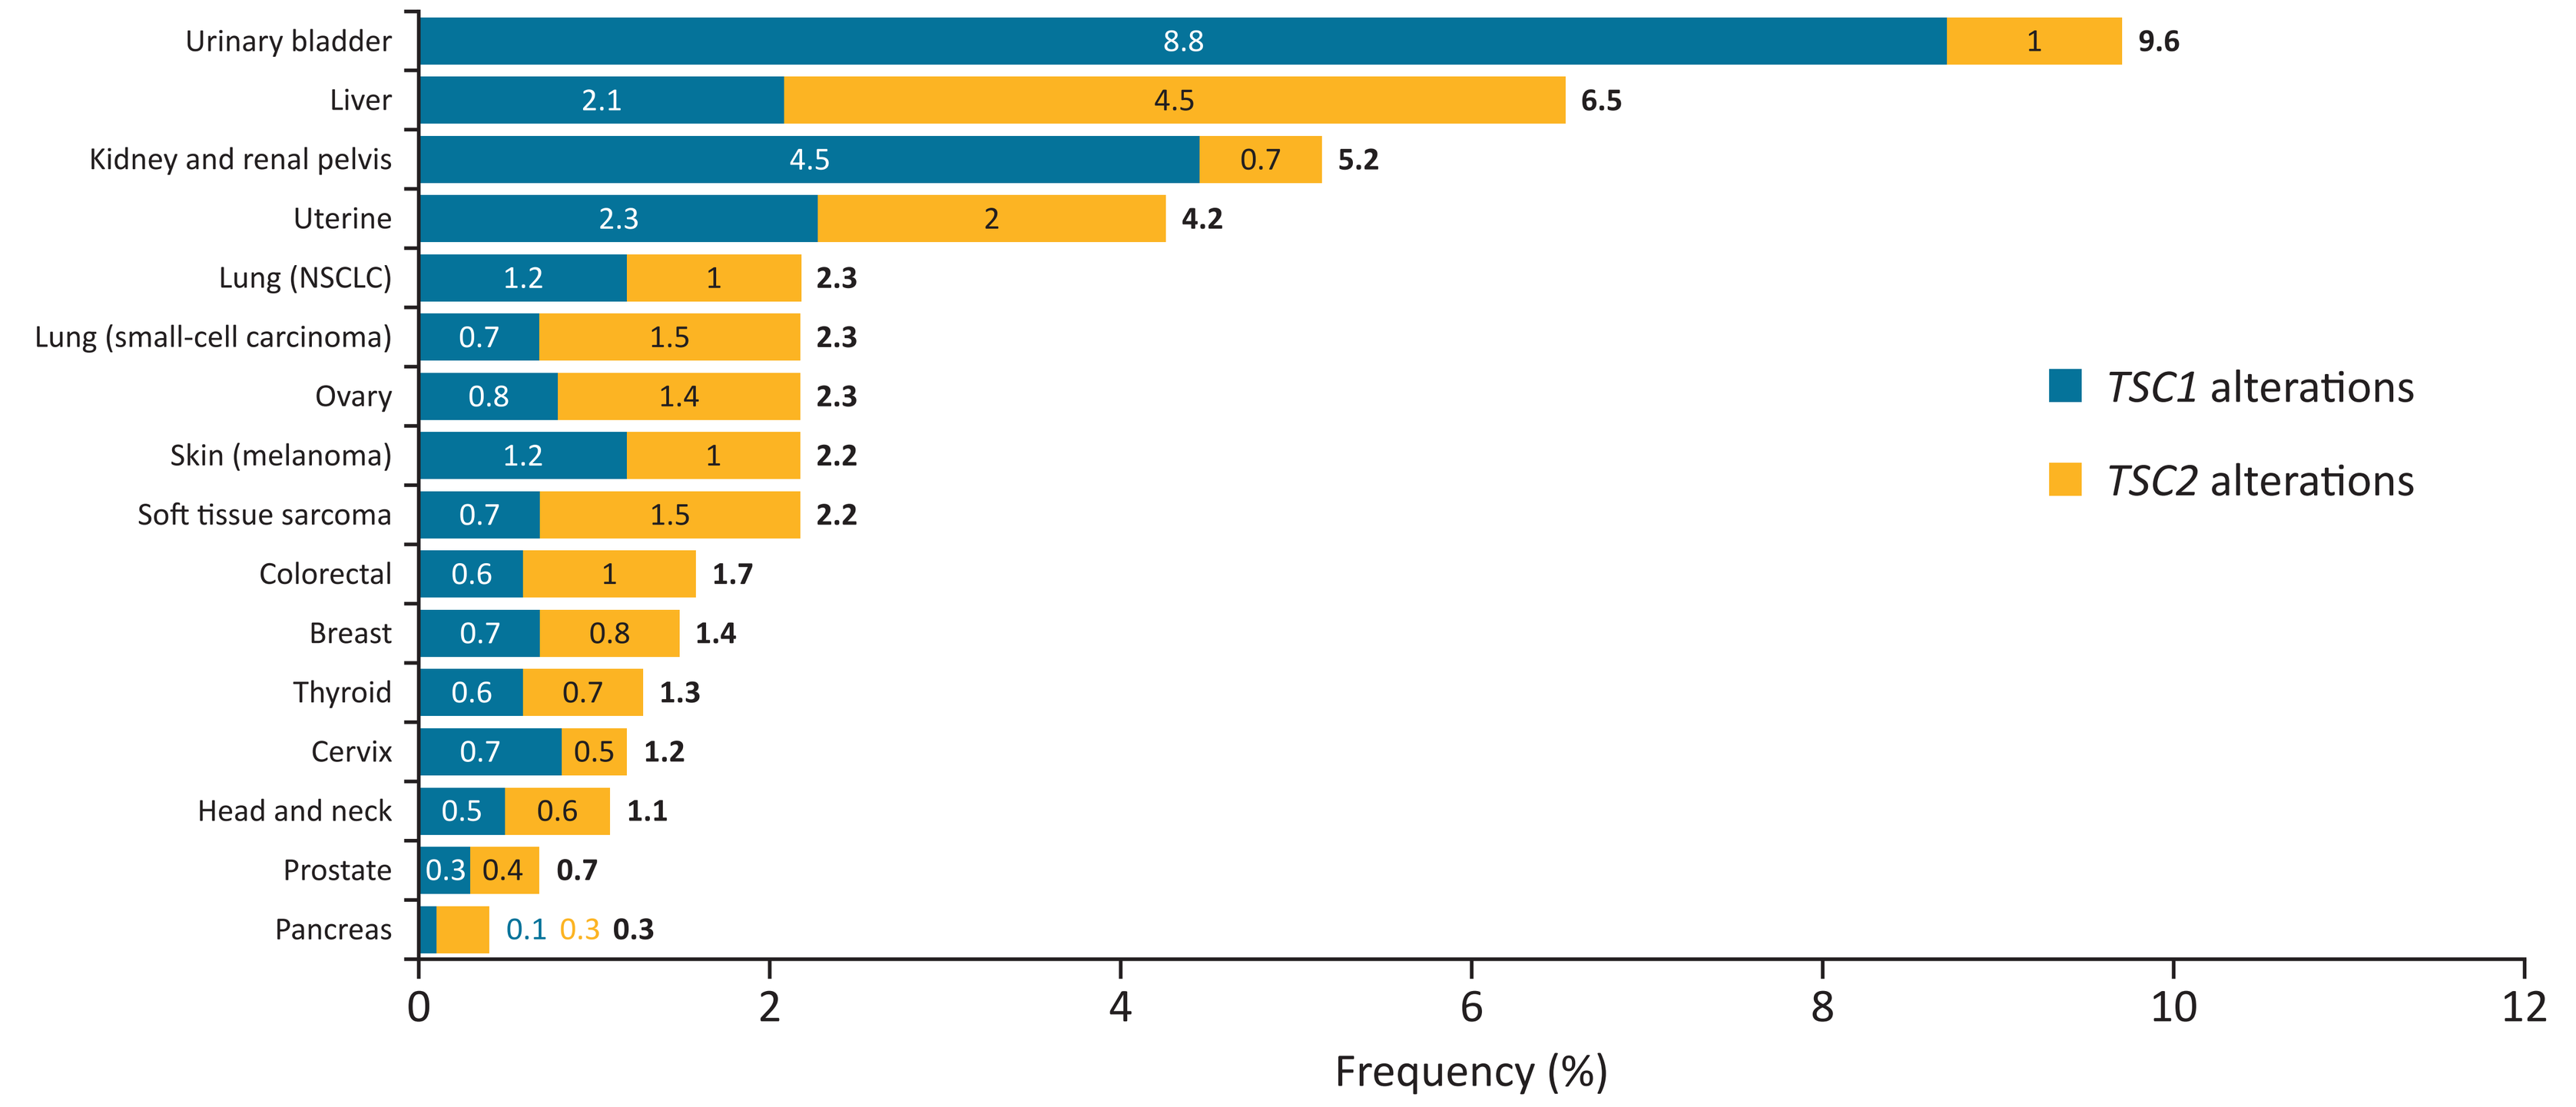
Urinary bladder 8.8 1 9.6
Liver 2.1 4.5 6.5
Kidney and renal pelvis 4.5 0.7 5.2
Uterine 2.3 2 4.2
Lung (NSCLC) 1.2 1 2.3
Lung (small-cell carcinoma) 0.7 1.5 2.3
Ovary 0.8 1.4 2.3
Skin (melanoma) 1.2 1 2.2
Soft tissue sarcoma 0.7 1.5 2.2
Colorectal 0.6 1 1.7
Breast 0.7 0.8 1.4
Thyroid 0.6 0.7 1.3
Cervix 0.7 0.5 1.2
Head and neck 0.5 0.6 1.1
Prostate 0.3 0.4 0.7
Pancreas 0.1 0.3 0.3
0
2
4
6
8
10
12
Frequency (%)
TSC1 alterations
TSC2 alterations

Note: Total percentages may not equal the combination of *TSC1* and *TSC2* due to some patients harboring more than one *TSC1* or *TSC2* mutation type and/or mutations in both *TSC1* and *TSC2*.
NSCLC, non-small cell lung cancer.

Figure 5. Other commonly occurring mutations in (A) patients with a *TSC1* or *TSC2* inactivating alteration and (B) the total patient population in the Foundation Medicine genomic database

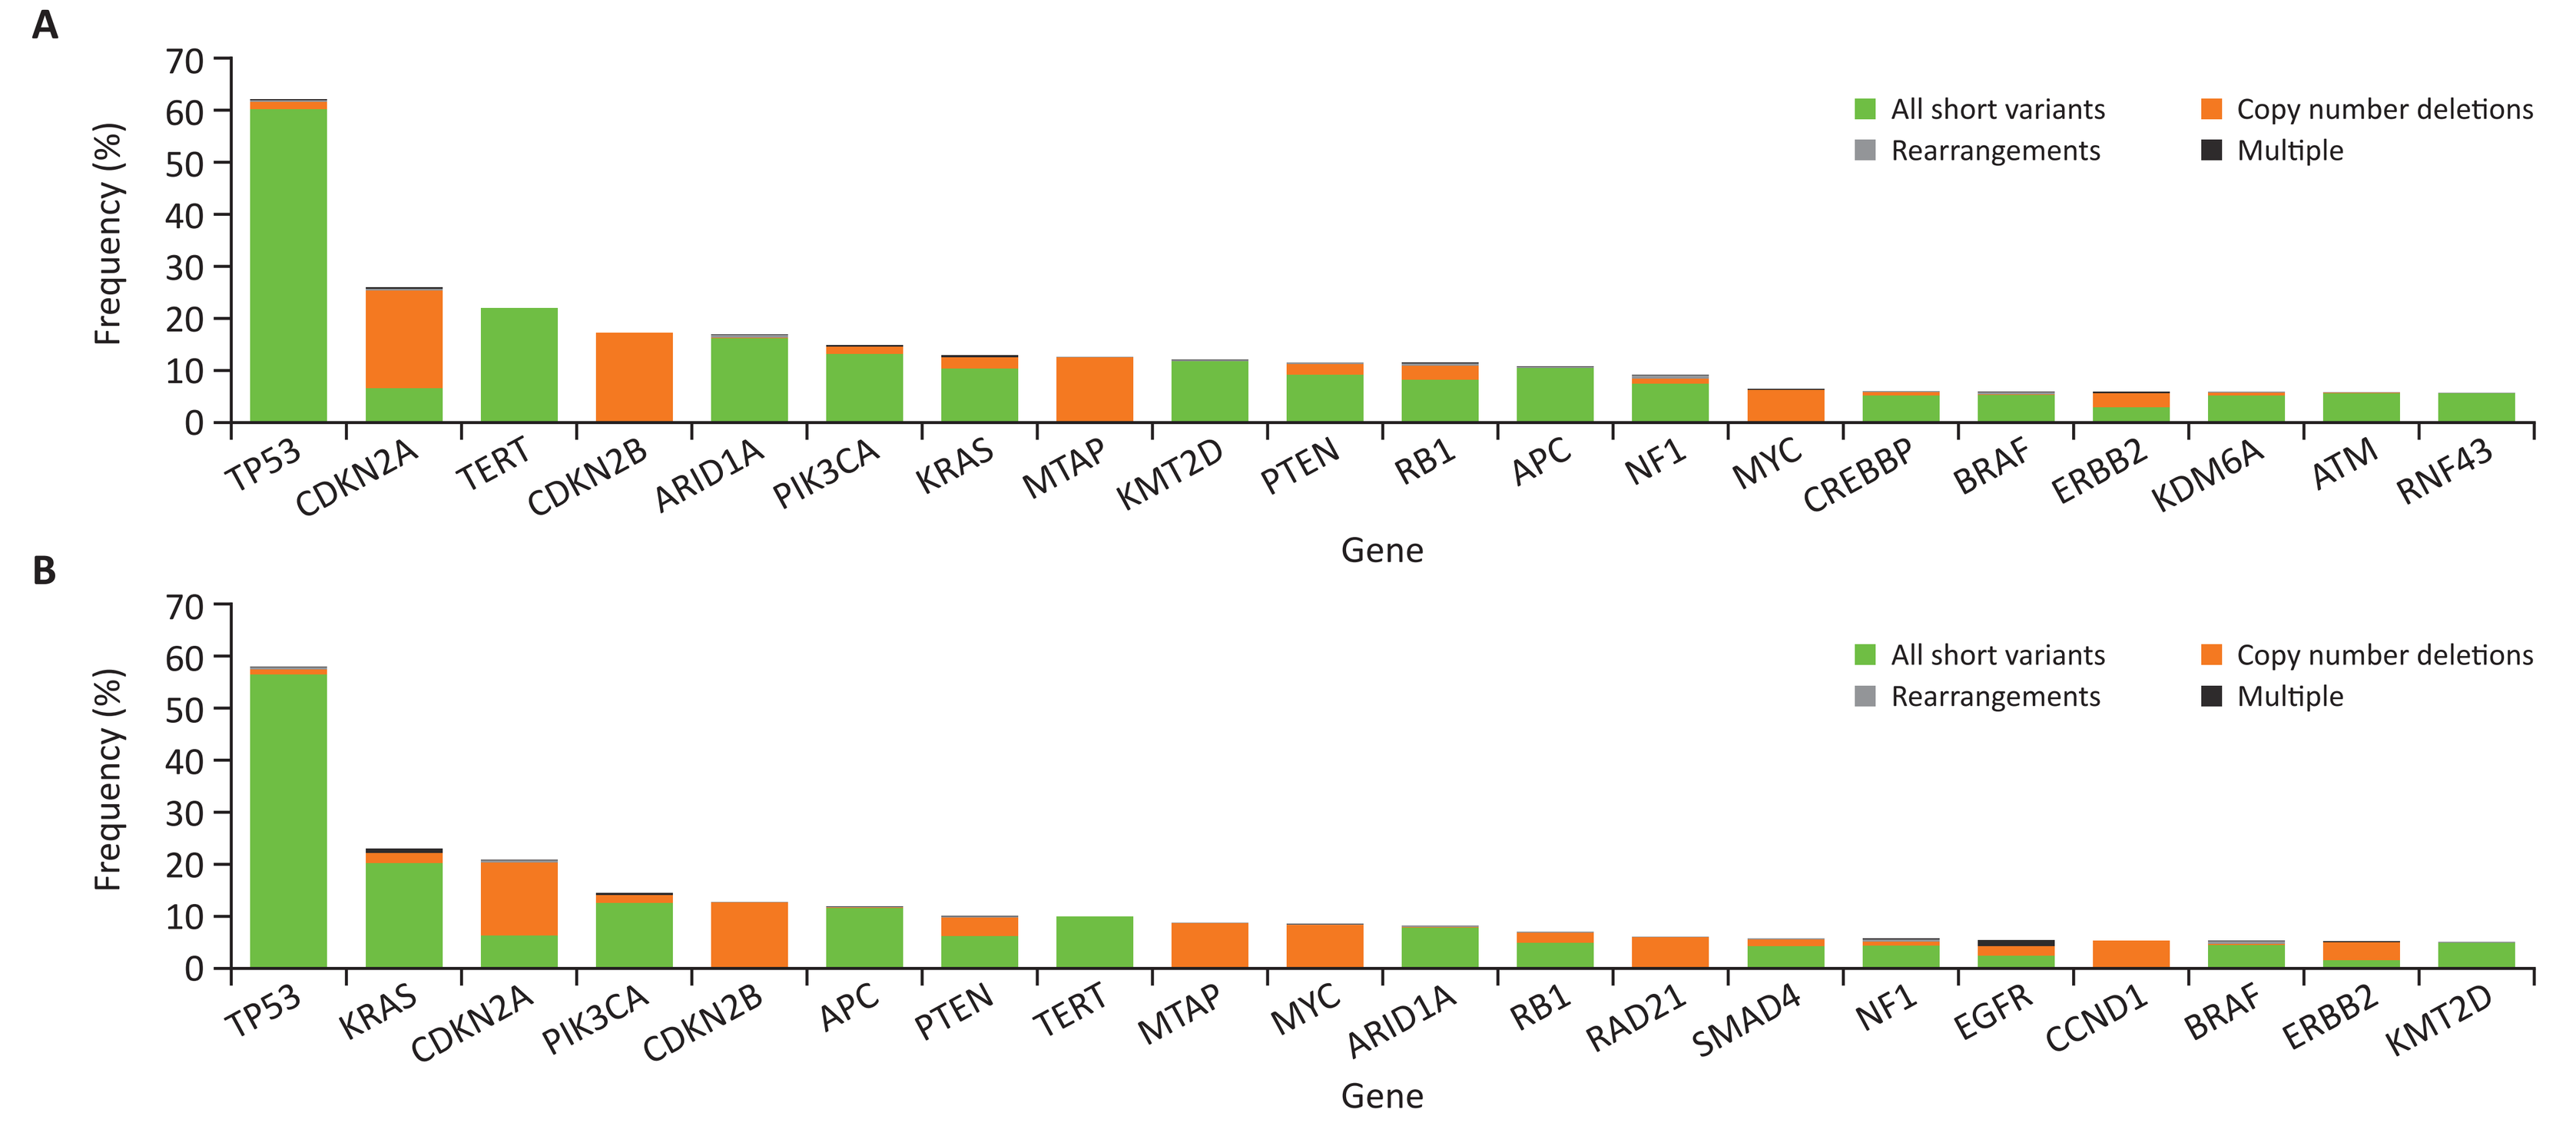
A
Frequency (%)
0
10
20
30
40
50
60
70
TP53
CDKN2A
TERT
CDKN2B
ARID1A
PIK3CA
KRAS
MTAP
KMT2D
PTEN
RB1
APC
NF1
MYC
CREBBP
BRAF
ERBB2
KDM6A
ATM
RNF43
Gene
All short variants
Rearrangements
Copy number deletions
Multiple
B
Frequency (%)
TP53
KRAS
CDKN2A
PIK3CA
CDKN2B
APC
PTEN
TERT
MTAP
MYC
ARID1A
RB1
RAD21
SMAD4
NF1
EGFR
CCND1
BRAF
ERBB2
KMT2D
Gene
All short variants
Rearrangements
Copy number deletions
Multiple

- *TSC1* or *TSC2* inactivating alterations were most frequently identified in urinary bladder (9.6%), liver (6.5%), kidney and renal pelvis (5.2%), and uterine (4.2%) tumors (**Figure 4**)
- In tumors with known/likely inactivating alterations in *TSC1* or *TSC2*, the frequency of mutations in *TP53* (62.2%) and *CDKN2A* (26.2%) (**Figure 5A**) were similar to those in the total population of patients with tumor data in the Foundation Medicine database (58.0% and 21.0%, respectively) where the predominant tumor types are lung, colorectal, and breast tumors overall (**Figure 5B**)
- The patients with *TSC1* or *TSC2* inactivating alterations had a lower frequency of mutations in *KRAS* (10.5%) compared with the total population in the Foundation Medicine database (20.2%)
- While TMB was high (>20) in 20% of patients with known/likely inactivating alterations in *TSC1* or *TSC2*, TMB was low (<6 mutations per megabase) in approximately half (51.6%) of patients with known/likely inactivating alterations in *TSC1* or *TSC2* (**Figure 6A**). TMB was low in the majority (71.1%) of the total population of patients in the Foundation Medicine database (**Figure 6B**)
- Tumor samples with known/likely inactivating alterations in *TSC1* or *TSC2* were mostly microsatellite stable (82.2%) across tumor types (**Figure 6C**), and this was similar to the total patient population of the Foundation Medicine database (90.3%) (**Figure 6D**)

Figure 6. Tumor mutational burden and microsatellite instability status in patients with known/likely inactivating alterations in *TSC1* or *TSC2* (A, C) and in the total patient population of the Foundation Medicine database (B, D)

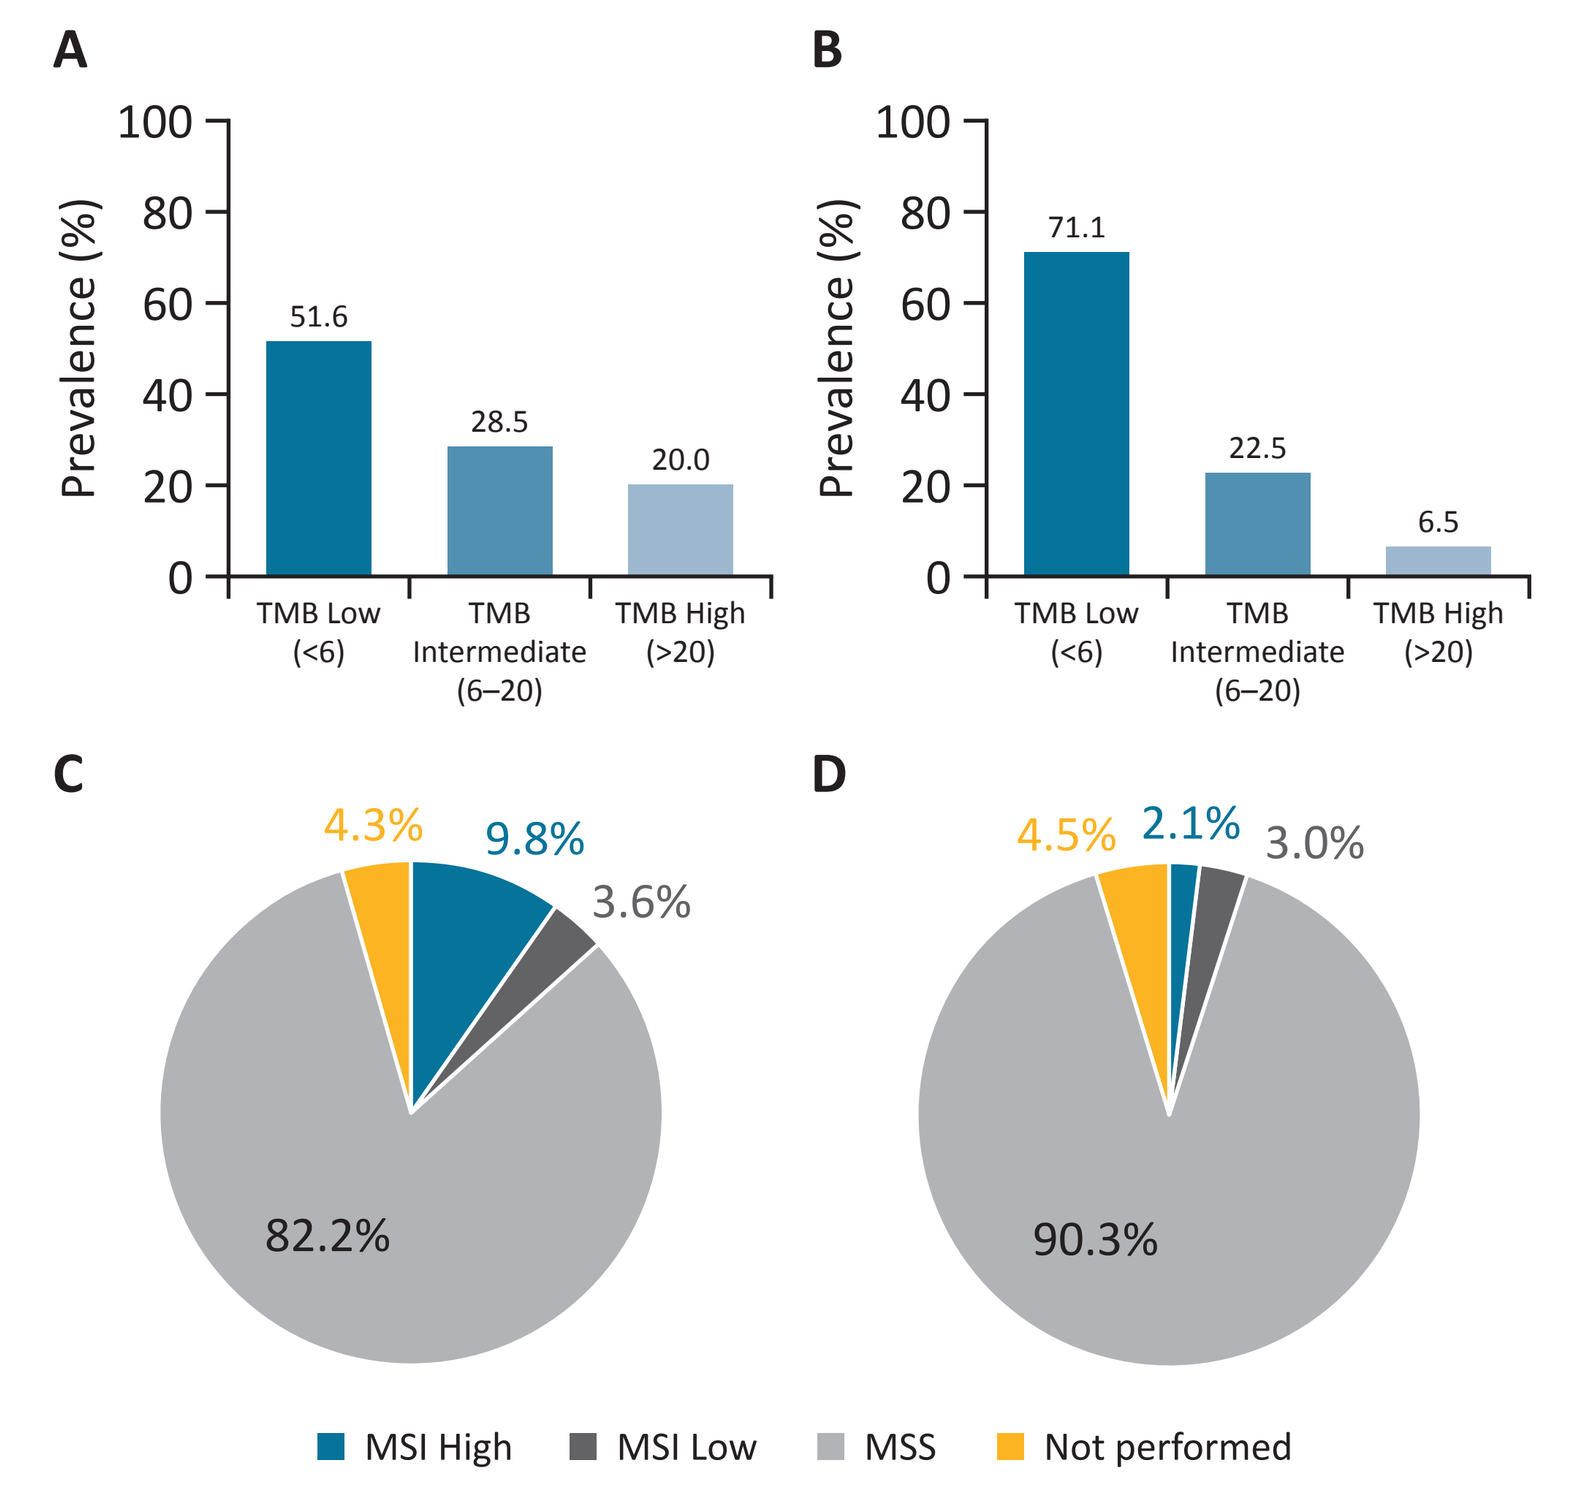
A
Prevalence (%)
0
20
40
60
80
100
51.6
28.5
20.0
TMB Low (<6)
TMB Intermediate (6–20)
TMB High (>20)
B
Prevalence (%)
71.1
22.5
6.5
TMB Low (<6)
TMB Intermediate (6–20)
TMB High (>20)
C
4.3%
9.8%
3.6%
82.2%
D
4.5%
2.1%
3.0%
90.3%
MSI High
MSI Low
MSS
Not performed

Note: Percentages do not add up to 100 due to rounding.
MSI, microsatellite instability; **MSS**, microsatellite stable; **TMB**, tumor mutational burden.

Acknowledgments & Disclosures

Medical writing and editorial assistance were provided by Heather Caballes, PhD, of Twist Medical, and were funded by Aadi Bioscience, Inc.

DJK has no conflicts of interest. **NAP** and **WHN** are employees of and stockholders in Aadi Bioscience, Inc. **GI** reports consulting or scientific advisory board membership for Mirati Therapeutics, Janssen, Bayer, Basilea, Flare Therapeutics, Aadi Bioscience, and LOXO at Lilly; speaker's fees for Gilead Sciences and the Lynx Group; and research funding from Mirati Therapeutics, Novartis, Debiopharm Group, Bayer, Janssen, Flare Therapeutics, Seagen, Aadi Bioscience, and LOXO at Lilly.

References

1. Peng Y, et al. *Front Oncol.* 2022;12:819128.
2. He Y, et al. *Signal Transduct Target Ther.* 2021:6:425.
3. Yang J, et al. *Mol Cancer.* 2019;18:26.
4. Rehbein U, et al. *Front Cell Dev Biol.* 2021;9:751892.
5. FYARRO® (sirolimus protein-bound particles for injectable suspension [albumin-bound]). Package insert. Aadi Bioscience, Inc.: Pacific Palisades, CA; December 2021.
6. Wagner AJ, et al. *J Clin Oncol.* 2021;39:3660–70.
7. Dickson MA, et al. *J Clin Oncol.* 2021;39:15_suppl, 3111.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from AACR-NCI-EORTC or the author of the poster