TPS585

PRECISION 1: A phase 2, multicenter, open-label tumor-agnostic trial of *nab*-sirolimus for malignant solid tumors harboring pathogenic inactivating alterations in *TSC1* and *TSC2*

Dustin Deming, MD[1]; David J. Kwiatkowski, MD, PhD[2]; Li Ding, MS, MA[3]; Anita N. Schmid, PhD[3]; Willis H. Navarro, MD[3]; Jordi Rodon Ahnert, MD, PhD[4]; Gopa Iyer, MD[5]

[1]University of Wisconsin Carbone Cancer Center, Madison, WI, USA; [2]Brigham and Women's Hospital, Boston, MA, USA; [3]Aadi Bioscience, Inc., Pacific Palisades, CA, USA; [4]MD Anderson Cancer Center, Houston, TX, USA; [5]Memorial Sloan Kettering Cancer Center, New York, NY, USA

Objective

- **This trial is designed to evaluate the efficacy, safety, and tolerability of *nab*-sirolimus in a patient population with advanced malignancies and limited therapeutic options**

KEY POINTS

- ***nab*-Sirolimus is an mTOR inhibitor (mTORi) approved in the US for the treatment of adult patients with advanced malignant PEComa**
- **Data from the AMPECT exploratory analysis and an expanded access program suggest *nab*-sirolimus may provide clinically relevant benefit with a manageable safety profile in patients with solid tumors harboring inactivating alterations in *TSC1* and/or *TSC2***
- ***TSC1* and/or *TSC2* inactivating alterations have been observed in patients with GI cancers with a frequency of up to 6.6% in liver cancer, 1.6% in colorectal adenocarcinoma, and 0.5% in pancreatic cancer; however, there are no specific treatment options for patients with these alterations**
- **PRECISION 1 (NCT05103358) is a registrational, tumor-agnostic trial for patients with solid tumors driven by *TSC1* or *TSC2* inactivating alterations; the trial is currently enrolling patients with solid tumors with *TSC1* or *TSC2* inactivating alterations**

Presented at the ASCO Gastrointestinal Cancers Symposium; San Francisco, CA; January 18–20, 2024

Correspondence: MedInfo@AadiBio.com

INTRODUCTION

- Gastrointestinal (GI) cancers have a poor prognosis as indicated by low 5-year survival rates in patients with advanced colorectal (15.6%) and liver (3.5%) cancers[1]
- Overactivation of the PI3K-Akt-mTOR pathway, has been implicated in a number of cancers, including GI cancers[2], and can result from inactivation of the tumor suppressor genes *TSC1* and *TSC2*[3] (**Figure 1**)

Figure 1. **PI3K-Akt-mTOR pathway[2,4]**

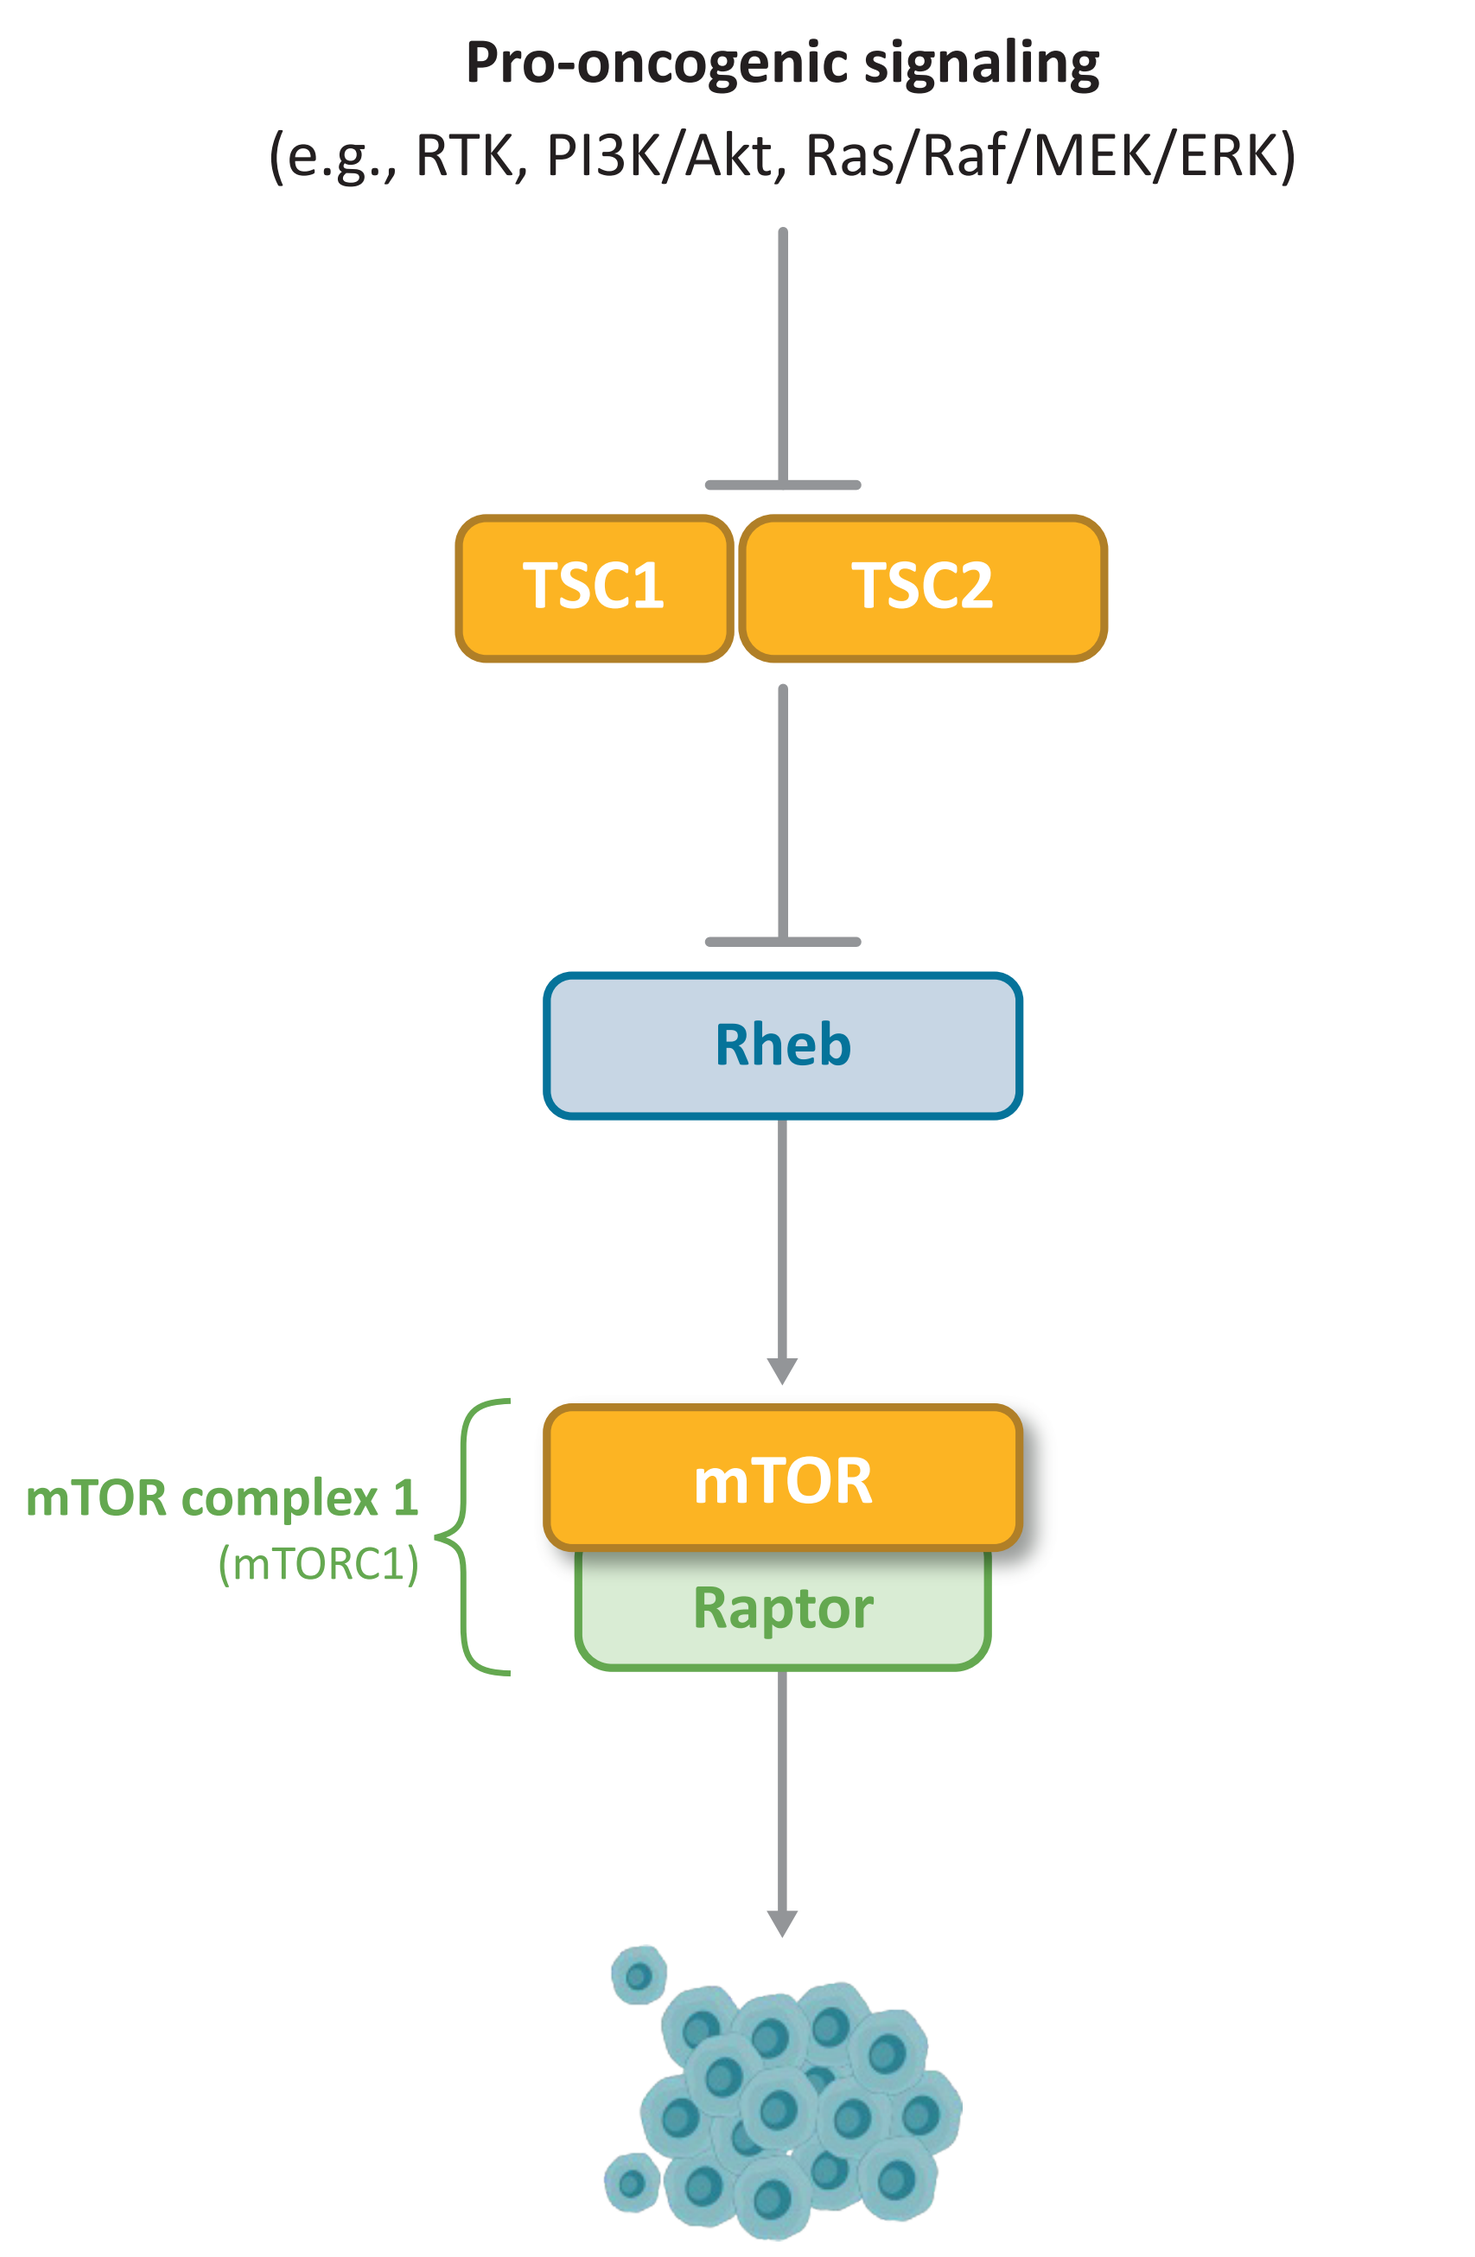
Pro-oncogenic signaling
(e.g., RTK, PI3K/Akt, Ras/Raf/MEK/ERK)
TSC1
TSC2
Rheb
mTOR complex 1
(mTORC1)
mTOR
Raptor

- *nab*-Sirolimus, a nanoparticle albumin-bound, IV-administered mTOR inhibitor, is approved in the United States for the treatment of adult patients with advanced, malignant perivascular epithelioid cell tumor (PEComa)[4] a group of rare aggressive tumors that can originate in GI tissues[5,6]
- In the open-label, phase 2 AMPECT trial (NCT02494570), patients treated with *nab*-sirolimus for malignant PEComa showed clinically meaningful overall response rate, median duration of response of more than 3 years, and durable disease control and survival [7]
- Results from the AMPECT exploratory biomarker analysis demonstrated rapid and durable responses in patients with *TSC1* or *TSC2* inactivating alterations and suggested significant clinical benefit (**Figure 2**)[8]

Figure 2. **Exploratory biomarker analysis from AMPECT demonstrated rapid and durable responses in the subset of patients with known *TSC1* or *TSC2* inactivating alterations treated with *nab*-sirolimus (n=14)**

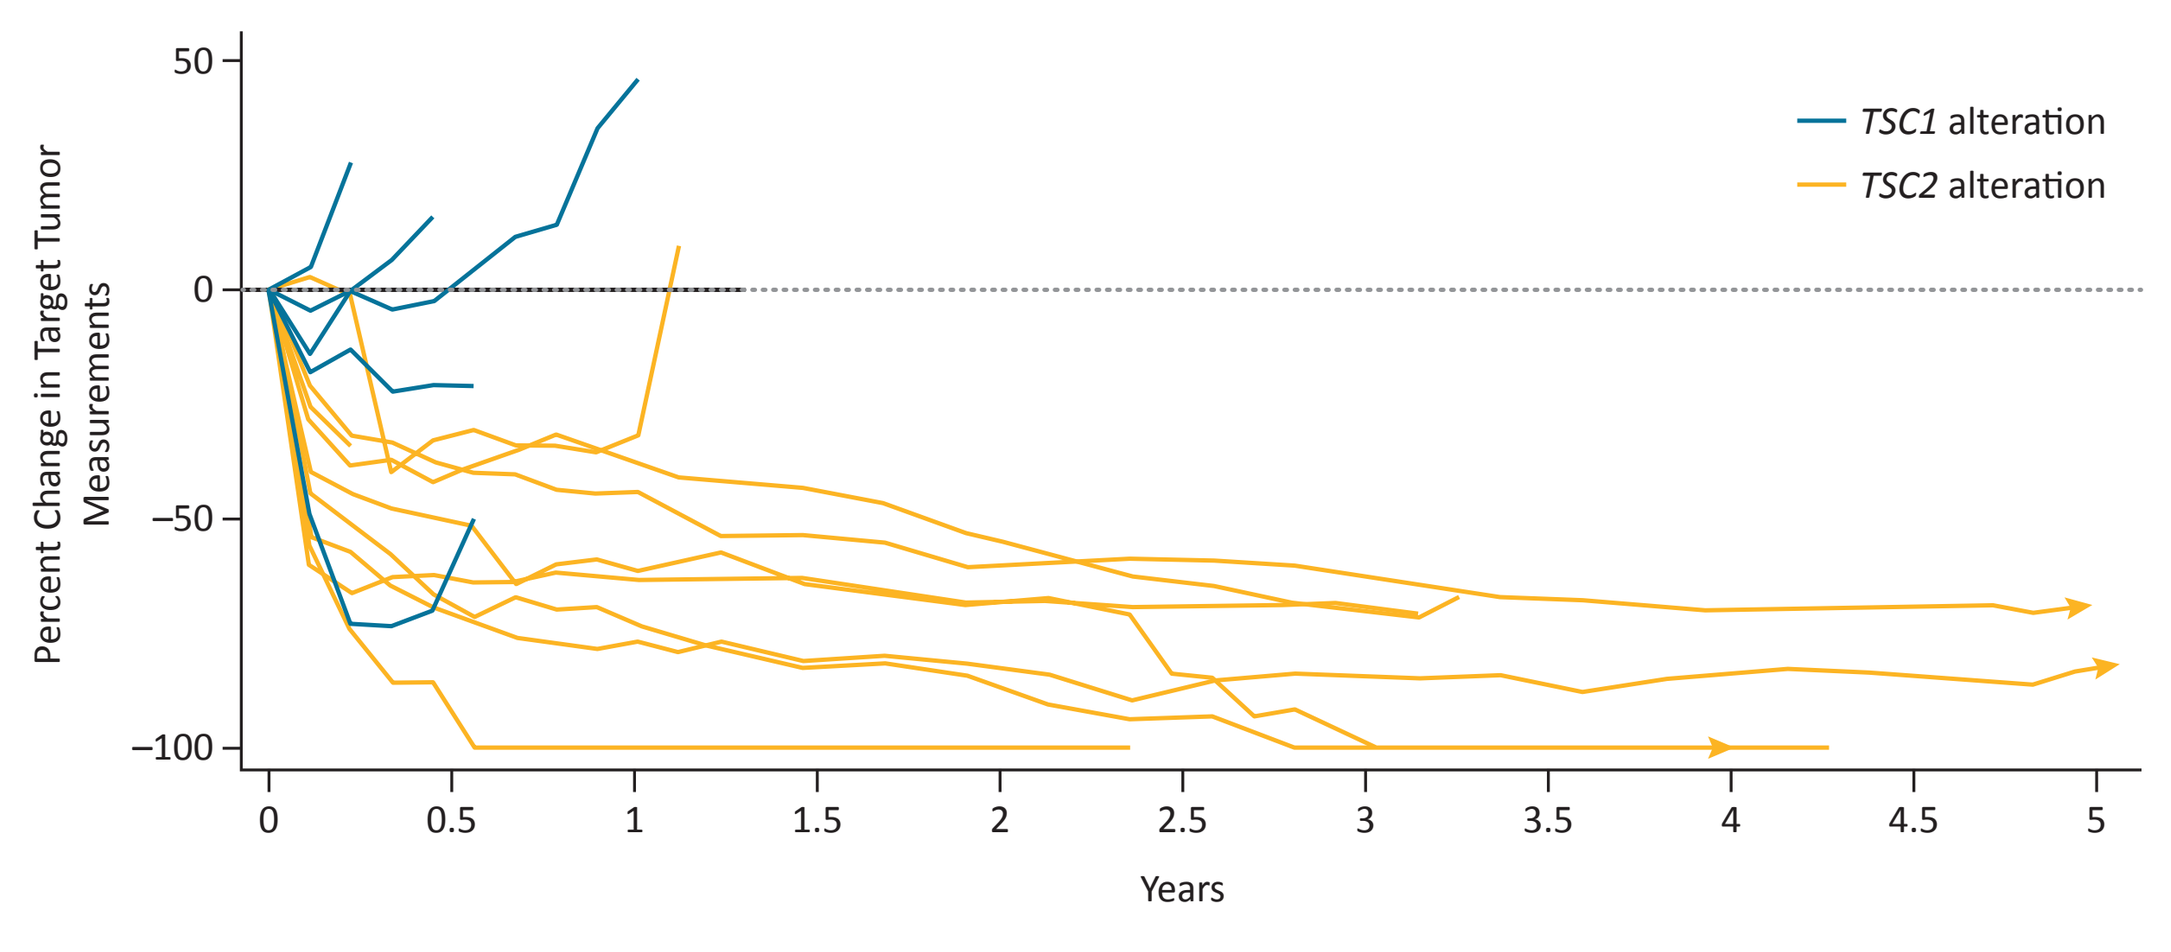
Percent Change in Target Tumor Measurements
50
0
−50
−100
0
0.5
1
1.5
2
2.5
3
3.5
4
4.5
5
Years
TSC1 alteration
TSC2 alteration

- The safety profile in the overall study population was consistent with the mTORi class with no new or unexpected safety signals[8]
- The most common any-grade, nonhematologic treatment-related adverse events (TRAEs) were stomatitis (28/34, [82%]), fatigue (21/34 [62%]), and rash (21/34 [62%]); and the most common, any-grade hematologic TRAEs were anemia (18/34 [53%]) and thrombocytopenia (12/34 [35%])
- Most TRAEs were grade 1/2 and were manageable for long-term treatment; no grade ≥4 TRAEs were observed

- Patients with various malignancies bearing *TSC1* or *TSC2* inactivating alterations treated with *nab*-sirolimus as part of the expanded access program (NCT03817515) showed evidence of response (partial response in 5/7 patients) and manageable toxicities[9]
- Inactivating alterations in *TSC1* and/or *TSC2* have been observed in GI cancers with a combined frequency of up to 6.6% in liver cancer, 1.6% in colorectal adenocarcinoma, and 0.5% in pancreatic cancer (**Figure 3**); however, no treatment options exist specifically for patients with these alterations
- The phase 2 PRECISION 1 trial was initiated to evaluate the potential of mTOR inhibition with *nab*-sirolimus for the treatment of patients with solid tumors harboring inactivating alterations in *TSC1* or *TSC2*

Figure 3. **Estimated frequency of pathogenic and/or inactivating alterations in *TSC1* or *TSC2* by tumor type**

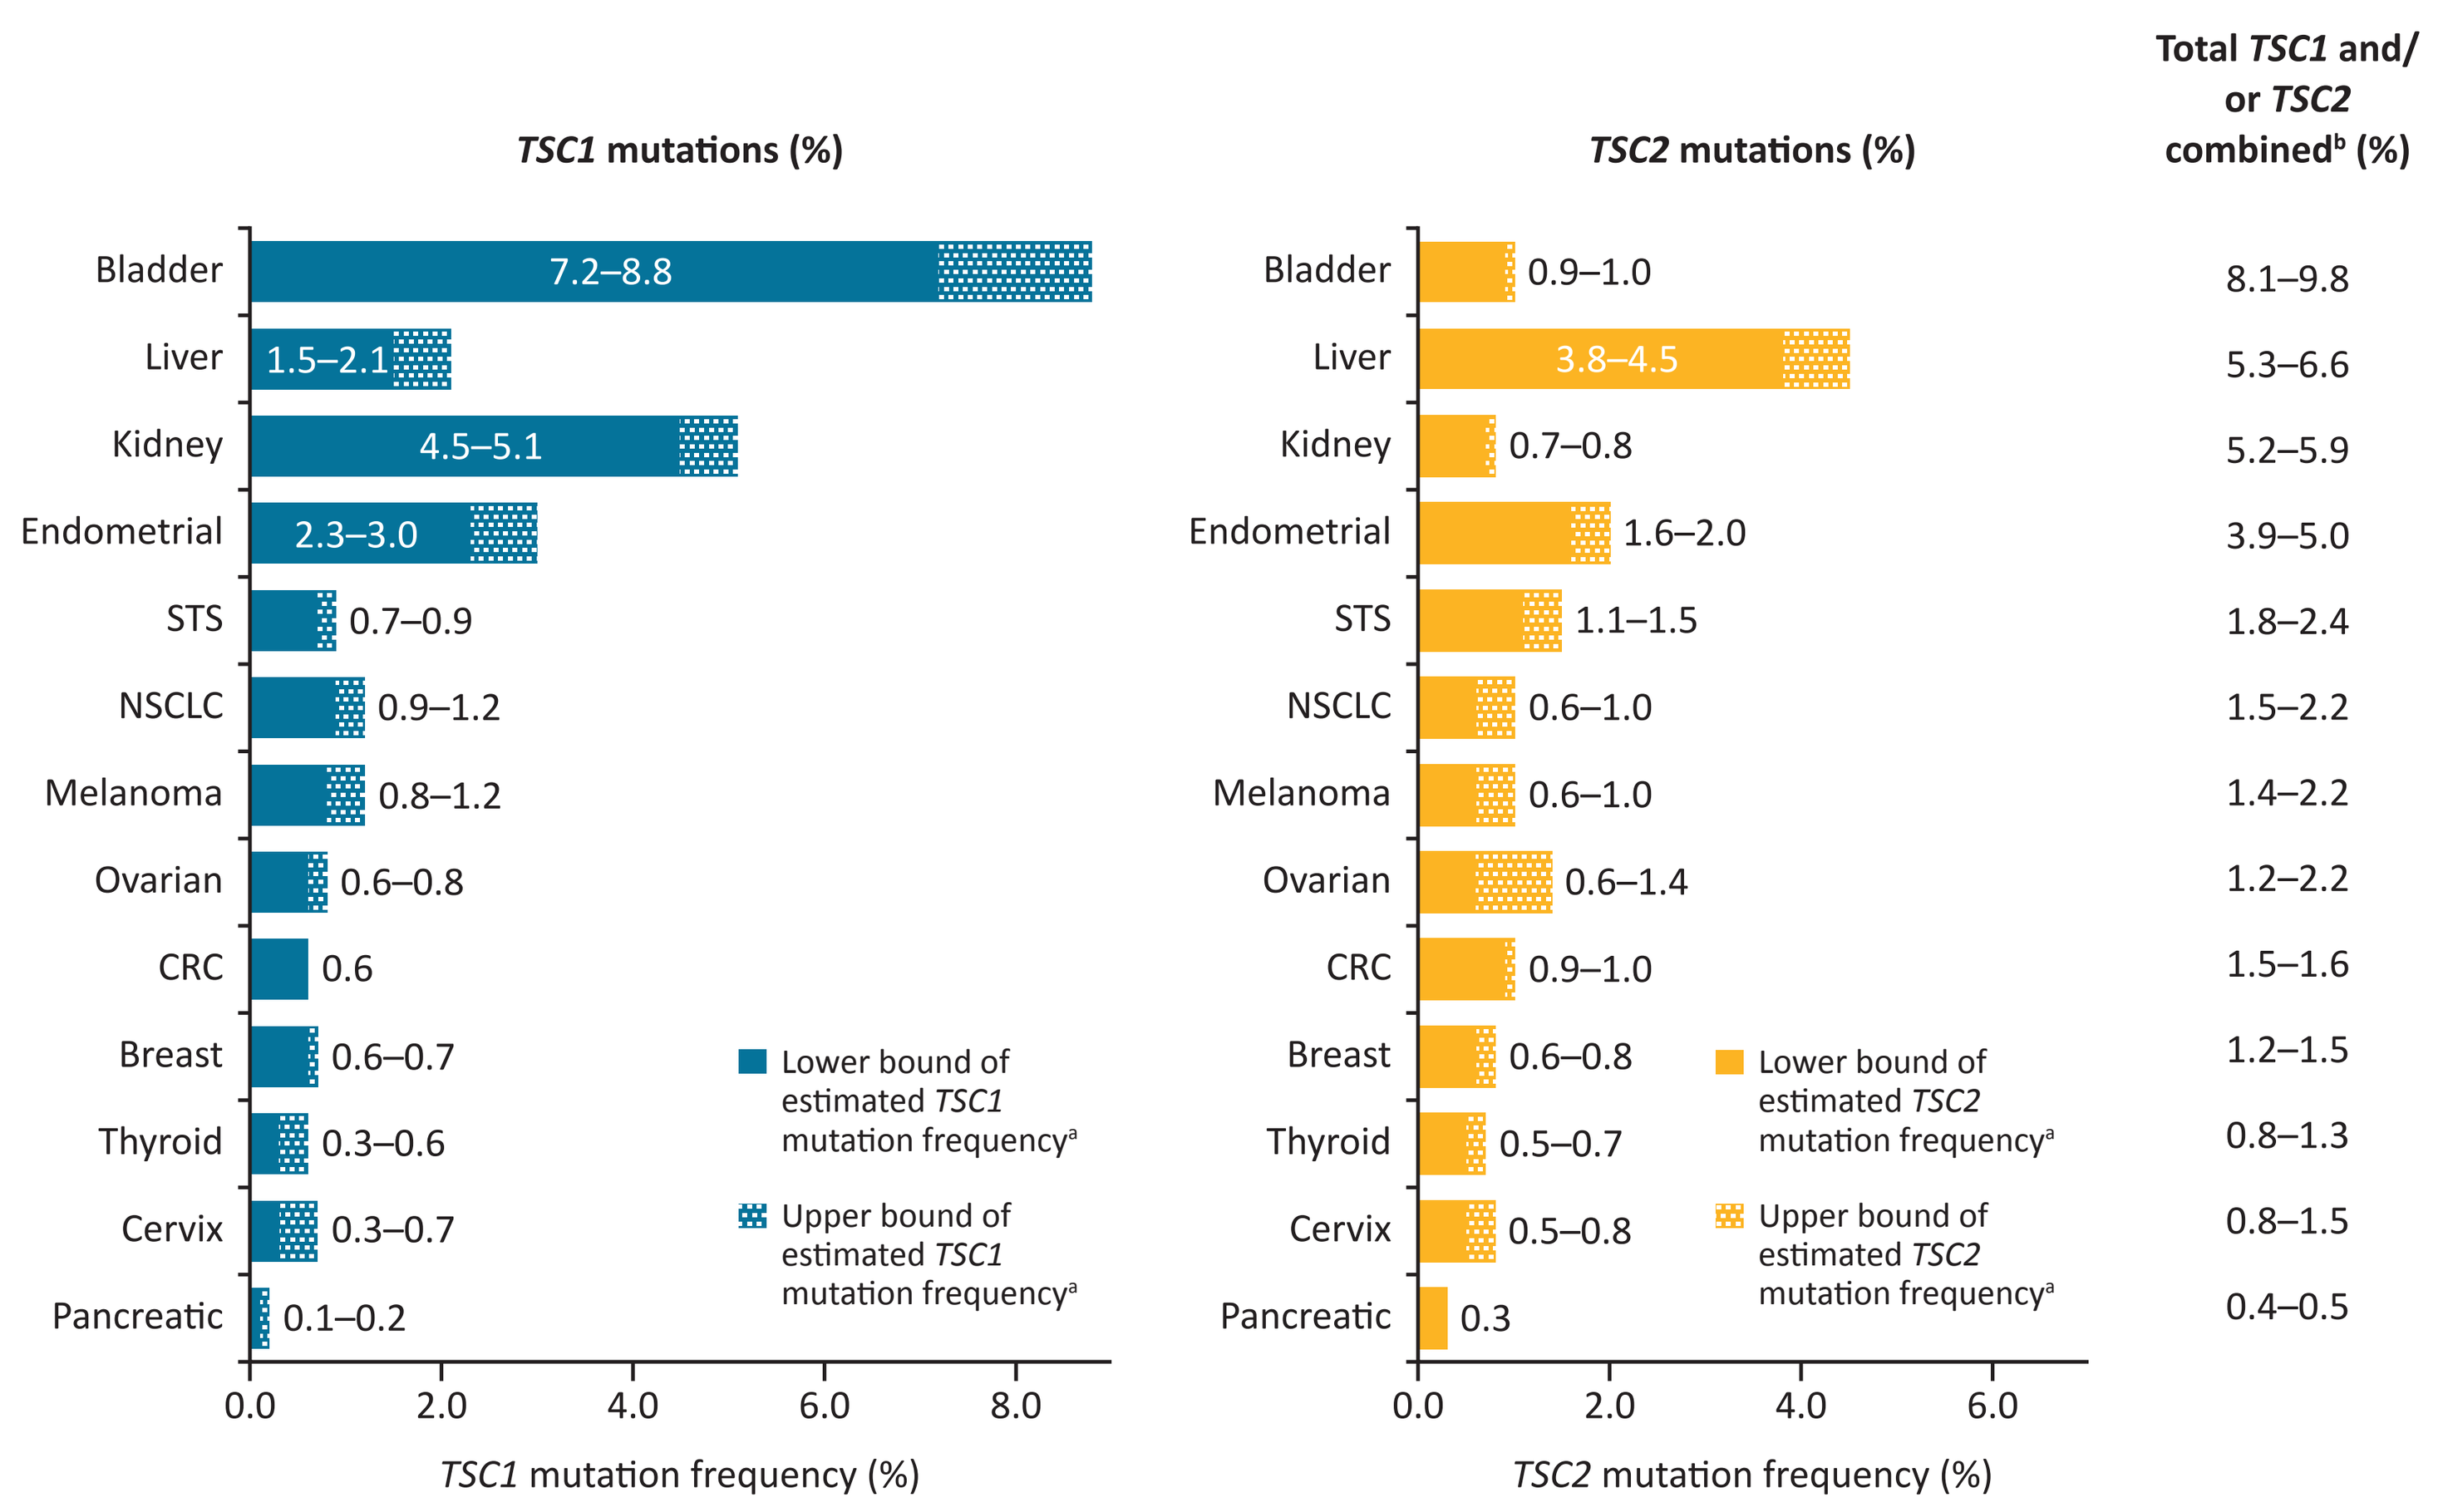
TSC1 mutations (%)
TSC2 mutations (%)
Total TSC1 and/or TSC2 combined[b] (%)
Bladder 7.2–8.8 | 0.9–1.0 | 8.1–9.8
Liver 1.5–2.1 | 3.8–4.5 | 5.3–6.6
Kidney 4.5–5.1 | 0.7–0.8 | 5.2–5.9
Endometrial 2.3–3.0 | 1.6–2.0 | 3.9–5.0
STS 0.7–0.9 | 1.1–1.5 | 1.8–2.4
NSCLC 0.9–1.2 | 0.6–1.0 | 1.5–2.2
Melanoma 0.8–1.2 | 0.6–1.0 | 1.4–2.2
Ovarian 0.6–0.8 | 0.6–1.4 | 1.2–2.2
CRC 0.6 | 0.9–1.0 | 1.5–1.6
Breast 0.6–0.7 | 0.6–0.8 | 1.2–1.5
Thyroid 0.3–0.6 | 0.5–0.7 | 0.8–1.3
Cervix 0.3–0.7 | 0.5–0.8 | 0.8–1.5
Pancreatic 0.1–0.2 | 0.3 | 0.4–0.5
Lower bound of estimated TSC1 mutation frequency[a]
Upper bound of estimated TSC1 mutation frequency[a]
Lower bound of estimated TSC2 mutation frequency[a]
Upper bound of estimated TSC2 mutation frequency[a]
0.0
2.0
4.0
6.0
8.0
TSC1 mutation frequency (%)
TSC2 mutation frequency (%)

[a]Real-world frequencies were derived from tissue-based NGS results for nonhematologic malignancies included in the Foundation Medicine FoundationInsights™ web platform (n=438,974) and data provided by Tempus Labs (n=128,914) with analysis restricted to known or likely oncogenic *TSC1* or *TSC2* inactivating alterations. Upper and lower bounds of frequency ranges were defined as the frequencies calculated from each dataset.
[b]The range is reported as the sum of the highest and lowest values for each gene reported in either dataset. **CRC**, colorectal cancer; **NSCLC**, non–small cell lung cancer; **STS**, soft tissue sarcoma; ***TSC1*/*TSC2***, tuberous sclerosis complex 1/2. Data on file. Aadi Bioscience [May 2023].

STUDY DESIGN

- PRECISION 1 (NCT05103358) is a prospective, phase 2, open-label, multi-institution tumor-agnostic trial evaluating *nab*-sirolimus in patients with malignant solid tumors with pathogenic inactivating alterations in *TSC1* (Arm A) or *TSC2* (Arm B) (**Figure 4**)

Figure 4. **Study design**

Key eligibility criteria

- Metastatic or locally advanced solid tumor with a pathogenic inactivating alteration in *TSC1* or *TSC2* identified using NGS in tumor tissue or liquid biopsy[a]
- ≥12 years of age
- mTORi–naïve
- Patients must have received appropriate standard treatments or be unlikely to tolerate or derive clinically meaningful benefit from standard therapy as determined by the investigator
- ECOG PS 0 or 1 (or KPS ≥80; or LPPS ≥80 for pediatric patients)
- ≥1 measurable target lesion by CT scan or MRI
- Patients with primary brain tumors or PEComa are excluded

Treatment phase

Arm A: Pathogenic inactivating alterations in *TSC1* n=~60	Arm B: Pathogenic inactivating alterations in *TSC2* n=~60

- *nab*-Sirolimus 100 mg/m² administered as an intravenous infusion over 30 minutes on Days 1 and 8 of each 21-day cycle
- Dose reduction allowed per protocol at 75, 56, 45, and 35 mg/m²

***nab*-Sirolimus dosing schedule**

21-day Cycle
Day 1 · Day 8 · Day 15
Repeat until disease progression, unacceptable toxicity, withdrawal of consent, or at investigator discretion

Patients are enrolled in the United States and globally - NCT05103358

Follow-up

- Response assessment every 6 weeks for the first year, then every 12 weeks thereafter, until disease progression
- Quarterly follow-up until EOS, death, or consent withdrawal[b]

Endpoints

Primary endpoint
- ORR by independent radiology review (RECIST v1.1)

Secondary endpoints
- DOR, DCR, PFS by independent radiology review, OS, QoL, safety and tolerability

Key exploratory endpoints
- Investigator-assessed response assessment; time on treatment; association between mutational profile and clinical outcomes; and rate of surgical resection if inoperable at baseline

[a]Central confirmation of *TSC1* and *TSC2* inactivating alterations is via evaluation of NGS reports. Patients will be enrolled only after central confirmation of eligibility. *TSC1* and *TSC2* alterations should be identified using NGS in tumor tissue or liquid biopsy and must be determined by analytically validated NGS tests performed in CLIA-certified laboratories. [b]Follow-up is for survival and initiation of anticancer therapy; it is initiated after the EOS visit.
CLIA, Clinical Laboratory Improvement Amendments; **DCR**, disease control rate; **DOR**, duration of response; **ECOG**, Eastern Cooperative Oncology Group; **EOS**, end of study; **KPS**, Karnofsky Performance Scale; **LPPS**, Lansky Play-Performance Scale; **NGS**, next generation sequencing; **ORR**, overall response rate; **OS**, overall survival; **PFS**, progression-free survival; **PS**, performance status; **QOL**, quality of life; **RECIST**, Response Evaluation Criteria for Solid Tumors; ***TSC1*/*TSC2***, tuberous sclerosis complex 1 or 2.

- Partnerships with leading next-generation sequencing companies (Foundation Medicine, Tempus, and Caris) and US Oncology will facilitate identification of patients with qualifying inactivating alterations in *TSC1* or *TSC2* and expand access to the study through just-in-time trial locations and accelerated site activation (**Figure 5**)

Figure 5. **Expanding PRECISION 1 access and enrollment through strategic partnerships**

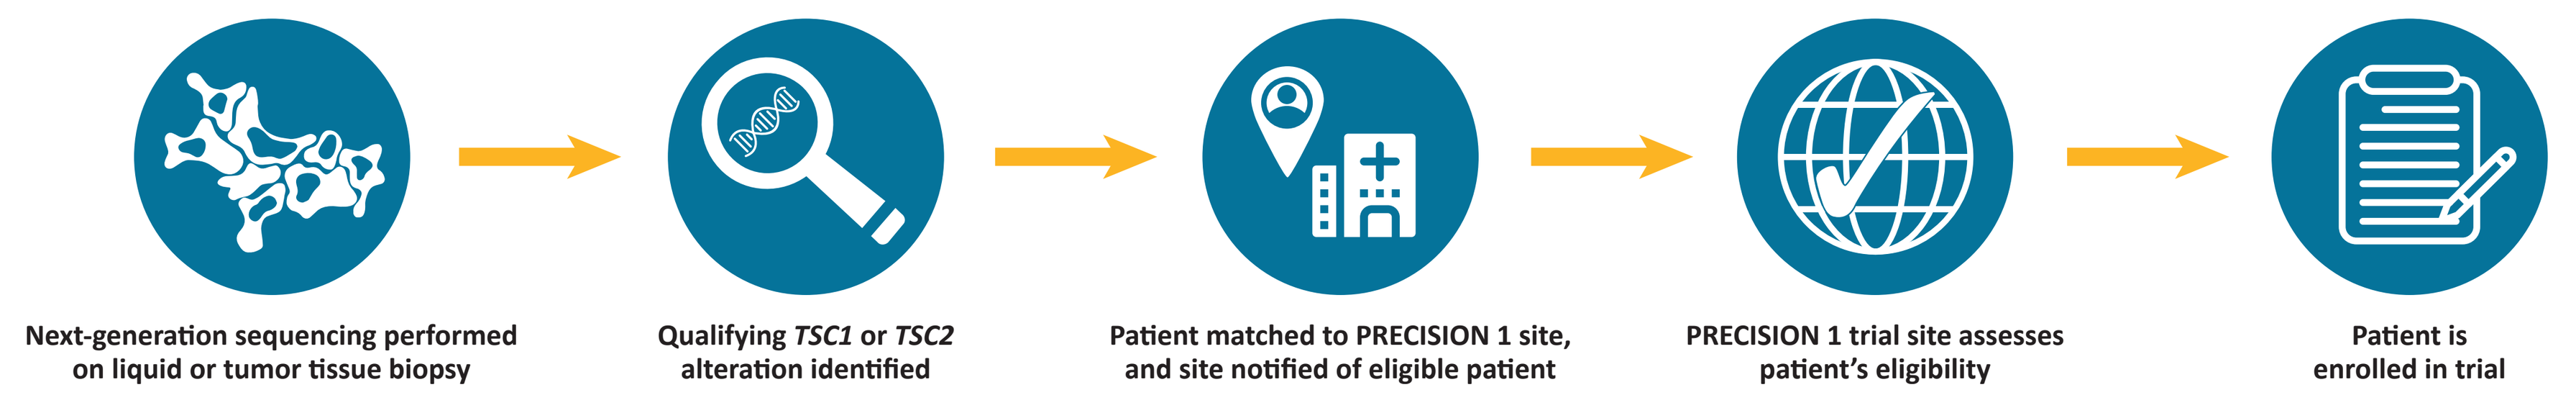
Next-generation sequencing performed on liquid or tumor tissue biopsy
Qualifying TSC1 or TSC2 alteration identified
Patient matched to PRECISION 1 site, and site notified of eligible patient
PRECISION 1 trial site assesses patient's eligibility
Patient is enrolled in trial

Acknowledgments & Disclosures

Editorial assistance was provided by Heather Caballes, PhD of Twist Medical and was funded by Aadi Bioscience, Inc.

Dr. Dustin Deming reports research funding from Merck, Genentech, Bristol Myers Squibb, Aadi Bioscience, Inc., Pfizer, Curegenix, Promega, Natera, STRATA Oncology, Cornerstone Pharmaceuticals, Arcus, Guardant Health, Ipsen, Takeda, Eli Lilly, and Revolution Medicine; consulting or advisory board membership for Bayer, Pfizer, Seagen, Eli Lilly, Foundation Medicine, Illumina, Regeneron, and Aadi Bioscience, Inc.; and participation in the data safety monitoring board for Seagen.

For the disclosures of his co-authors please refer to the abstract.

References

1. Surveillance, Epidemiology, and End Results (SEER) Program SEER*Stat Database: Incidence – SEER Research Data, 22 Registries, November 2022 Sub (1975-2020). https://seer.cancer.gov/statistics-network/explorer accessed on November 3, 2023.
2. Millis SZ, et al. *Cancer.* 2019;125:1185-1199.
3. Huang J, Manning BD. *Biochem J.* 2008:412(2):179-190.
4. FYARRO® (sirolimus protein-bound particles for injectable suspension [albumin-bound]). Package insert. Aadi Bioscience, Inc.: Pacific Palisades, CA; December 2021.
5. Armah HB, Parwani AV. *Arch Pathol Lab Med.* 2009;133:648-654.
6. Bleeker JS, et al. *Sarcoma.* 2012;2012:541626.
7. Wagner AJ, et al. CTOS, November 16–19, 2022, Vancouver, BC. Poster 167.
8. Cranmer, LD, et al. AACR, April 14–19, 2023, Orlando, FL. Poster LB288.
9. Dickson MA, et al. ASCO, June 4–8, 2021 [virtual]. Abstract 339405.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster