Response to Treatment With *nab*-Sirolimus Among Patients With Malignant PEComa of Uterine Origin: A Subanalysis From AMPECT

B010

Vinod Ravi, MD[1]; Mark A. Dickson, MD[2]; Kristen Ganjoo, MD[3]; Li Ding, MS, MA[4]; Anita N. Schmid, PhD[4]; Norma A. Palma, PhD[4]; Willis H. Navarro, MD[4]; Andrew J. Wagner, MD, PhD[5]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Memorial Sloan Kettering Cancer Center, New York, NY; [3]Stanford University, Stanford, CA; [4]Aadi Bioscience, Pacific Palisades, CA; [5]Dana -Farber Cancer Institute, Boston, MA

Objectives

- **To describe the outcomes of a subset of patients with malignant PEComa of uterine origin in the AMPECT trial**

KEY FINDINGS

Patients with malignant PEComa of uterine origin in AMPECT attained a response rate of 42.9%

- **This is similar to the response rate of 38.7% observed in the overall AMPECT population and substantially higher than the response rate of 12.5% reported in a retrospective study of patients with uterine PEComas treated with other mTORis**
- **Responses were rapid and durable**

The safety profile for this subset of patients was consistent with what has been observed with *nab*-sirolimus and other mTORis; TRAEs were manageable and did not result in drug discontinuation

***nab*-Sirolimus is being further evaluated in mTOR driven cancers, including PRECISION 1 (NCT05103358), a tumor-agnostic trial of patients with *TSC1* or *TSC2* inactivating alterations, and in an open-label, single arm phase 2 study in advanced or recurrent endometrioid endometrial cancer (NCT05997017)**

AACR Special Conference in Cancer Research: Endometrial Cancer: Transforming Care Through Science; Boston, MA; November 16–18, 2023

Correspondence: MedInfo@AadiBio.com

BACKGROUND

- Malignant perivascular epithelioid cell tumor (PEComa) is an aggressive, rare sarcoma, with a strong female predominance, for which cytotoxic chemotherapies provide limited patient benefit[1–3]
- PEComas most frequently affect the uterus within the female genital tract[3]
- Inactivation of *TSC1* or *TSC2* tumor suppressor genes, upstream of mTORC1, is commonly associated with malignant PEComa; therefore, the mTOR pathway presents an opportunity for targeted therapy
- High response rates have been reported in a small number of patients with PEComas of uterine or gynecologic origin who were treated with mTOR inhibitors (mTORis)[4–9]
- However, a retrospective analysis reported that patients with PEComas of uterine origin had numerically lower response rates to mTORis (12.5%) compared to those with extrauterine primary tumors (51.6%)[1], confirming that further studies are needed in the uterine PEComa subset
- *nab*-Sirolimus is an intravenous mTORi that utilizes albumin-bound nanoparticle technology to achieve greater tumor growth inhibition, higher intratumoral drug levels and more complete pathway suppression than oral mTORis[10]
- *nab*-Sirolimus is approved by the US Food and Drug Administration for the treatment of adults with advanced malignant PEComa based on the primary analysis results of the AMPECT trial, which showed a confirmed overall response rate (ORR) of 38.7%, regardless of mutation status[11]
- Here, we describe outcomes of a subset of patients with malignant PEComa of uterine origin, representing 23% of the efficacy-evaluable patients in AMPECT

METHODS

- AMPECT (NCT02494570) was an open-label, multicenter, phase 2 registration study in adult patients (≥18 years) with a histologically confirmed diagnosis of malignant PEComa by a central pathologist, and Eastern Cooperative Oncology Group (ECOG) performance status (PS) score 0 or 1
- Patients received *nab*-sirolimus 100 mg/m^2 intravenously on days 1 and 8 of a 21-day cycle until disease progression or unacceptable toxicity (**Figure 1**)
- The primary endpoint was ORR (defined as the number of patients with a partial response [PR] and complete response [CR]) by independent radiology review
- Secondary endpoints included time to response, duration of response (DOR), progression-free survival, and safety, including treatment-related adverse events (TRAEs), serious TRAEs, and TRAEs leading to drug withdrawal or dose reduction
- The disease control rate (DCR), an exploratory endpoint, was defined as CR + PR + stable disease (SD) of ≥12 weeks

Figure 1. AMPECT study design

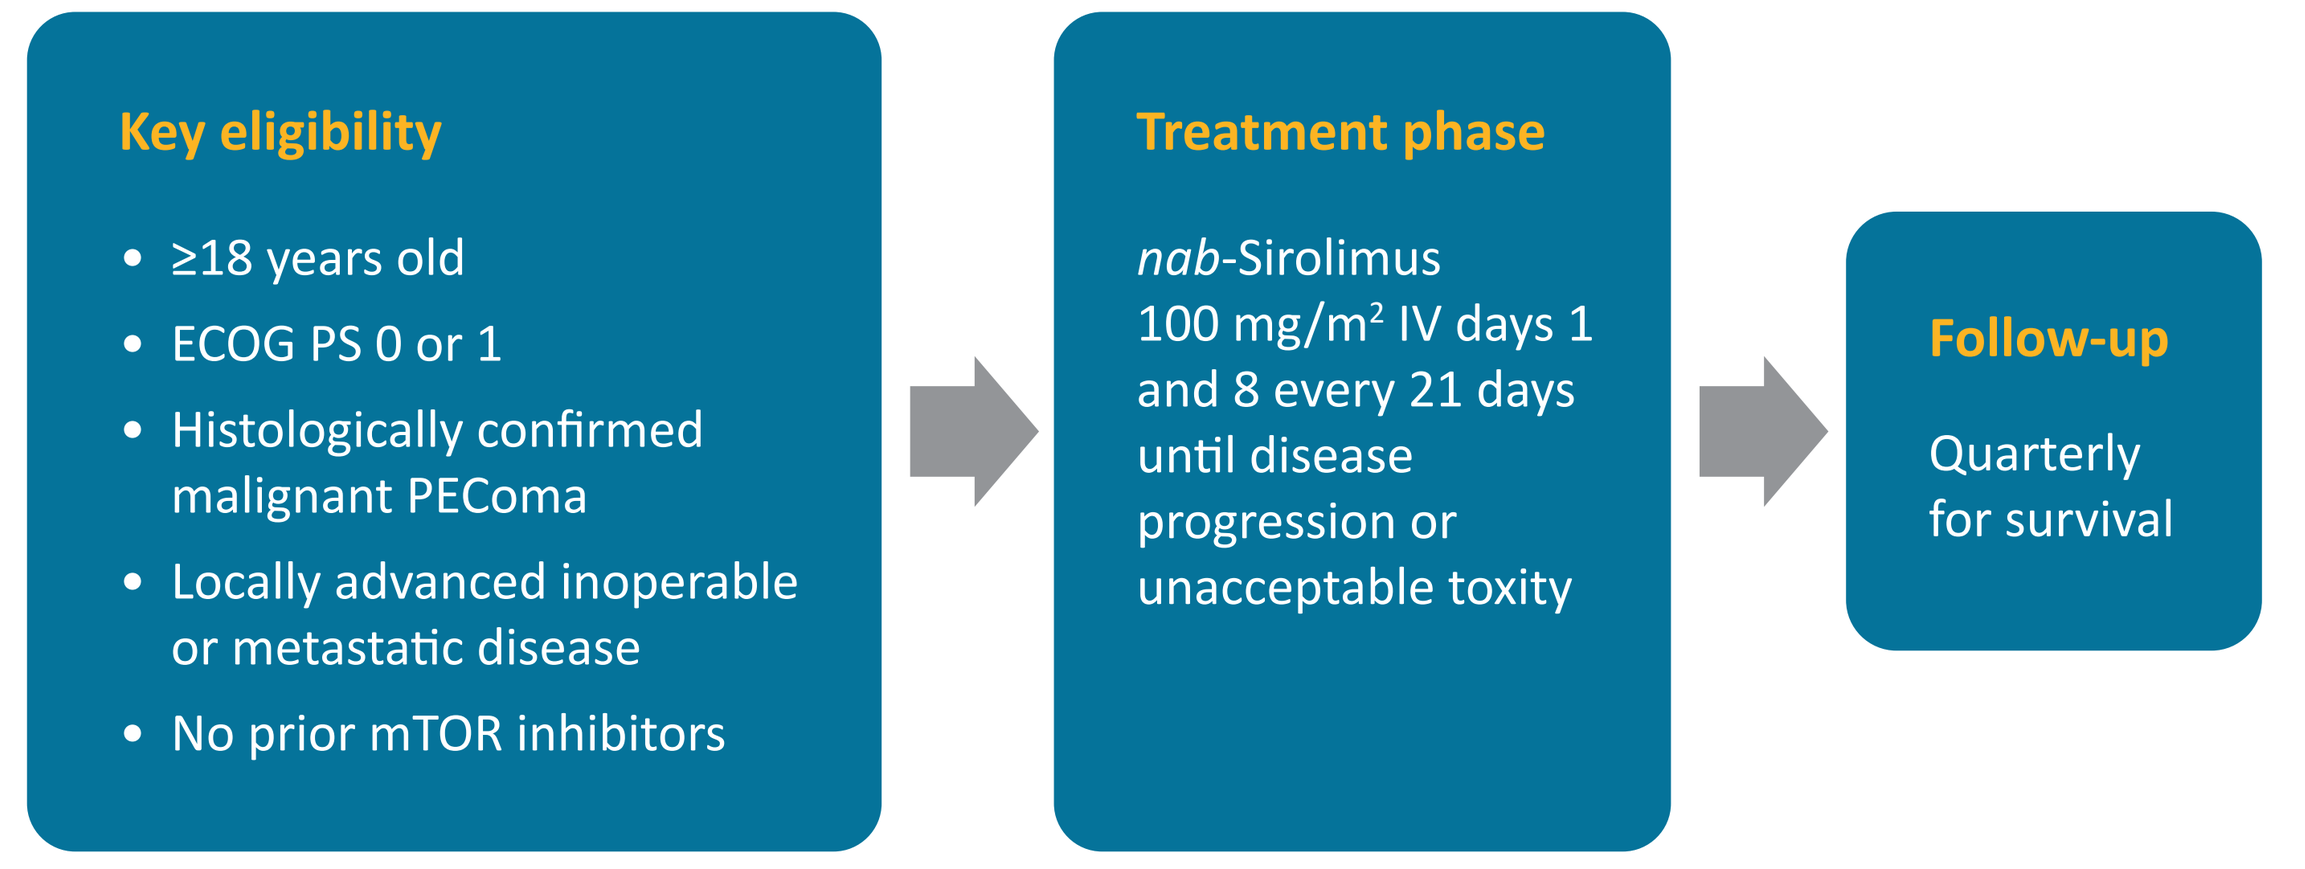
Key eligibility
≥18 years old
ECOG PS 0 or 1
Histologically confirmed malignant PEComa
Locally advanced inoperable or metastatic disease
No prior mTOR inhibitors
Treatment phase
nab-Sirolimus 100 mg/m2 IV days 1 and 8 every 21 days until disease progression or unacceptable toxity
Follow-up
Quarterly for survival

ECOG, Eastern Cooperative Oncology Group; **IV**, intravenously; **mTOR**, mammalian/mechanistic target of rapamycin; **PEComa**, perivascular epithelioid cell tumor; **PS**, performance status.

RESULTS

- Of the 31 efficacy-evaluable patients in AMPECT, 7 patients with malignant PEComa of uterine origin were treated with *nab*-sirolimus and were evaluable for both safety and efficacy
- The median age of all 7 females was 64 years, and 71% (n=5) were White
- Of the 7 patients, 2 had *TSC2*, and 1 had *TSC1* inactivating alterations
- Six of the 7 patients presented with 1 or 2 metastatic sites at baseline, and 1 patient presented with >3 metastatic sites
- Prior treatment included surgery for all 7 patients (100%) and prior chemotherapy for 1 patient (14%)
- The ECOG PS at baseline was 0 or 1 for all patients
- The ORR was 42.9% (95% CI, 9.9%–81.6%)
 - Three patients (42.9%; 95% CI, 9.9%–81.6%) had a confirmed PR, of which all 3 responses (**Figure 2**) were seen in patients with tumors harboring somatic inactivating alterations in *TSC1* or *TSC2*
 - Two patients (28.6%; 95% CI, 3.7%–71.0%) had confirmed SD with at least 12-week duration, and 2 patients (28.6%; 95%CI, 3.7–71.0%) had progressive disease
- DCR (n=5) was 71.4% (95% CI, 29.0%–96.3%)
- Time to response in the 3 responders was rapid (1.4, 1.4, and 1.5 months; **Figure 3**)
- DOR was 5.6, 36.0+, and 39.7 months for the 3 responders (**Figure 3**)
 - Median DOR was 39.7 months (95% CI, 5.6 months to not evaluable [NE]) for responders
- Median progression-free survival was 6.9 months (95% CI, 1.3 months to not reached)

Figure 2. Target tumor response

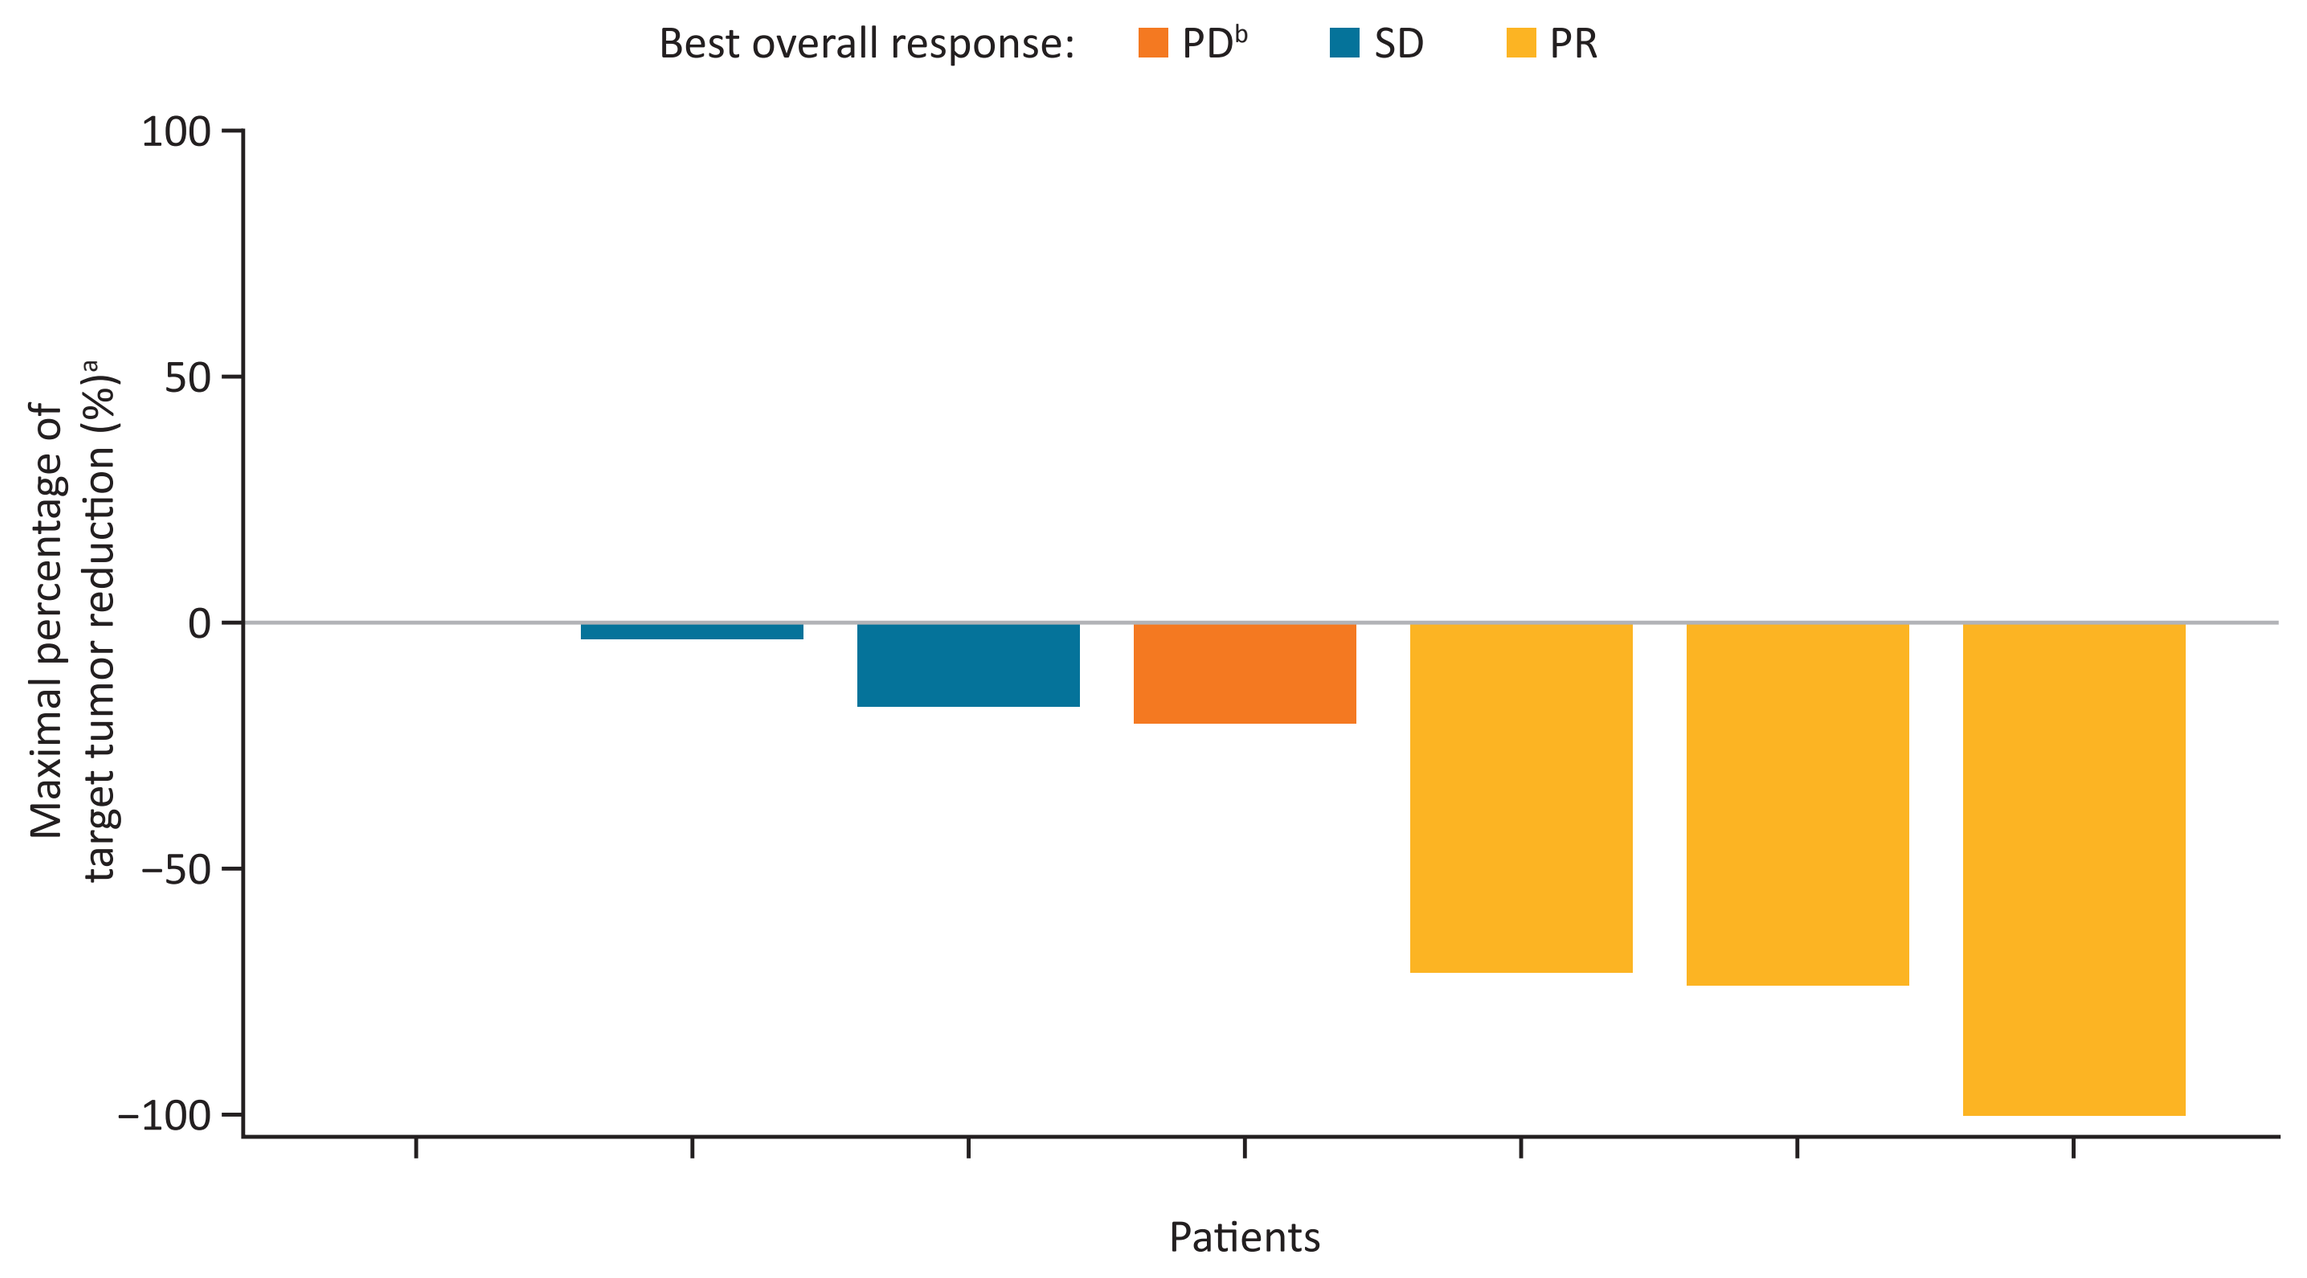
Best overall response:
PD[b]
SD
PR
100
50
0
−50
−100
Maximal percentage of target tumor reduction (%)[a]
Patients

[a]Maximal percentage of tumor reduction, the minimum percent change in the sum of diameters from baseline based on all target lesions across all post-baseline assessments for each patient whose best overall response was not PD. [b]One patient did not have a baseline target tumor lesion but was classified as having PD by independent radiology review. Target tumor reduction may not match best overall response assessment, which takes into consideration nontarget lesions and observations of new lesions per RECIST v1.1. **PD**, progressive disease; **PR**, partial response; **SD**, stable disease.

Figure 3. Rapid and durable responses

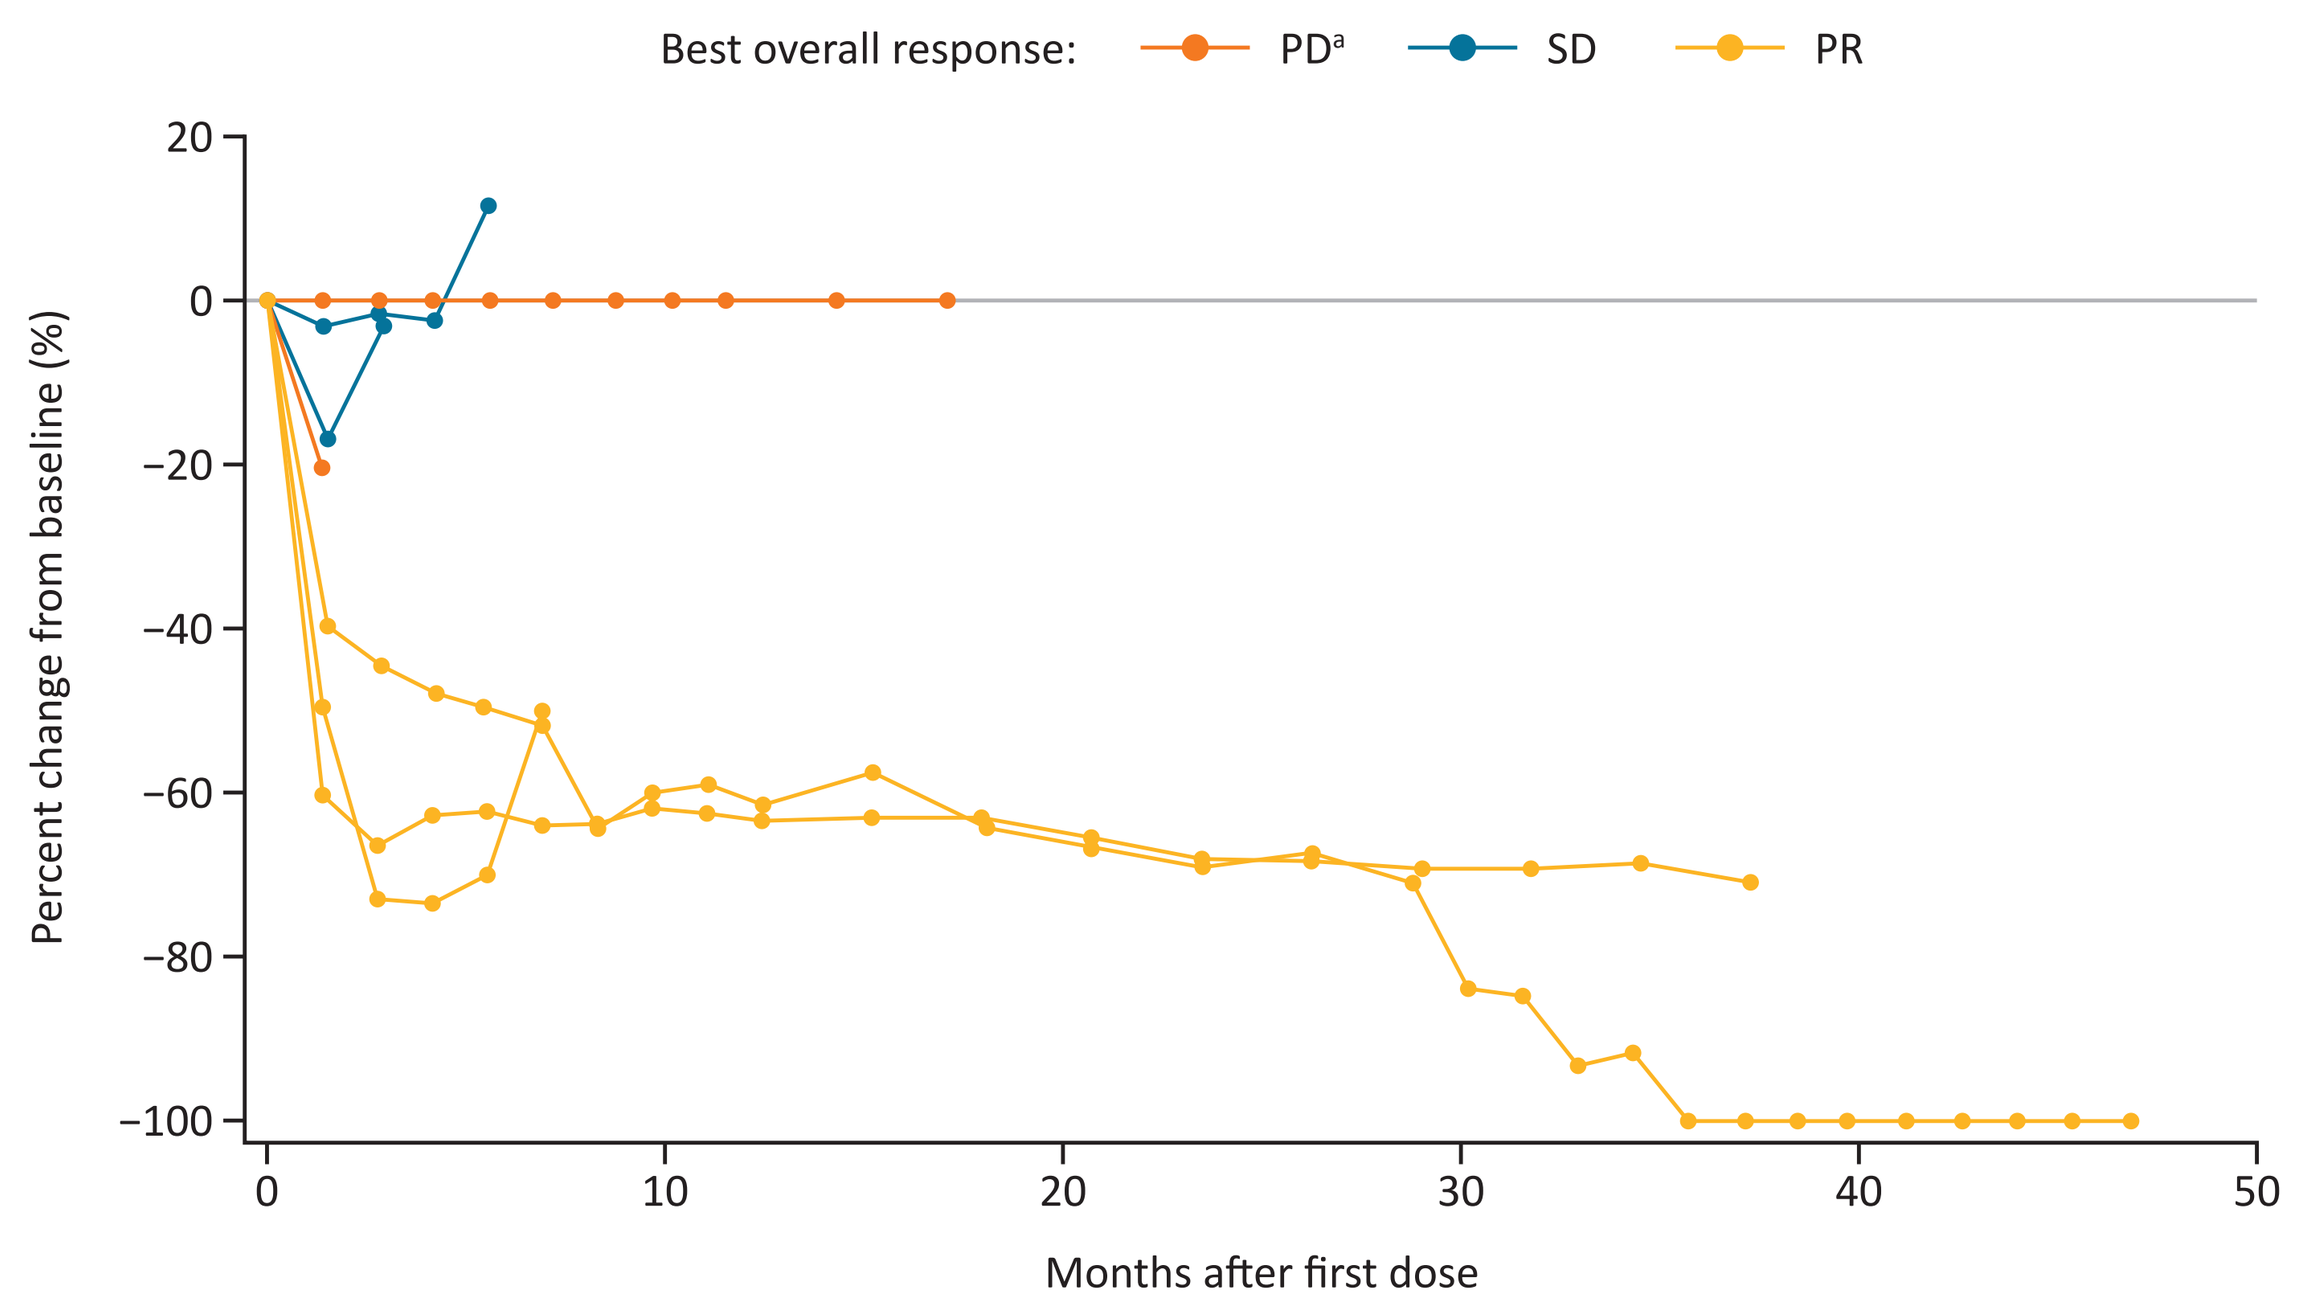
Best overall response:
PD[a]
SD
PR
20
0
−20
−40
−60
−80
−100
Percent change from baseline (%)
0
10
20
30
40
50
Months after first dose

[a]One patient did not have a baseline target tumor lesion but was classified as having PD by independent radiology review. Target tumor reduction may not match best overall response assessment, which takes into consideration nontarget lesions and observations of new lesions per RECIST v1.1. **PD**, progressive disease; **PR**, partial response; **SD**, stable disease.

- All 7 patients had ≥1 TRAE (grade 3, 43%); no grade ≥4 or serious TRAEs were observed
- Four patients (57%) and 2 patients (29%) experienced a TRAE that led to drug interruption or dose reduction, respectively; however, no patients discontinued treatment due to a TRAE
- TRAEs are shown in **Table 1**
 - The most common hematological TRAEs were: anemia (43%), leukopenia, lymphopenia, and neutropenia (29% each)
 - The most common nonhematological TRAEs were: stomatitis (86%); edema and rash (71% each); and decreased appetite, fatigue, and nausea (57% each)

Table 1. TRAEs Occurring in ≥25% of Patients (N=7)

TRAE by Preferred Term, n (%)	Any grade	Grade 3
Patients with any TRAE	7 (100)	3 (42.9)[a]
Hematologic TRAEs		
Anemia[b]	3 (42.9)	0
Leukopenia[b]	2 (28.6)	0
Lymphopenia[b]	2 (28.6)	0
Neutropenia[b]	2 (28.6)	0
Nonhematologic TRAEs		
Stomatitis[b]	6 (85.7)	2 (28.6)
Edema[b]	5 (71.4)	0
Rash[b]	5 (71.4)	0
Decreased appetite	4 (57.1)	0
Fatigue	4 (57.1)	0
Nausea	4 (57.1)	0
Alopecia	3 (42.9)	0
Diarrhea[b]	3 (42.9)	0
Hyperglycemia[b]	3 (42.9)	0
Weight decreased	3 (42.9)	0
Constipation	2 (28.6)	0
Dry mouth	2 (28.6)	0
Hypercholesterolemia[b]	2 (28.6)	0
Hypomagnesemia[b]	2 (28.6)	0
Nail disorder	2 (28.6)	0
Peripheral neuropathy[b]	2 (28.6)	0
Vomiting	2 (28.6)	0

Percentages are based on number of patients in the Safety Analysis Population. Patients with multiple events from the same system organ class or preferred term are counted only once. System organ class and preferred terms are assigned based on MedDRA version 24.0. Toxicity is graded with NCI CTCAE version 4.03. [a]One patient reported Grade 3 elevated amylase and insomnia. [b]Adverse Events of Special Interest and related referred terms are grouped.

TRAE, treatment-related adverse event.

Acknowledgments & Disclosures

ACKNOWLEDGMENTS: Editorial assistance was provided by Heather Caballes, PhD of TWIST Medical, and was funded by Aadi Bioscience, Inc.

DISCLOSURES: VR is a consultant or on the advisory board for Aadi Bioscience, Inc. and Daichi Sankyo; has received research grants from Aadi Bioscience, Athenex, Novartis, and TRACON Pharma; and owns ownership interest in AstraZeneca, Merck, Moderna Therapeutics, Pfizer, and TRACON Pharma. **MAD** has received research grants from Aadi Bioscience, Inc, Eli-Lilly, and Sumitomo. **KG** has no conflicts of interest. **LD**, **ANS**, **NAP**, and **WHN** are employees of and own stock in Aadi Bioscience. **AJW** is a consultant or on the advisory board for Aadi Bioscience, BioAlta, Boehringer-Ingelheim, Cogent Biosciences, Daiichi Sankyo, Deciphera, Five Prime Therapeutics, Lilly, Mundipharma, NanoCarrier, and Servier; and has received research grants to his institution from Aadi Bioscience, Cogent Biosciences, Deciphera, Foghorn Therapeutics, Karyopharm Therapeutics, Plexxikon, and Rain Therapeutics.

References

1. Sanfilippo R, et al. *Clin Cancer Res.* 2019;25(17):5295–5300.
2. Bleeker JS, et al. *Sarcoma.* 2012;2012:5416–26.
3. Armah HB, Parwani AV. *Diagn Pathol.* 2007;2:45.
4. Starbuck KD, et al. *Anticancer Res.* 2016;36(11):6161–4.
5. Dickson MA, et al. *Int J Cancer.* 2013;132(7):1711–7.
6. Wagner AJ, et al. *J Clin Oncol.* 2010;28(5):835–40.
7. Italiano A, et al. *Ann Oncol.* 2010;21(5):1135–7.
8. Benson C, et al. *Anticancer Res.* 2014;34(7):3663-8.
9. Ghosh I, et al. *Indian J Med Paediatr Oncol.* 2014;35(1):99–102.
10. Hou S, et al. AACR-NCI-EORTC Virtual International Conference on Molecular Targets and Cancer Therapeutics, October 7–10, 2021, Virtual.
11. Wagner AJ, et al. *J Clin Oncol.* 2021;39(33):3660–70.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.