Phase 2, Multicenter, Global, Open-Label Basket Trial of *nab*-Sirolimus for Patients With Inactivating Alterations in *TSC1* and *TSC2* (PRECISION 1)

B011

Debra L Richardson, MD[1]; Gopa Iyer, MD[2]; Li Ding, MS, MA[3]; Anita N. Schmid, PhD[3]; Willis H. Navarro, MD[3]; David J. Kwiatkowski, MD, PhD[4]; Jordi Rodon Ahnert, MD, PhD[5]

[1]Stephenson Cancer Center, OU Health, Oklahoma City, OK; [2]Memorial Sloan Kettering Cancer Center, New York, NY; [3]Aadi Bioscience, Pacific Palisades, CA; [4]Brigham and Women's Hospital, Boston, MA; [5]MD Anderson Cancer Center, Houston, TX

Objective

- **This trial is designed to evaluate the efficacy, safety, and tolerability of *nab*-sirolimus in a patient population with advanced malignancies and limited therapeutic options**

KEY POINTS

- ***nab*-Sirolimus is an mTORi utilizing nanoparticle albumin technology to enhance antitumor activity as shown in animal models**
- **Data from the AMPECT exploratory analysis and an expanded access program suggest *nab*-sirolimus may provide clinically relevant benefit with a manageable safety profile in patients with solid tumors harboring inactivating alterations in *TSC1* and/or *TSC2***
- **Combined, *TSC1* and/or *TSC2* inactivating alterations have been found in ~3% of patients with endometrial cancers; however, there are no specific treatment options for patients with these alterations**
- **PRECISION 1 (NCT05103358) is a registrational, tumor-agnostic trial for patients with solid tumors driven by *TSC1* or *TSC2* inactivating alterations; enrollment began in March 2022, and is open to patients with endometrial cancers with *TSC1* or *TSC2* inactivating alterations**

AACR Special Conference in Cancer Research: Endometrial Cancer: Transforming Care Through Science; Boston, MA; November 16–18, 2023

Correspondence: MedInfo@AadiBio.com

INTRODUCTION

- *TSC1* and *TSC2* form a protein complex and together are critical negative regulators of mTOR complex 1 activation[1] (**Figure 1**)

Figure 1. PI3K-AKT-mTOR Pathway[2,4]

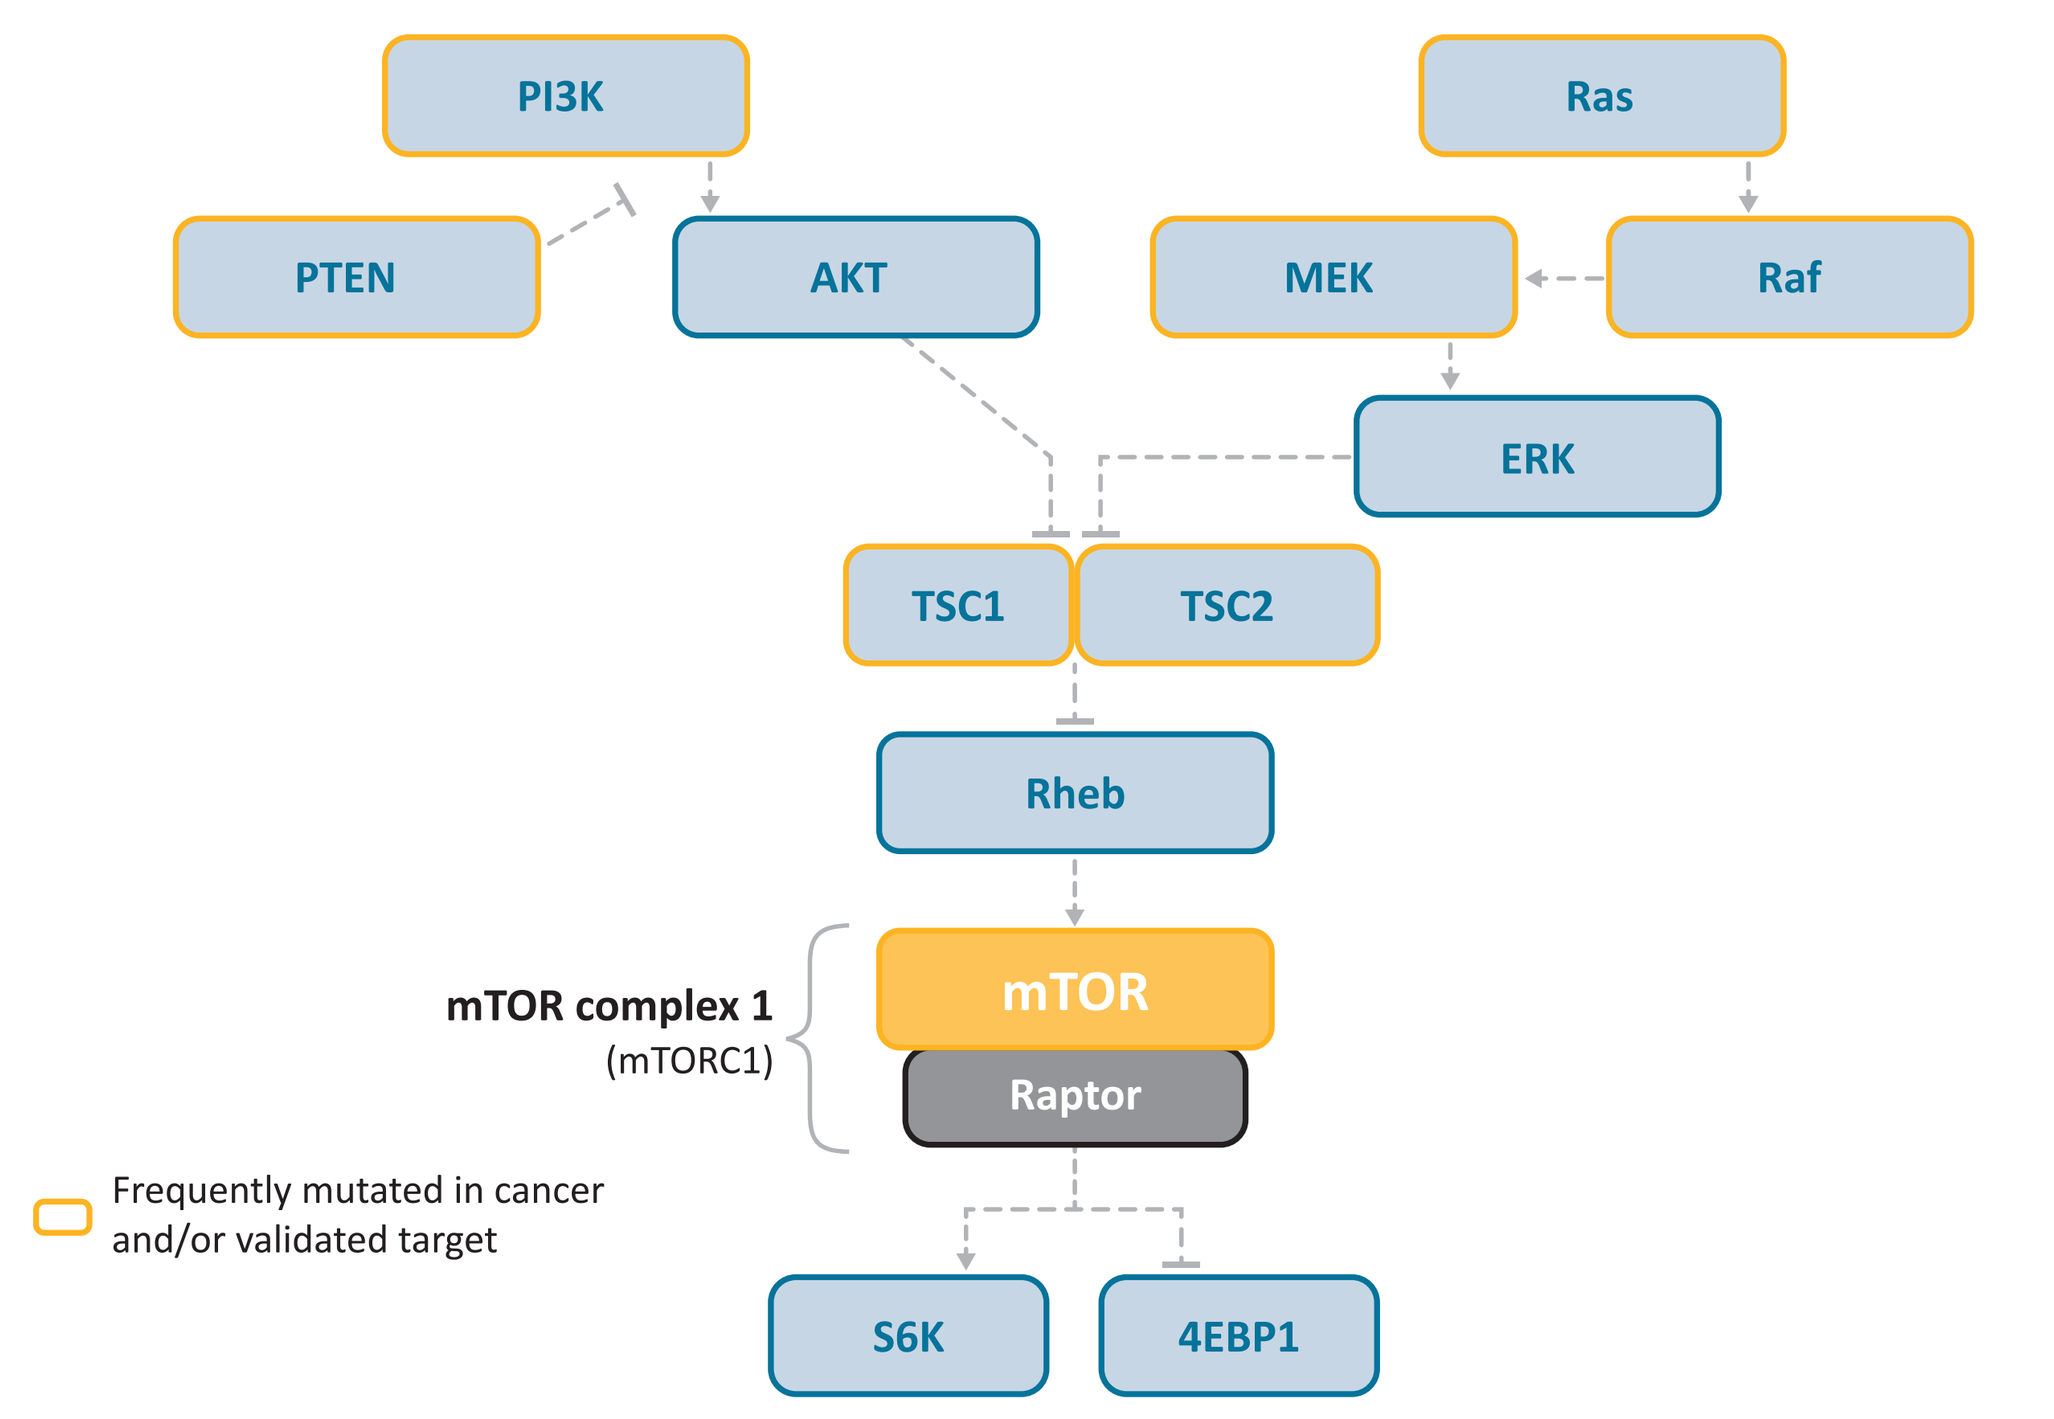
PI3K
Ras
PTEN
AKT
MEK
Raf
ERK
TSC1
TSC2
Rheb
mTOR complex 1
(mTORC1)
mTOR
Raptor
Frequently mutated in cancer and/or validated target
S6K
4EBP1

- The utility of oral mTOR inhibitors (mTORis), such as sirolimus, as pan-cancer agents may be restricted by low bioavailability and dose-limiting toxicity[2,3]
- To improve the pharmacologic properties of sirolimus, *nab*-sirolimus, a nanoparticle albumin-bound form of sirolimus, was developed for intravenous use
- In animal models, *nab*-sirolimus demonstrated significantly higher intratumor drug concentrations, greater tumor growth inhibition, improved survival, and greater inhibition of the downstream marker of mTOR activity, phosphorylated-S6 ribosomal protein, relative to equal weekly doses of sirolimus and everolimus[4]
- *nab*-Sirolimus is approved in the United States for the treatment of adult patients with advanced, malignant perivascular epithelioid cell tumor (PEComa)[5] based on clinical efficacy and safety results from the AMPECT trial (NCT02494570)[6]
- Results from the AMPECT exploratory biomarker analysis demonstrated rapid and durable responses in patients with *TSC1* or *TSC2* inactivating alterations and suggested significant clinical benefit (**Figure 2**)[7]
- The safety profile in the overall study population was consistent with other mTORis with no new or unexpected safety signals[8]
 - The most common any grade, nonhematologic treatment-related adverse events (TRAEs) were stomatitis (28/34, [82%]), fatigue (21/34 [62%]), and rash (21/34 [62%]); and the most common, any-grade hematologic TRAEs were anemia (18/34 [53%]) and thrombocytopenia (12/34 [35%])
 - Most TRAEs were grade 1/2 and were manageable for long-term treatment; no grade ≥4 TRAEs were observed
- Patients with various malignancies bearing *TSC1* or *TSC2* inactivating alterations treated with *nab*-sirolimus as part of the expanded access program (NCT03817515) showed evidence of response (partial response in 5/7 patients) and manageable toxicities[9]
- Inactivating alterations in *TSC1* and/or *TSC2* have been observed in several types of cancer, including in ~3% of patients with endometrial cancers, combined; however, no treatment options exist specifically for patients with these alterations (**Figure 3**)
- The phase 2 PRECISION 1 trial was initiated to evaluate the potential of mTOR inhibition with *nab*-sirolimus for the treatment of patients with solid tumors harboring *TSC1* or *TSC2* inactivating alterations

Figure 2. Exploratory Biomarker Analysis From AMPECT Demonstrated Rapid and Durable Responses in the Subset of Patients with Known *TSC1* or *TSC2* Inactivating Alterations Treated with *nab*-Sirolimus (n=14)

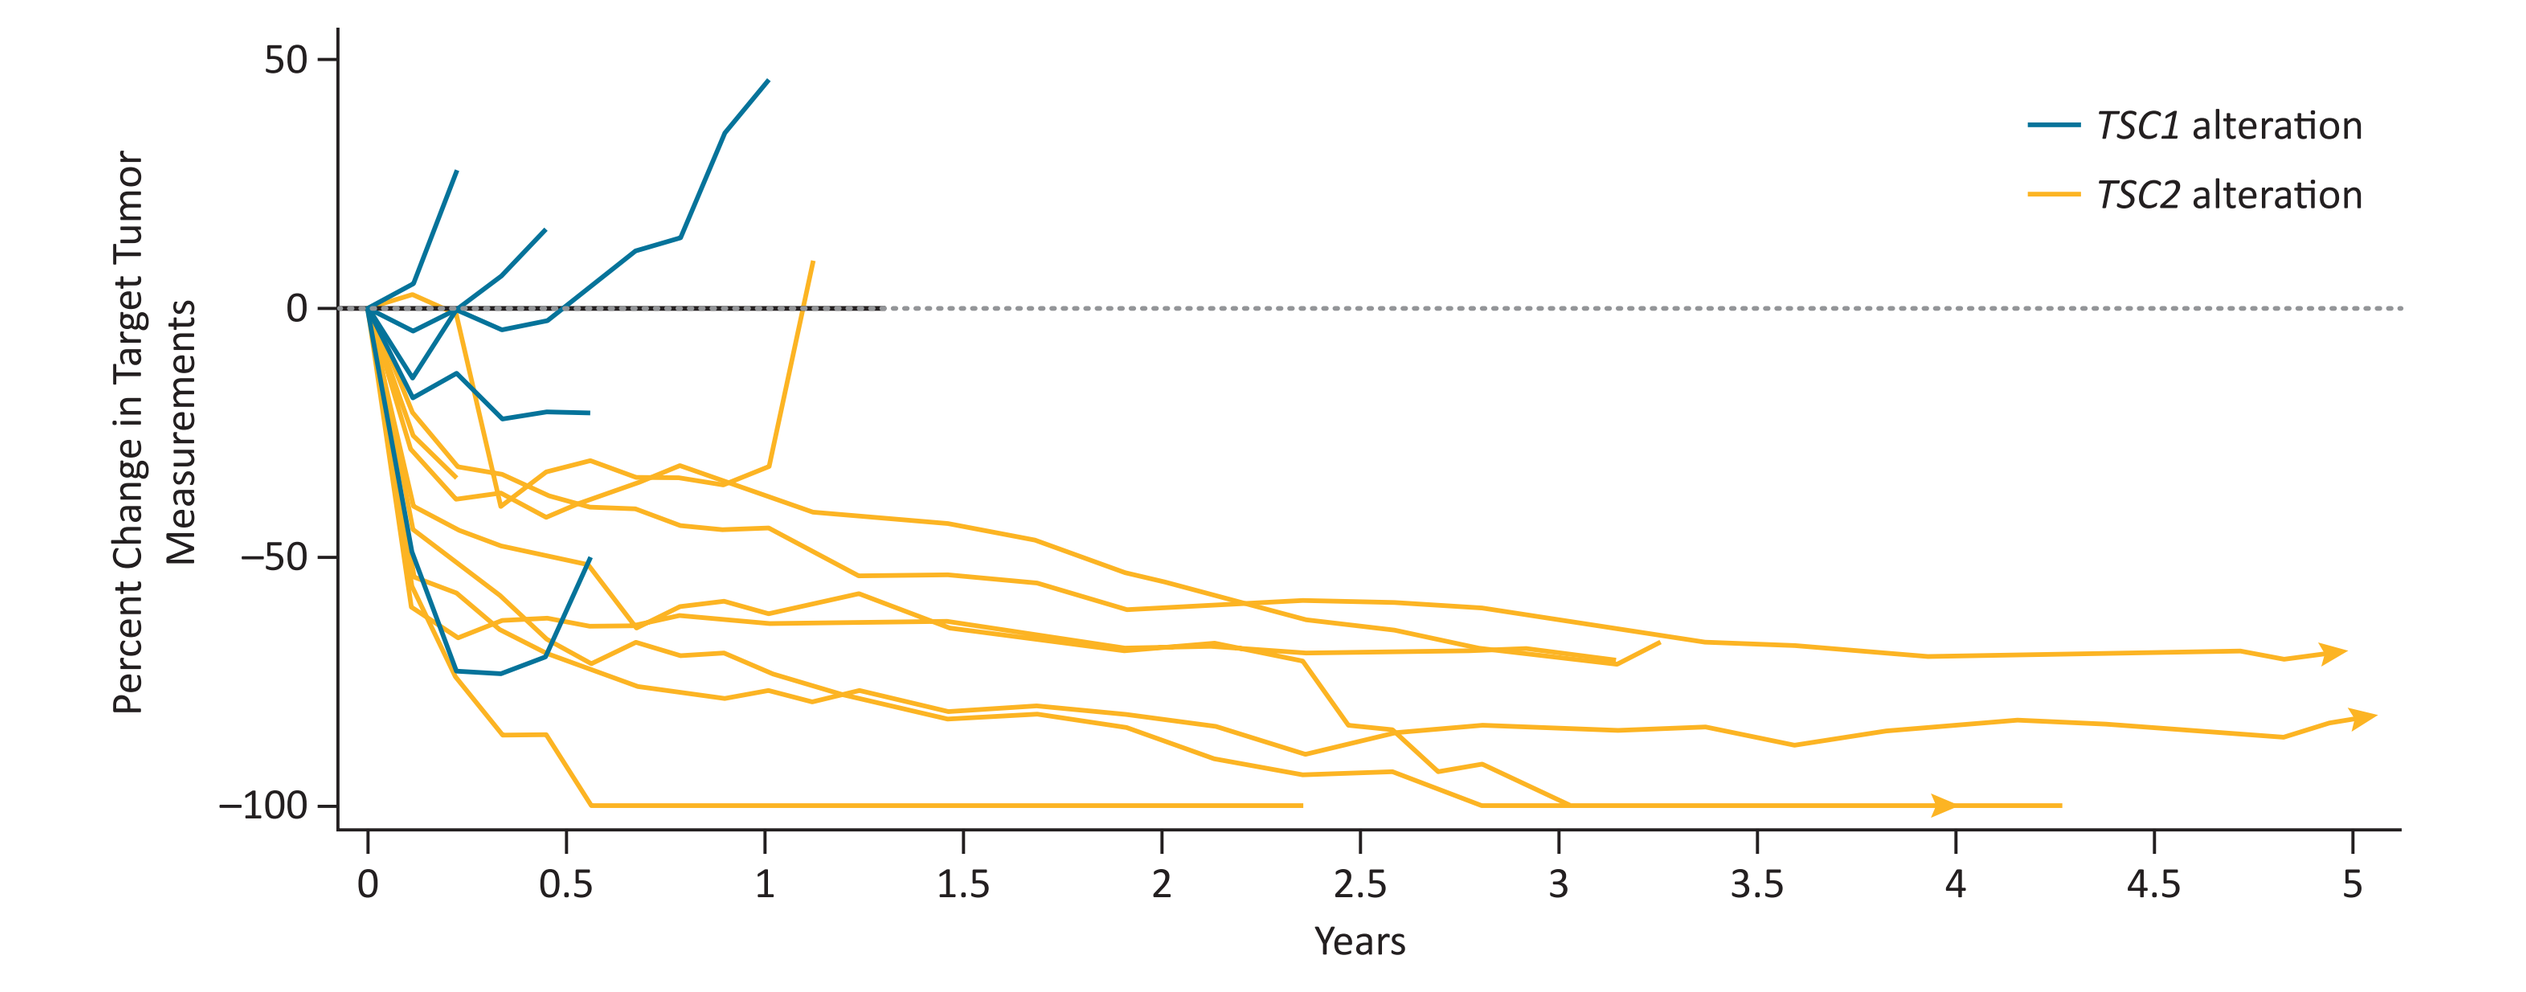
Percent Change in Target Tumor Measurements
50
0
−50
−100
0
0.5
1
1.5
2
2.5
3
3.5
4
4.5
5
Years
TSC1 alteration
TSC2 alteration

Figure 3. Estimated Frequency of Pathogenic and/or Inactivating Alterations in *TSC1* or *TSC2* by Tumor Type

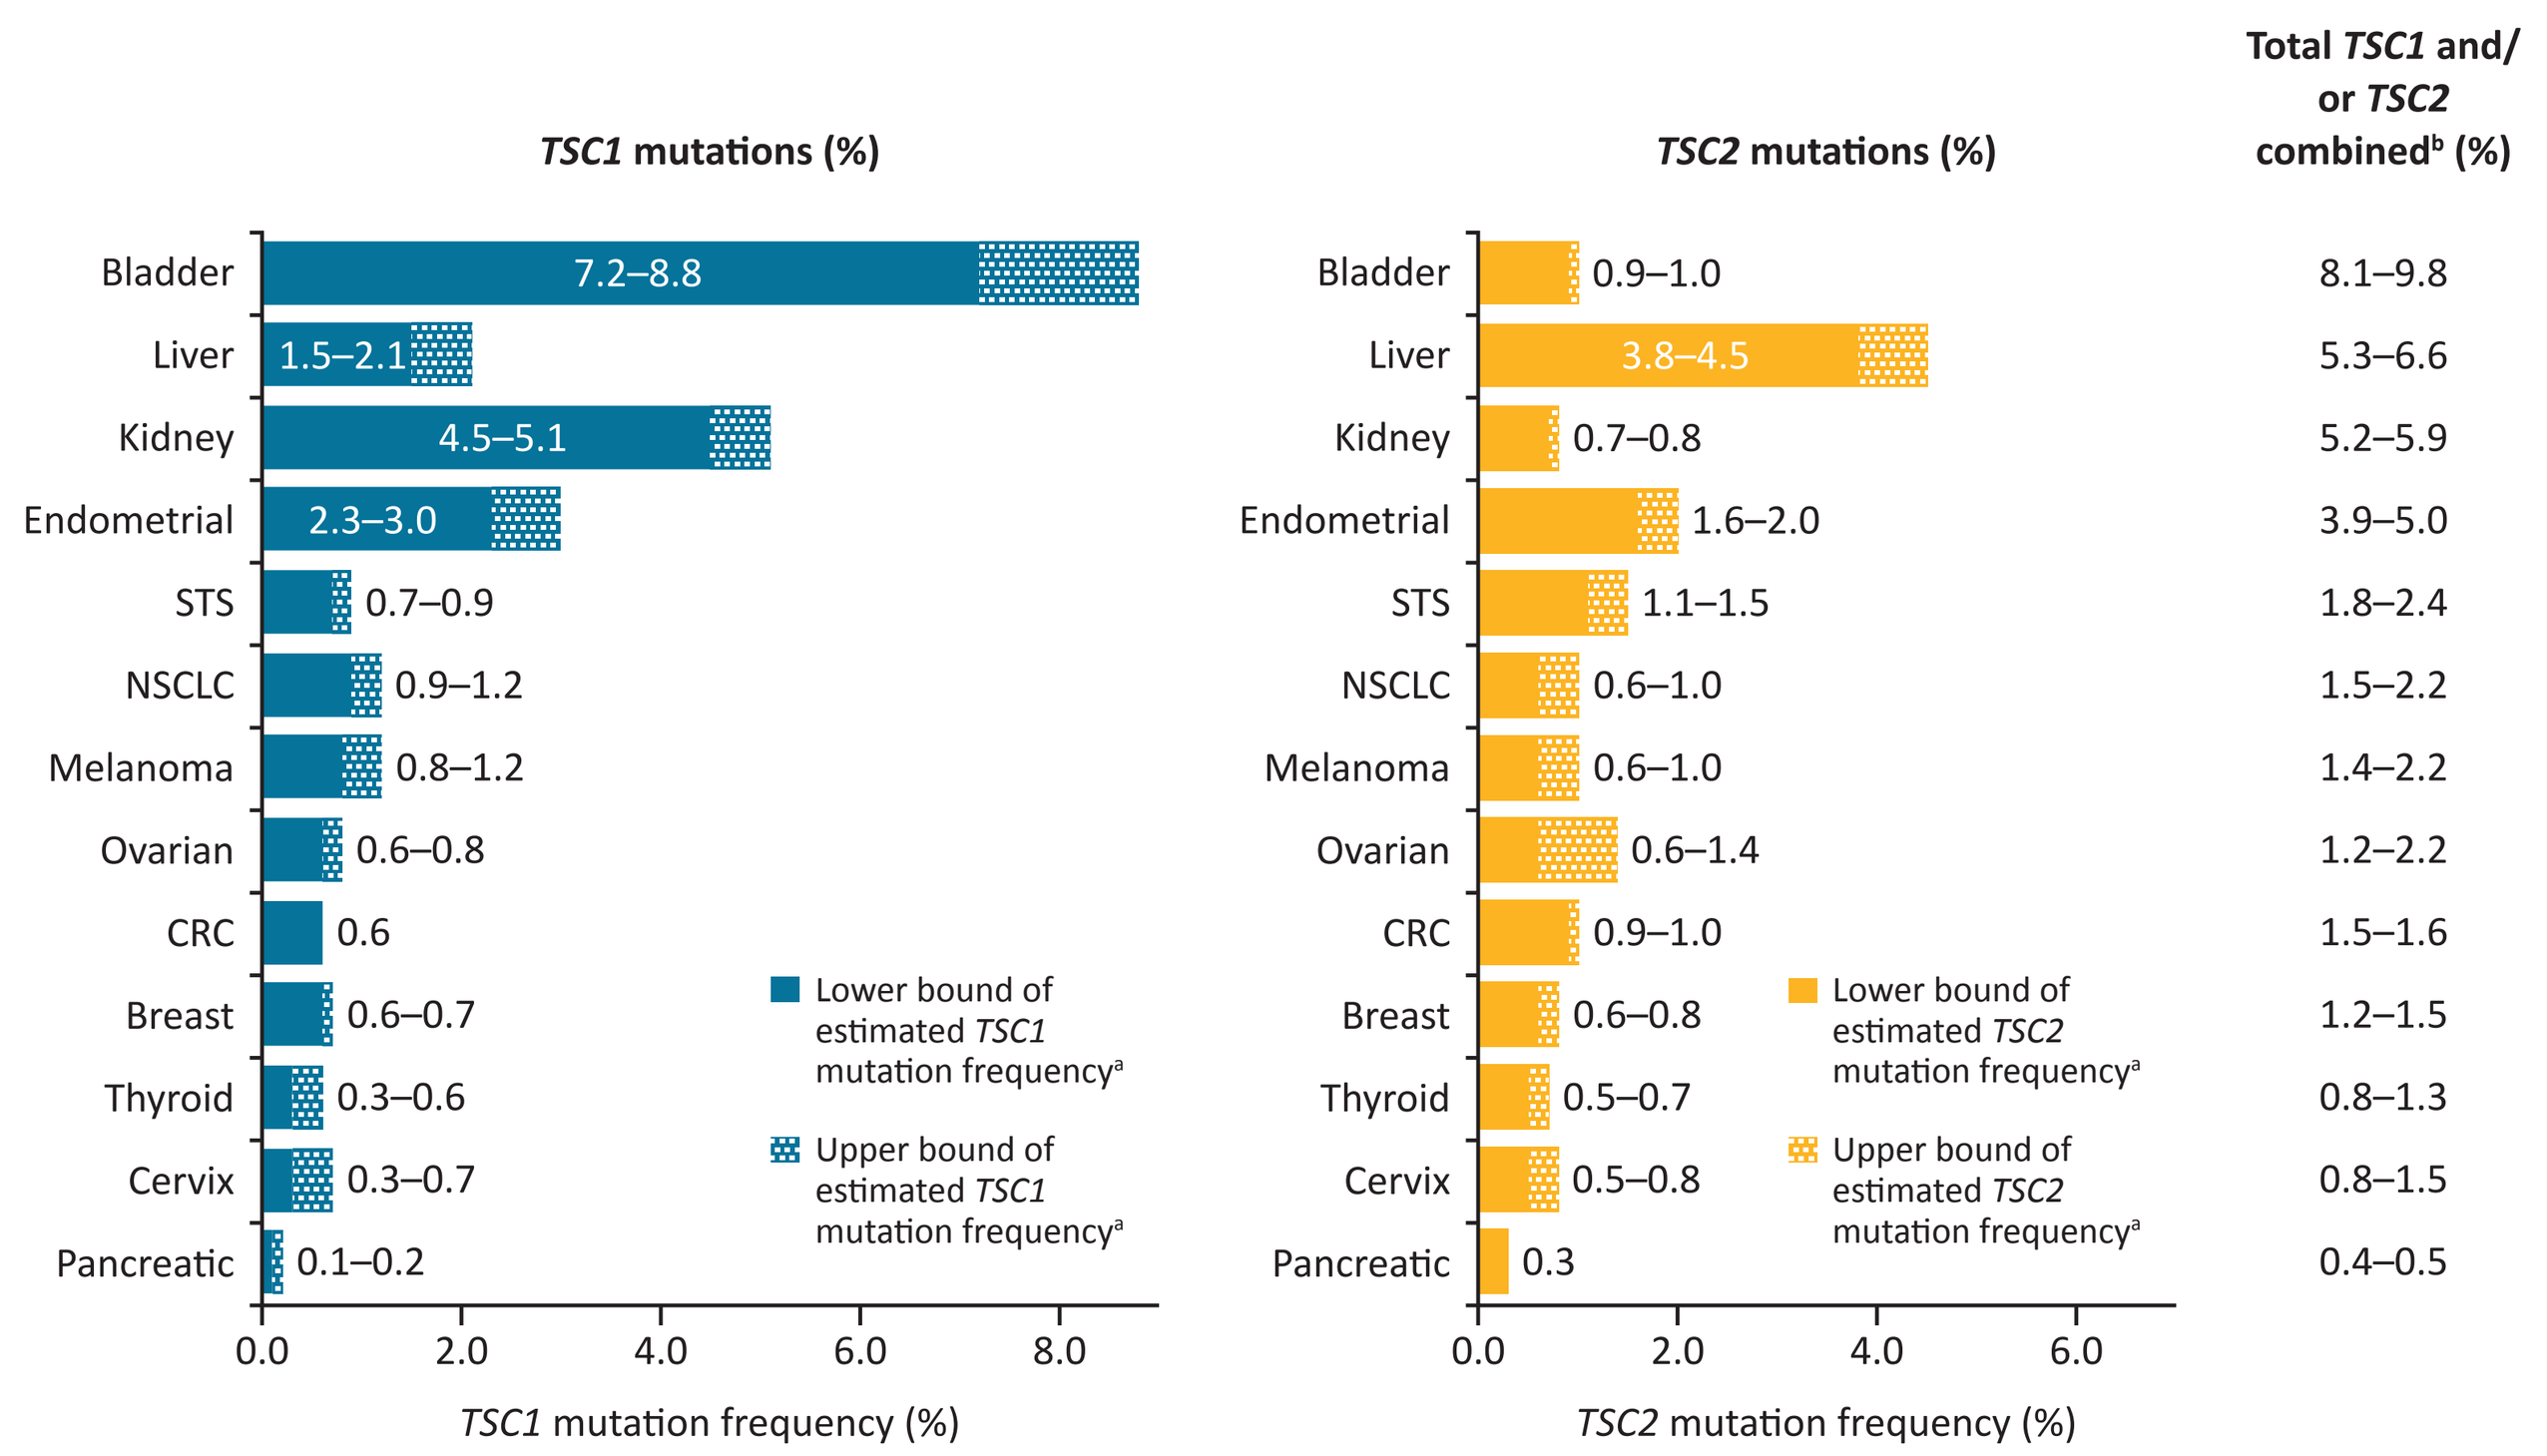
TSC1 mutations (%)
TSC2 mutations (%)
Total TSC1 and/or TSC2 combined[b] (%)
Bladder 7.2–8.8 0.9–1.0 8.1–9.8
Liver 1.5–2.1 3.8–4.5 5.3–6.6
Kidney 4.5–5.1 0.7–0.8 5.2–5.9
Endometrial 2.3–3.0 1.6–2.0 3.9–5.0
STS 0.7–0.9 1.1–1.5 1.8–2.4
NSCLC 0.9–1.2 0.6–1.0 1.5–2.2
Melanoma 0.8–1.2 0.6–1.0 1.4–2.2
Ovarian 0.6–0.8 0.6–1.4 1.2–2.2
CRC 0.6 0.9–1.0 1.5–1.6
Breast 0.6–0.7 0.6–0.8 1.2–1.5
Thyroid 0.3–0.6 0.5–0.7 0.8–1.3
Cervix 0.3–0.7 0.5–0.8 0.8–1.5
Pancreatic 0.1–0.2 0.3 0.4–0.5
0.0 2.0 4.0 6.0 8.0
TSC1 mutation frequency (%)
0.0 2.0 4.0 6.0
TSC2 mutation frequency (%)
Lower bound of estimated TSC1 mutation frequency[a]
Upper bound of estimated TSC1 mutation frequency[a]
Lower bound of estimated TSC2 mutation frequency[a]
Upper bound of estimated TSC2 mutation frequency[a]

[a]Real-world frequencies were derived from tissue-based NGS results for nonhematologic malignancies included in the Foundation Medicine FoundationInsights™ web platform (n=438,974) and data provided by Tempus Labs (n=128,914) with analysis restricted to known or likely oncogenic *TSC1* or *TSC2* inactivating alterations. Upper and lower bounds of frequency ranges were defined as the frequencies calculated from each dataset. [b]The range is reported as the sum of the highest and lowest values for each gene reported in either dataset. **CRC**, colorectal cancer; **NSCLC**, non–small cell lung cancer; **STS**, soft tissue sarcoma; ***TSC1/TSC2***, tuberous sclerosis complex 1/2. Data on file. Aadi Bioscience [May 2023].

STUDY DESIGN

- PRECISION 1 (NCT05103358) is a prospective, phase 2, open-label, multi-institution basket trial evaluating *nab*-sirolimus in patients with malignant solid tumors with pathogenic inactivating alterations in *TSC1* (Arm A) or *TSC2* (Arm B) (**Figure 4**)

Figure 4. Study Design

Key eligibility criteria

- Metastatic or locally advanced solid tumor with a pathogenic inactivating alteration in *TSC1* or *TSC2* identified using NGS in tumor tissue or liquid biopsy[a]
- ≥12 years of age
- mTORi–naïve
- Patients must have received appropriate standard treatments or be unlikely to tolerate or derive clinically meaningful benefit from standard therapy as determined by the investigator
- ECOG PS 0 or 1 (or KPS ≥80; or LPPS ≥80 for pediatric patients)
- ≥1 measurable target lesion by CT scan or MRI

Treatment phase

Arm A: Pathogenic inactivating alterations in *TSC1* n=~60	**Arm B:** Pathogenic inactivating alterations in *TSC2* n=~60

- *nab*-Sirolimus 100 mg/m² administered as an intravenous infusion over 30 minutes on Days 1 and 8 of each 21-day cycle
- Dose reduction allowed per protocol at 75, 56, 45, and 35 mg/m²

***nab*-Sirolimus Dosing Schedule**

21-day Cycle: Day 1, Day 8, Day 15

Repeat until disease progression, unacceptable toxicity, withdrawal of consent, or at investigator discretion

Patients will be enrolled in the United States and globally

NCT05103358

Follow-up

- Response assessment every 6 weeks for the first year, then every 12 weeks thereafter, until disease progression
- Quarterly follow-up until EOS, death, or consent withdrawal[b]

Endpoints[b]

Primary Endpoint
- ORR by independent radiology review (RECIST v1.1)

Secondary Endpoints
- DOR, DCR, PFS by independent radiology review, OS, QoL, safety and tolerability

Key Exploratory Endpoints
- Investigator-assessed ORR, DOR, DCR, and PFS; time on treatment; association between mutational profile and clinical outcomes

[a]Central confirmation of *TSC1* and *TSC2* inactivating alterations is via evaluation of NGS reports. Patients will be enrolled only after central confirmation of eligibility. *TSC1* and *TSC2* alterations should be identified using NGS in tumor tissue or liquid biopsy and must be determined by analytically validated NGS tests performed in CLIA-certified laboratories. [b]Follow-up is for survival and initiation of anticancer therapy; it is initiated after the EOS visit. **CLIA**, Clinical Laboratory Improvement Amendments; **DCR**, disease control rate; **DOR**, duration of response; **ECOG**, Eastern Cooperative Oncology Group; **EOS**, end of study; **KPS**, Karnofsky Performance Scale; **LPPS**, Lansky Play-Performance Scale; **NGS**, next generation sequencing; **ORR**, overall response rate; **OS**, overall survival; **PFS**, progression-free survival; **PS**, performance status; **QOL**, quality of life; **RECIST**, Response Evaluation Criteria for Solid Tumors; ***TSC1/TSC2***, tuberous sclerosis complex 1 or 2.

- Partnerships with leading next-generation sequencing companies (Foundation Medicine, Tempus, and Caris) and US Oncology will facilitate identification of patients with qualifying inactivating alterations in *TSC1* or *TSC2* and expand access to the study through just-in-time trial locations and accelerated site activation (**Figure 5**)

Figure 5. Expanding PRECISION 1 Access and Enrollment Through Strategic Partnerships

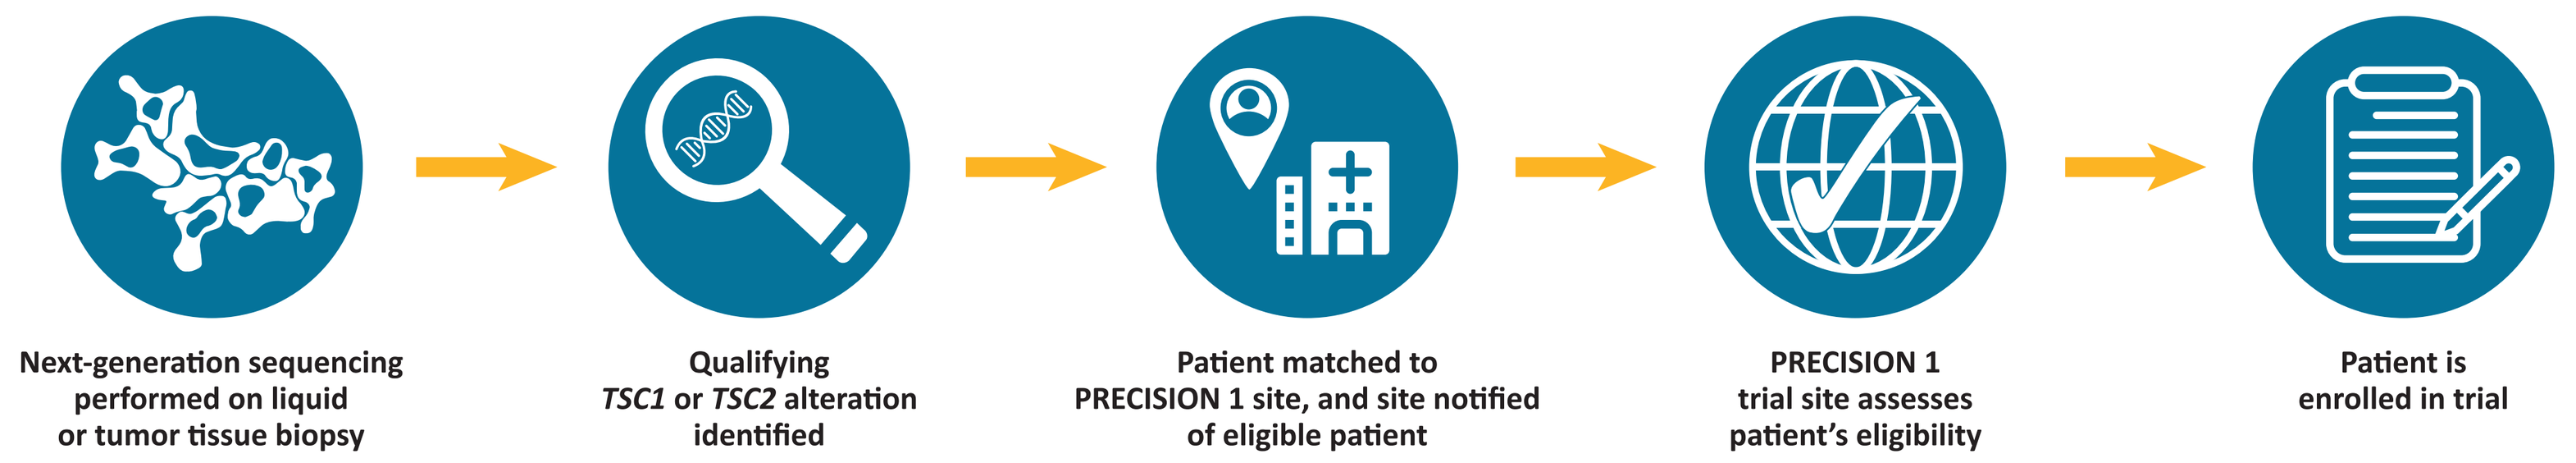
Next-generation sequencing performed on liquid or tumor tissue biopsy
Qualifying TSC1 or TSC2 alteration identified
Patient matched to PRECISION 1 site, and site notified of eligible patient
PRECISION 1 trial site assesses patient's eligibility
Patient is enrolled in trial

Acknowledgments & Disclosures

Editorial assistance was provided by Heather Caballes, PhD of Twist Medical and was funded by Aadi Bioscience, Inc.

DLR reports advisory boards/consulting for Merana, Immunogen, AstraZeneca, GlaxoSmithKline, Eisai and Profound Bio. **GI** reports consulting/advisory for Janssen, Mirati Therapeutics, Basilea, Flare Therapeutics, LOXO at Lilly; speakers fees from Gilead Sciences, the Lynx Group; Research Funding: Mirati Therapeutics, Novartis, Debiopharm Group, Bayer, Janssen, Seagen, Flare Therapeutics. **LD**, **ANS**, and **WHN** are employees of and own stock in Aadi Bioscience. **DJK** reports research funding from Aadi Biosciences, Genentech, Revolution Medicines; and consulting fees from Aadi Biosciences, Bridgebio, Genentech, and Guidepoint. **JRA** reports research funding from Bayer, Novartis; and personal fees from Eli Lilly, Peptomyc, and Servier.

References

1. He Y, et al. *Signal Transduct Target Ther.* 2021;6(1):425.
2. Saxton RA, et al. *Cell.* 2017;169(2):361–371.
3. Pavavra F, et al. *Oxid Med Cell.* 2017;9820181.
4. Hou S, et al. AACR-NCI-EORTC, October 7–10, 2021 [virtual]. Poster P138.
5. FYARRO (sirolimus albumin-bound particles for injectable suspension). Package insert. Aadi Bioscience, Inc.: Pacific Palisades, CA; 2021.
6. Wagner AJ, et al. *J Clin Oncol.* 2021;39(33):3660–3670.
7. Cranmer, LD, et al. AACR, April 14–19, 2023, Orlando, FL. Poster LB288.
8. Wagner AJ, et al. CTOS, November 16–19, 2022, Vancouver, BC. Poster 167.
9. Dickson MA, et al. ASCO, June 4–8, 2021 [virtual]. Abstract 339405.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.